TRIM21 is a novel endogenous partner of the inhibitory myeloid checkpoint CLEC-1 involved in tumor antigen cross-presentation

AACR 2023
Poster #6365

Irène Baccelli[1 ‡], Vanessa Gauttier[1 ‡], Marion Drouin[1 ‡], Emmanuelle Wilhelm[1], Caroline Mary[1], Stéphanie Neyton, Mylène Deramé[1], Isabelle Girault[1], Julien Taurelle[1], Ariane Desselle[1], Marion Colonello[1], Sabrina Pengam[1], Géraldine Teppaz[1], Virginie Thépénier[1], Nicolas Poirier[1 ‡], Elise Chiffoleau[2 ‡]

[1]Ose Immunotherapeutics, Nantes, France; [2]Université de Nantes, [CHU Nantes], Inserm, Centre de Recherche en Transplantation et Immunologie, UMR 1064, ITUN, F-44000 Nantes, France, ; ‡ equal contribution

INTRODUCTION

Myeloid cells represent the most abundant immune component of the tumor microenvironment, where they often assume immunosuppressive roles. Therefore, understanding the mechanisms regulating the immunosuppressive activity of myeloid cells is of major interest in oncology.

We have identified CLEC-1, a member of the C-type lectin receptor (CLR) family, as a myeloid cell-driven brake for anti-tumor T cell response in different murine models, including mesothelioma, hepatocellular and colorectal carcinomas[1]. The molecular mechanism underlying the immunomodulatory role of CLEC-1 remained to be fully elucidated.

Here, using both murine and human cells, we showed that CLEC-1 recognized a protein expressed in cells following stress. We identified TRIM21 as a novel endogenous ligand for CLEC-1, shedding new light on CLEC-1 driven inhibitory myeloid checkpoint activity.

RESULTS

1. Single cell RNA sequencing identifies subpopulations of CLEC-1 expressing human myeloid cells, such as cDC1 dendritic cells and TAMs

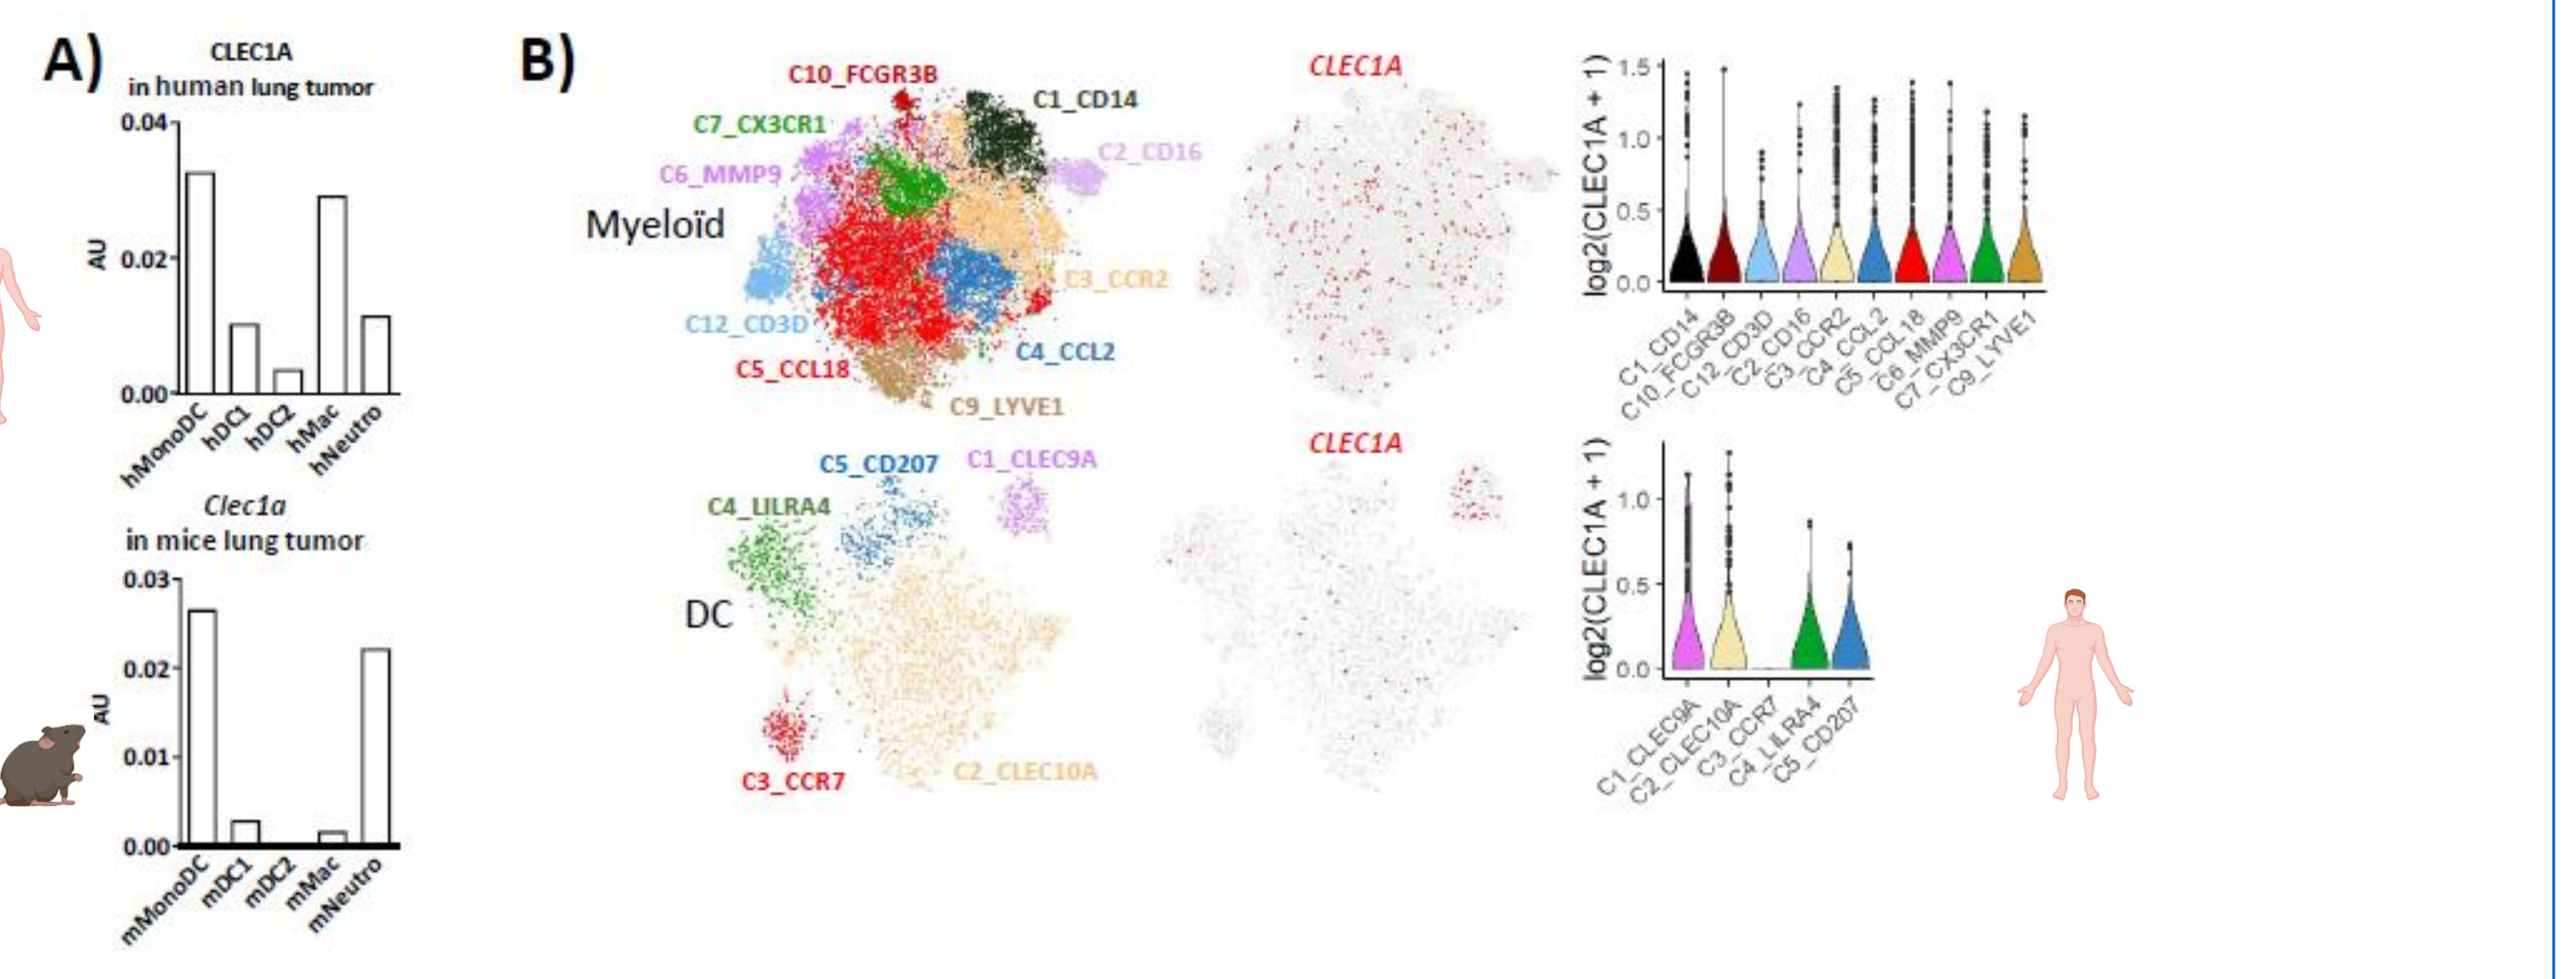
A)
CLEC1A
in human lung tumor
Clec1a
in mice lung tumor
B)
Myeloïd
DC
CLEC1A

Using scRNA-seq dataset of pan-cancer (breast, colorectal, lung, and ovarian), we confirmed that CLEC-1 is expressed by human and murine myeloid cells, especially by cDC1 cells and by tumor-associated macrophages (TAMs) in tumors **(A and B)**.

2. CLEC-1 genetic deletion enhances cDC1 cross priming of CD8+T cells

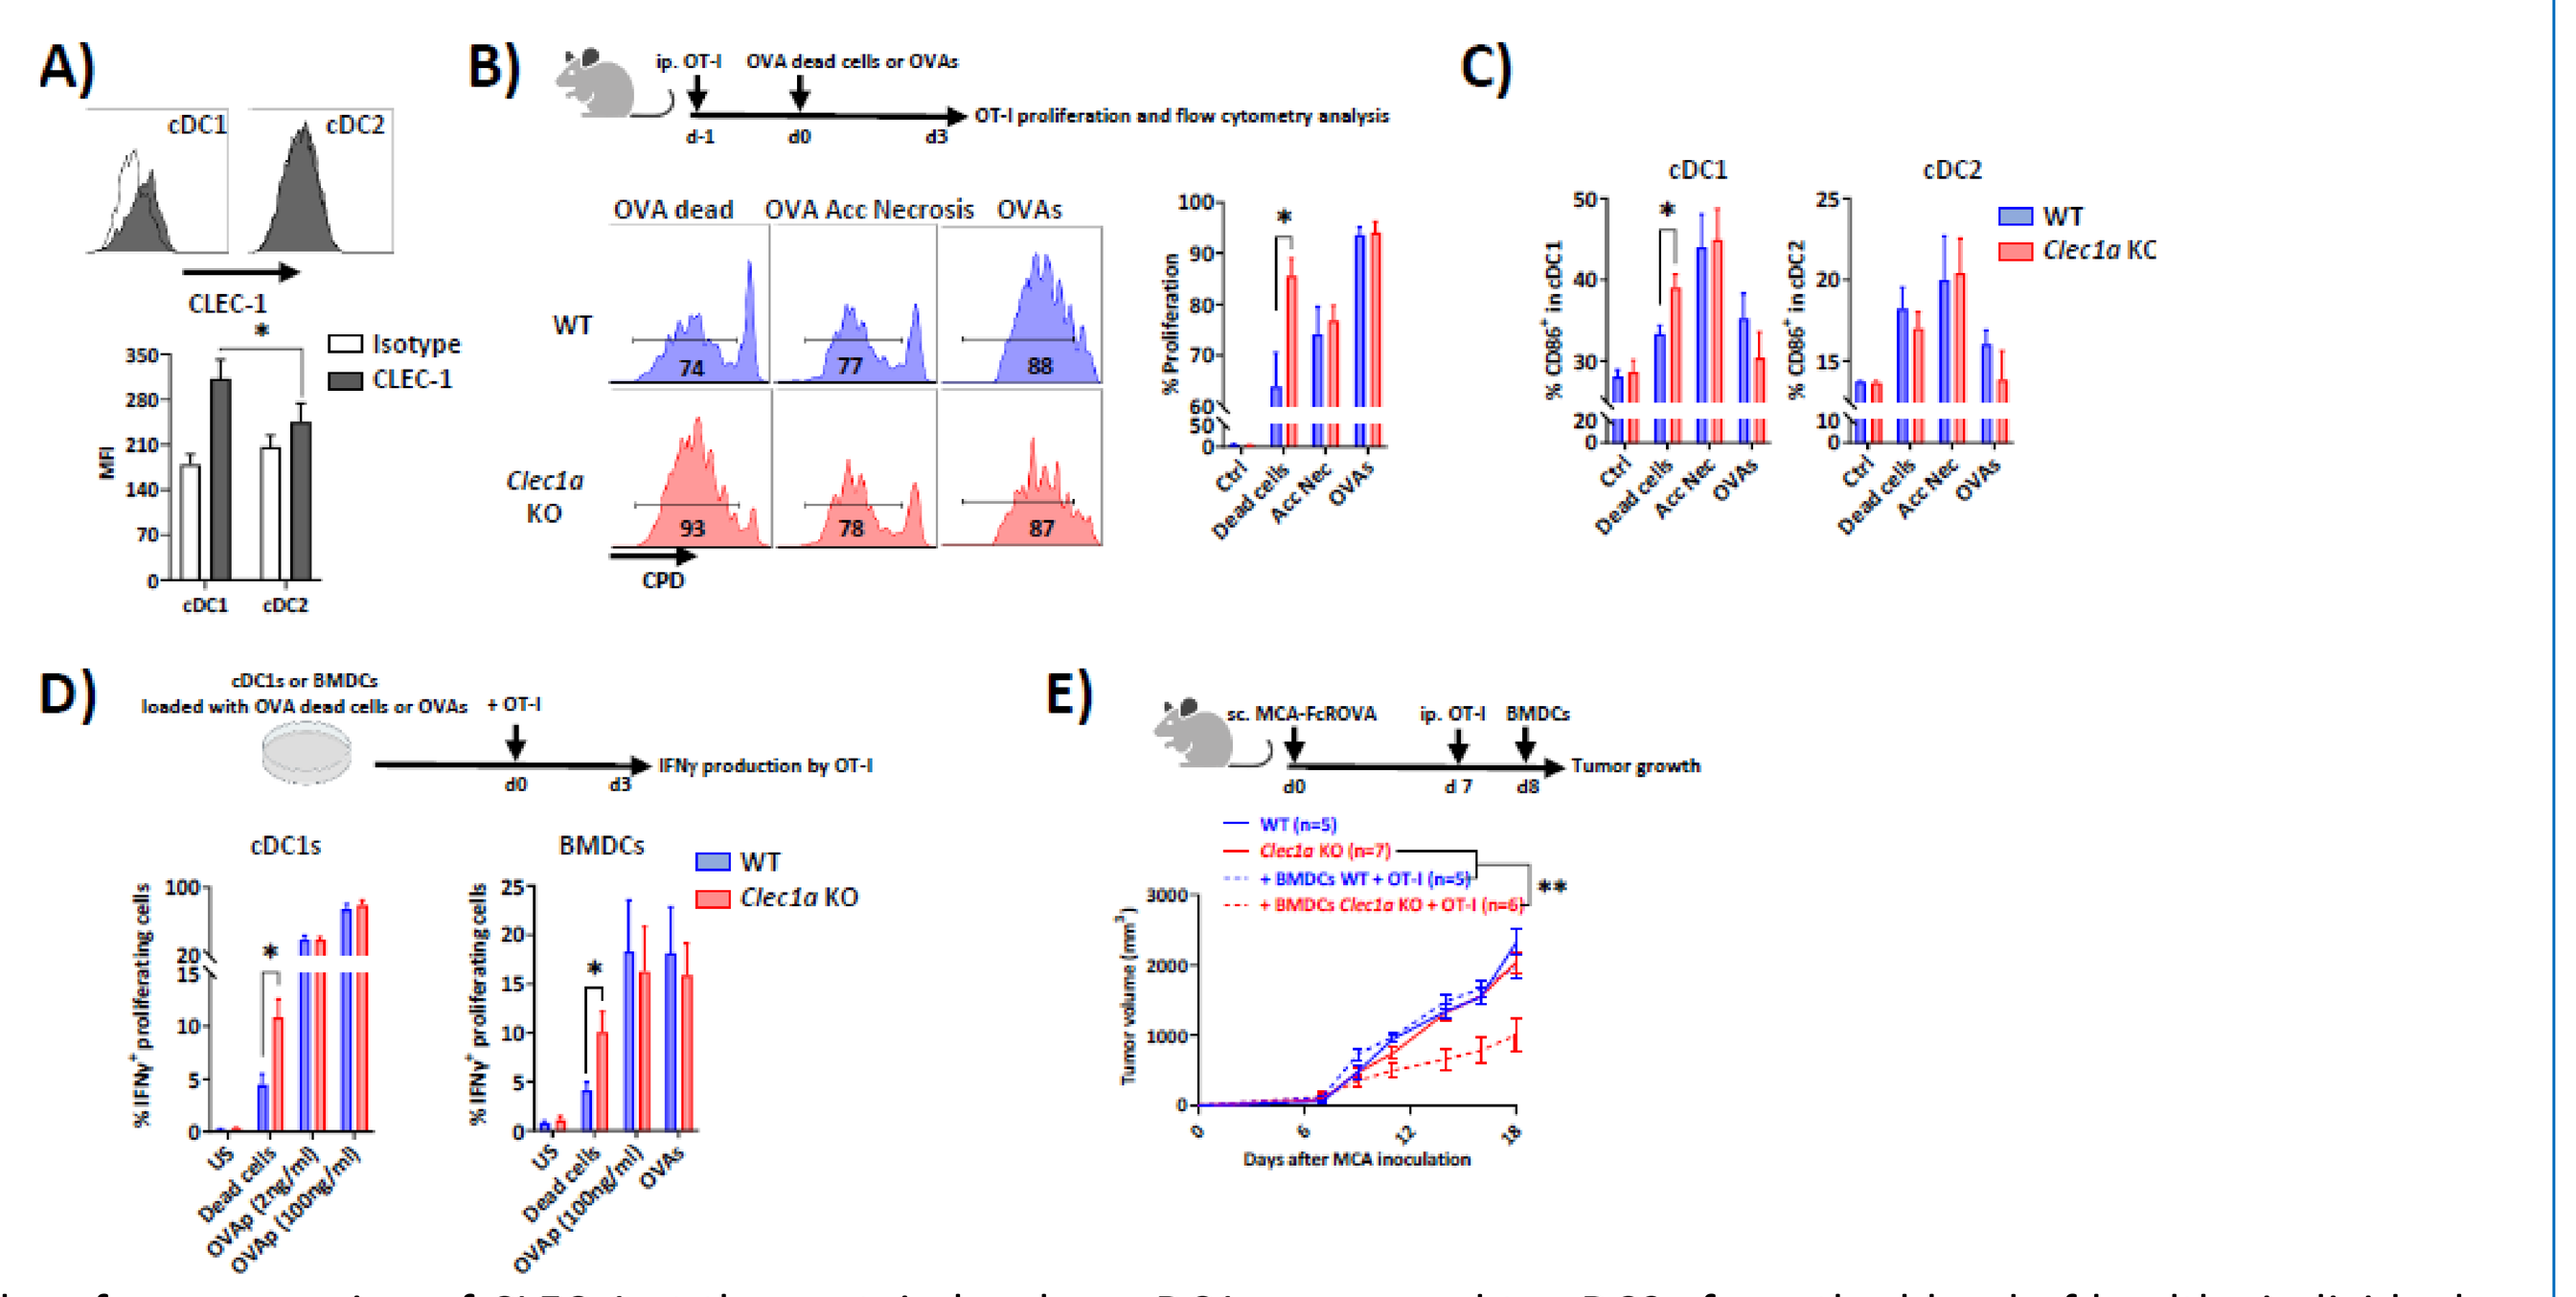
A)
B)
C)
D)
E)
OT-I proliferation and flow cytometry analysis
IFNγ production by OT-I
Tumor growth
Days after MCA inoculation

A higher cell-surface expression of CLEC-1 at the protein level on cDC1s compared to cDC2s from the blood of healthy individuals was observed (**A**). After co-injection with dead MCA101-FcR OVA cells and OT-I CD8+ T cells, an increase of OT-I proliferation was observed in *Clec1a* KO (**B**). cDC1s from the *Clec1a* KO mice further up-regulated the costimulation marker CD86 (**C**). Results were confirmed *in vitro* with a co-culture with DCs loaded with dead MCA101-FcR OVA cells and OT-I CD8+ T cells (**D**). Co-injection of preactivated OT-I cells with *Clec1a* KO DCs loaded with necrotic dead cell–associated mOVA antigen in mice previously injected with MCA tumors expressing mOVA delayed the growth (**E**).

3. An endogenous CLEC-1 ligand is exposed by dying cells under stress conditions

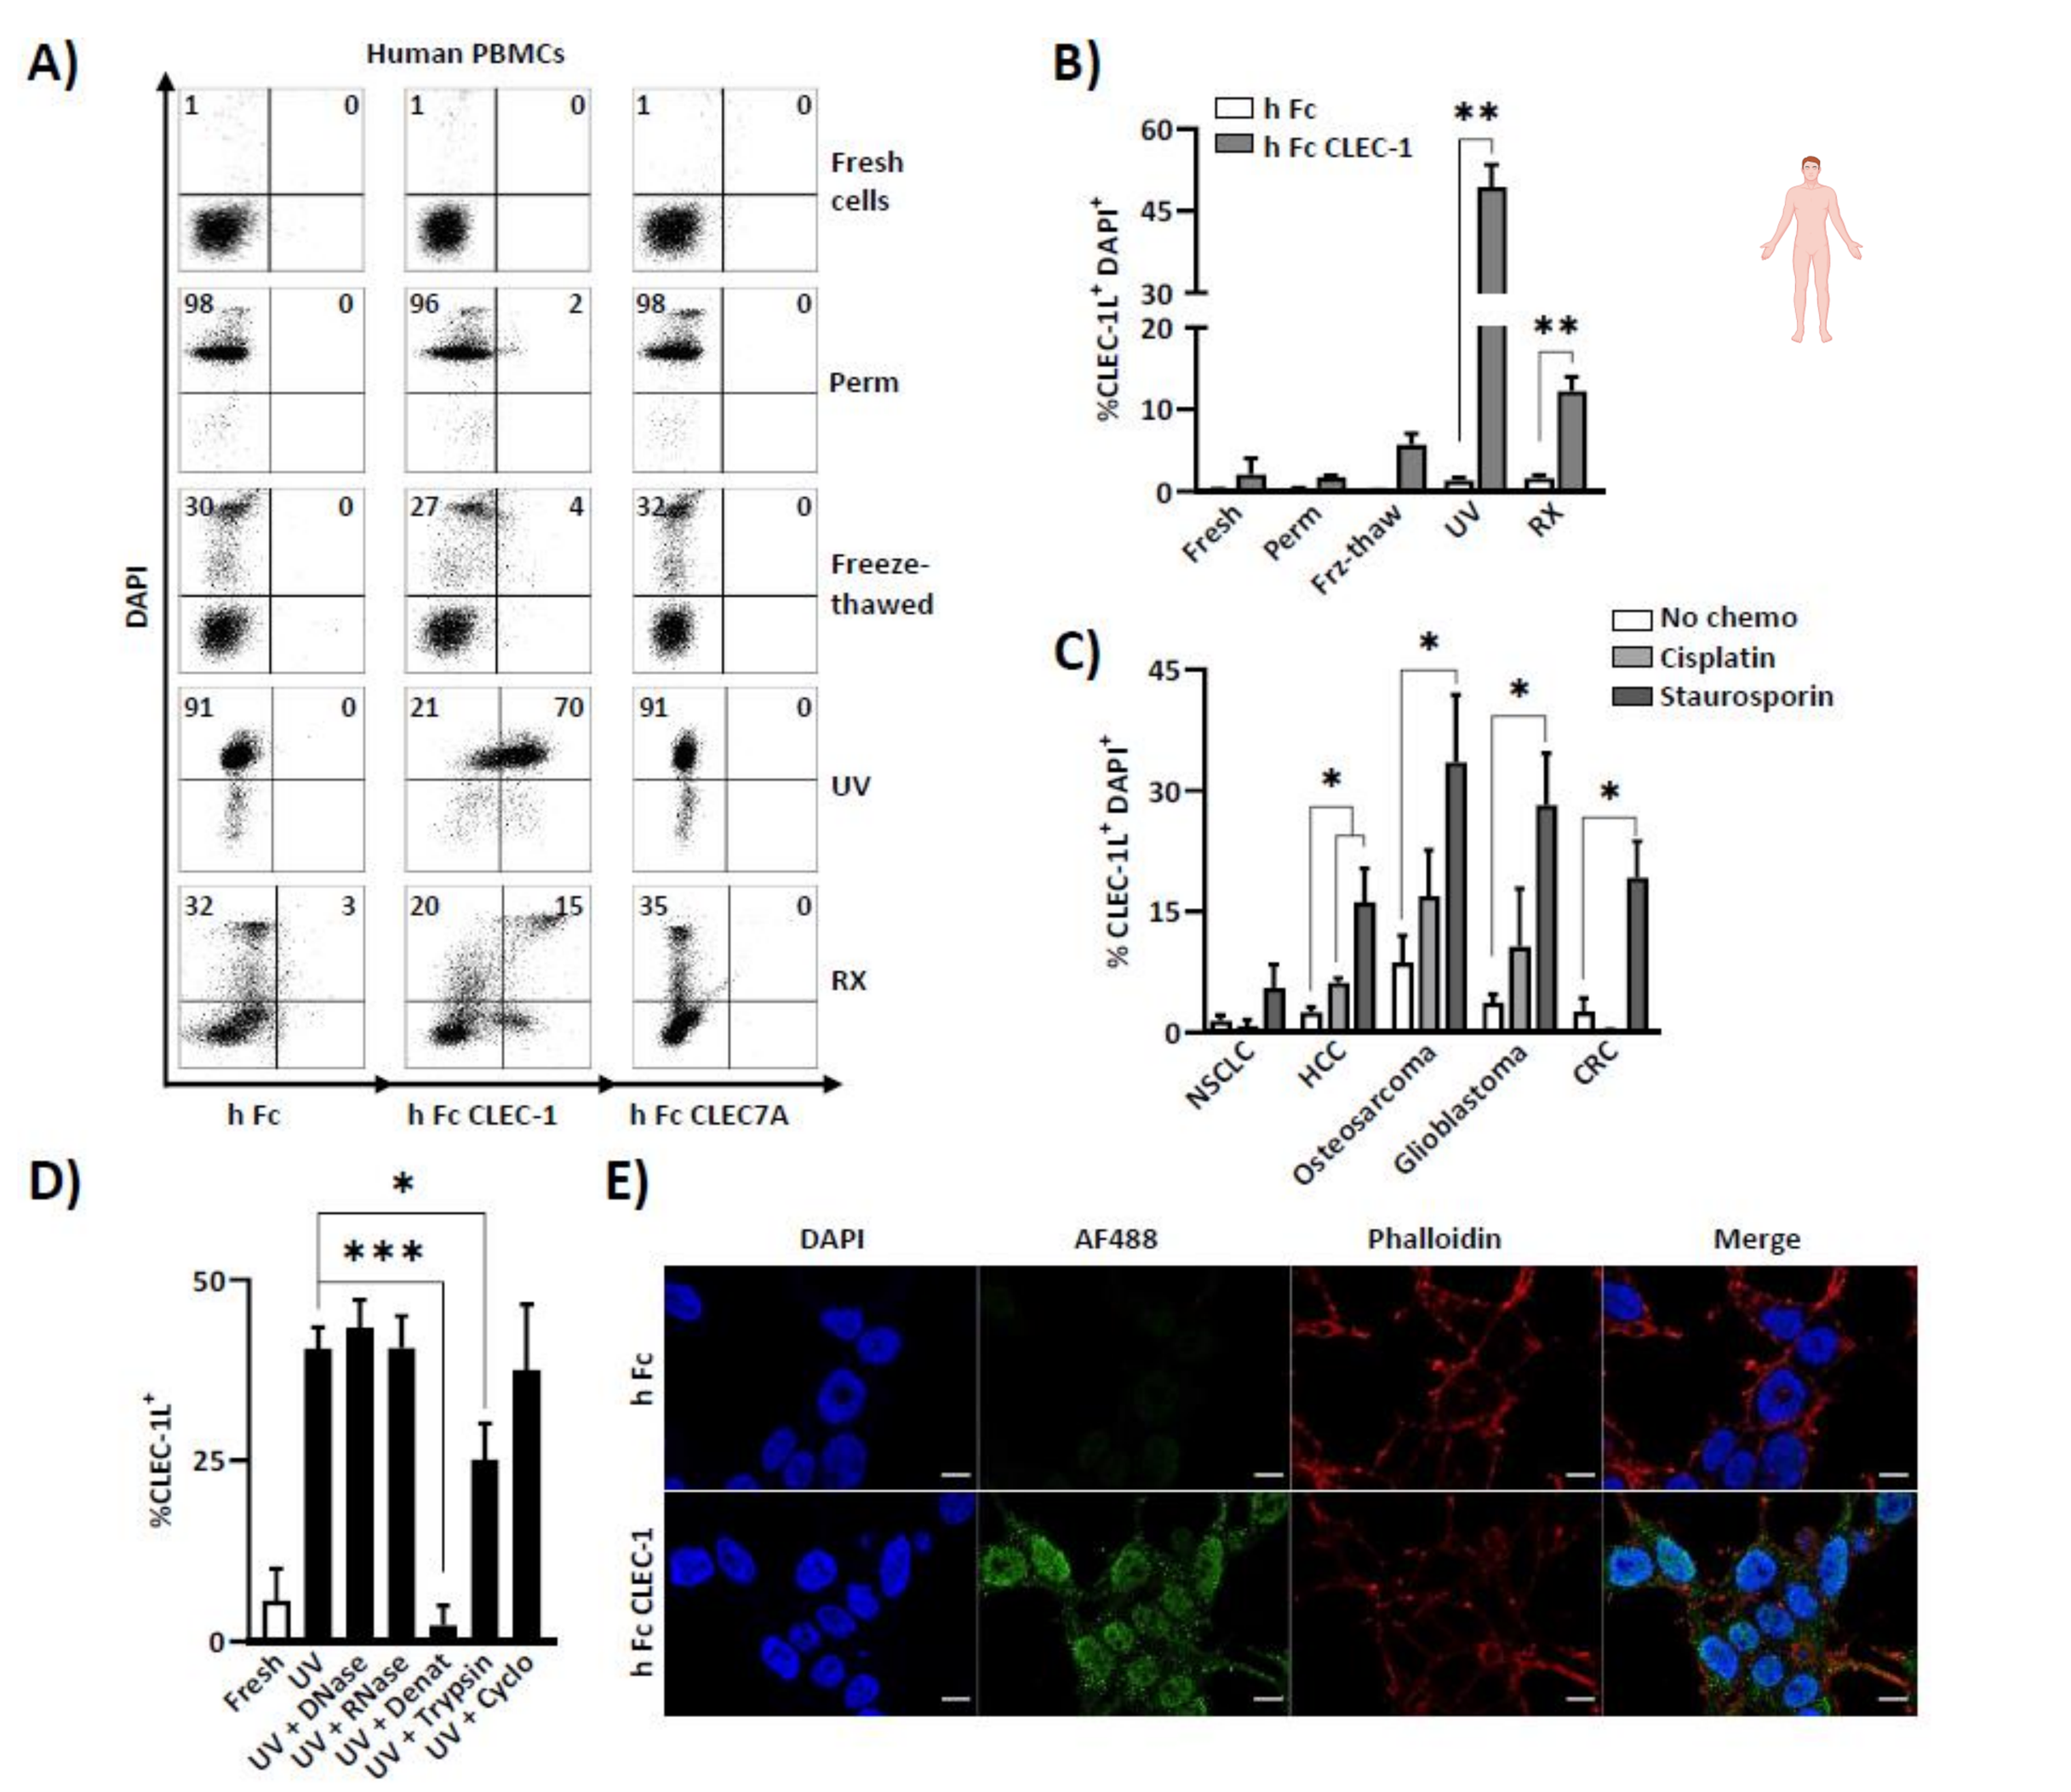
A)
Human PBMCs
Fresh cells
Perm
Freeze-thawed
UV
RX
h Fc
h Fc CLEC-1
h Fc CLEC7A
DAPI
B)
h Fc
h Fc CLEC-1
C)
No chemo
Cisplatin
Staurosporin
NSCLC
HCC
Osteosarcoma
Glioblastoma
CRC
D)
E)
DAPI
AF488
Phalloidin
Merge

Using Fc Ctrl, Fc CLEC7A (an irrelevant CLR), or Fc CLEC-1 staining, we observed that CLEC1 recognizes necrotic human PBMCs previously treated by 150mJ/cm² UV radiation or 10Gy X–radiation 18 hours early (**A and B**). Human tumor cell lines treated with 20µM of cisplatin or 1µM of staurosporin expressed strongly CLEC-1 ligands (**C**). After treatment with DNase A (10 µg/ml), RNase I (50 U/ml), heat-denaturated (20 min at 65°C), trypsin (100 µg/ml) or cycloheximide (100 µg/ml), the binding of human Fc CLEC-1 decreased when dead cells were heat-denaturated or treated with trypsin to digest proteins (**D**). Confocal microscopy showed that CLEC-1 ligands localized in the cytoplasm and the nucleus of dying cells (**E**).
(NSCLC : non-small cells lung cancer A549; HCC : hepatocellular carcinoma Huh7; OS : osteosarcoma U2OS; GB : glioblastoma U373; CRC : colorectal cancer HT-29)

4. CLEC-1 interaction with its endogenous ligand is functional

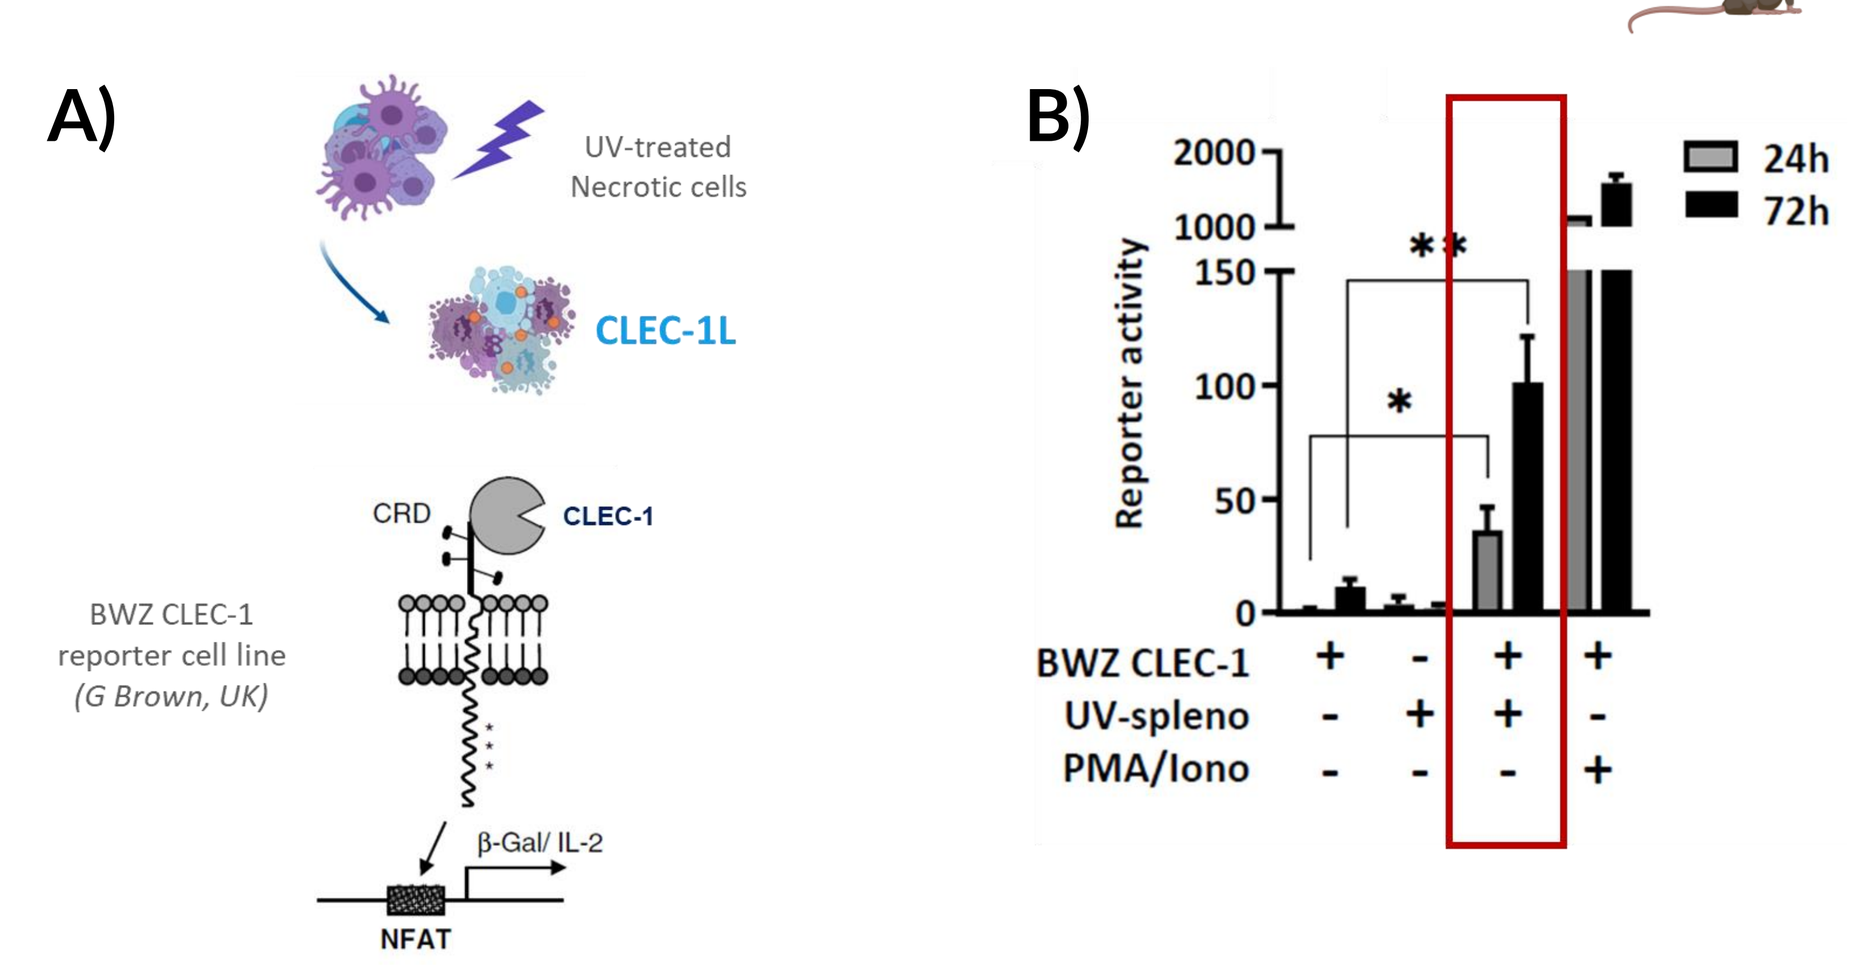
A)
UV-treated
Necrotic cells
CLEC-1L
CRD
CLEC-1
BWZ CLEC-1
reporter cell line
(G Brown, UK)
β-Gal/ IL-2
NFAT
B)
24h
72h
Reporter activity
BWZ CLEC-1
UV-spleno
PMA/Iono

The binding of necrotic cells to CLEC-1 was able to trigger a time-dependent signal in a mouse CLEC-1 reporter cell line expressing the extracellular and transmembrane domains of murine CLEC-1 fused to CD3 zeta (**A and B**).

5. Inhibition of CLEC-1 with its endogenous ligand promotes anti-tumor response

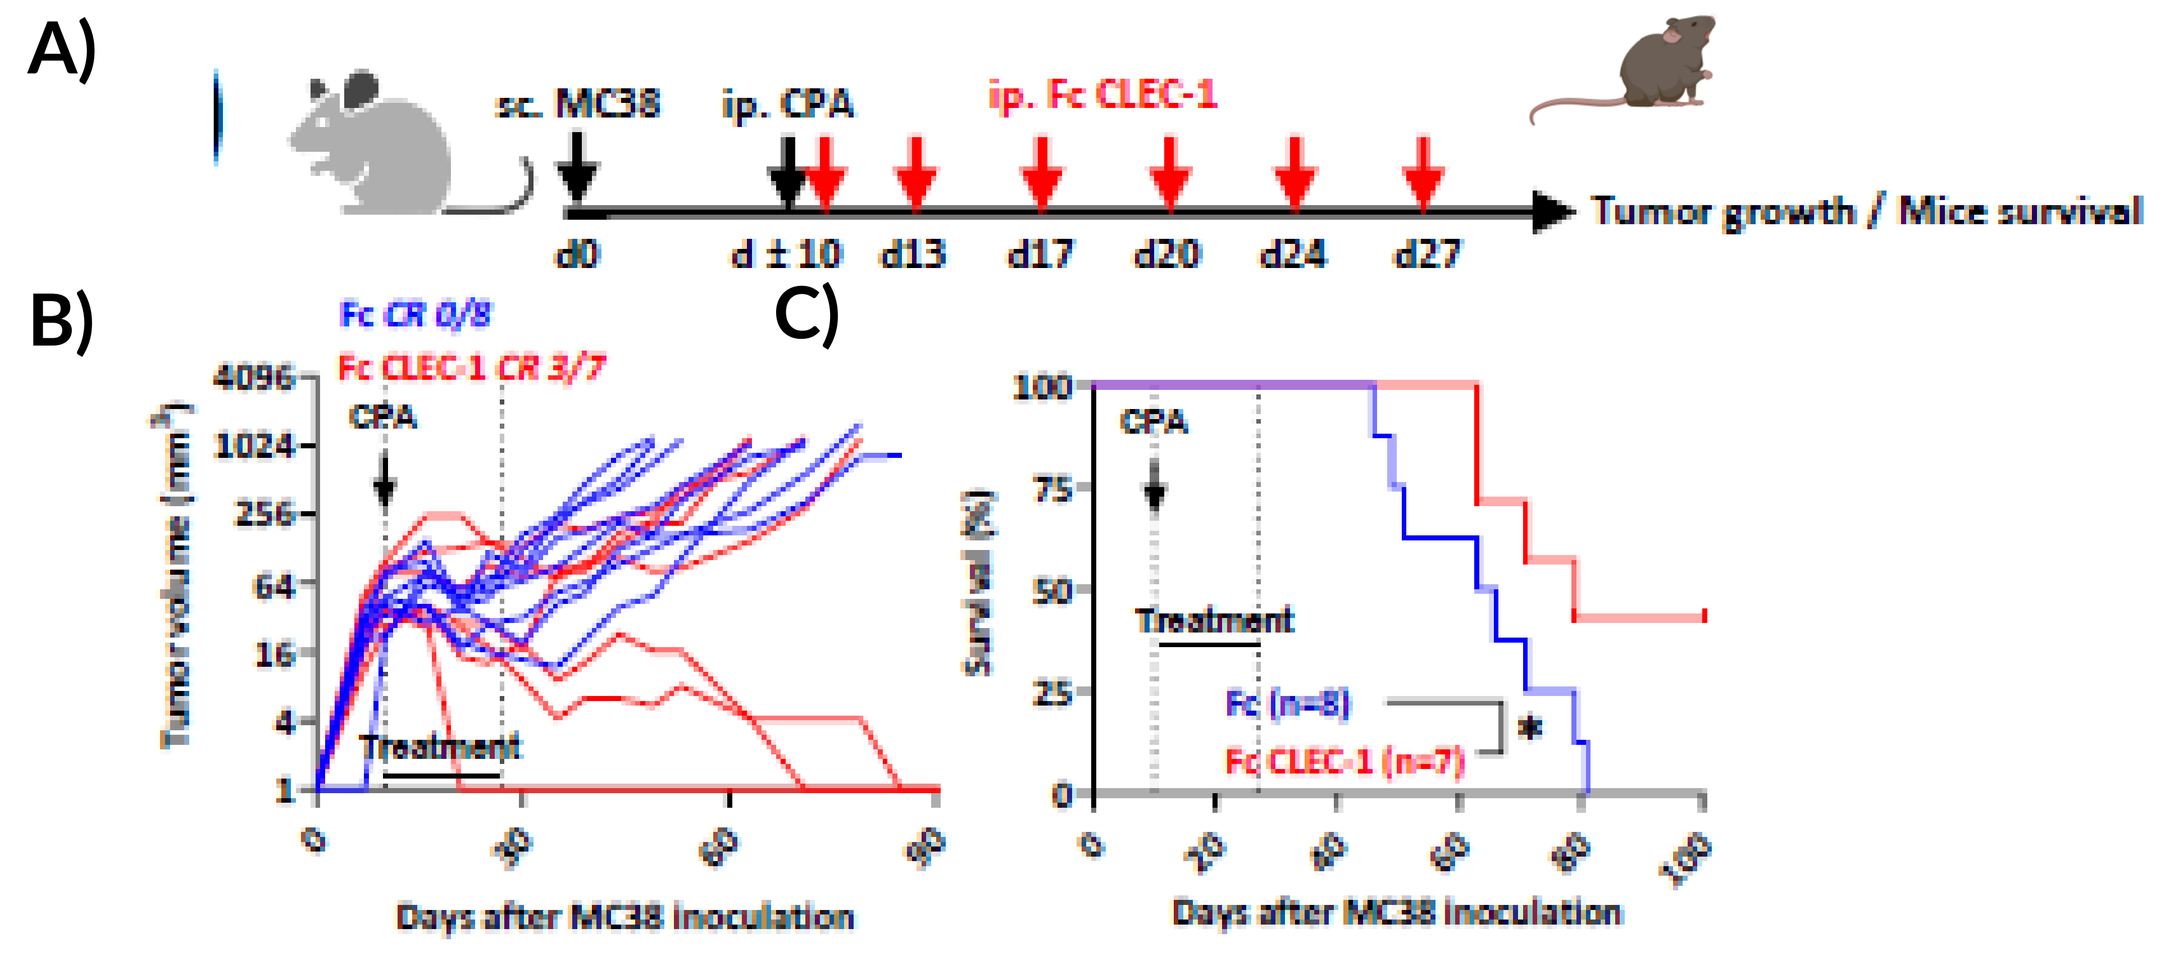
A)
sc. MC38
ip. CPA
ip. Fc CLEC-1
d0
d ±10
d13
d17
d20
d24
d27
Tumor growth / Mice survival
B)
Fc CR 0/8
Fc CLEC-1 CR 3/7
CPA
Treatment
Days after MC38 inoculation
C)
Fc (n=8)
Fc CLEC-1 (n=7)
Days after MC38 inoculation

The blockade of CLEC-1 signaling with mouse Fc CLEC-1 fusion protein prolonged survival of WT mice subcutaneously injected with 1x10⁶ MC38 colorectal cancer cells and treated with Chemotherapy (150mg/kg) cyclophosphamide administered intraperitoneally (i.p.) once at d12. Mouse Fc-CLEC-1 recombinant protein was intraperitoneally injected twice a week for three weeks from d4 at 5mg/kg (**A and B**).

6. Identification of TRIM21 as a novel endogenous CLEC-1 ligand

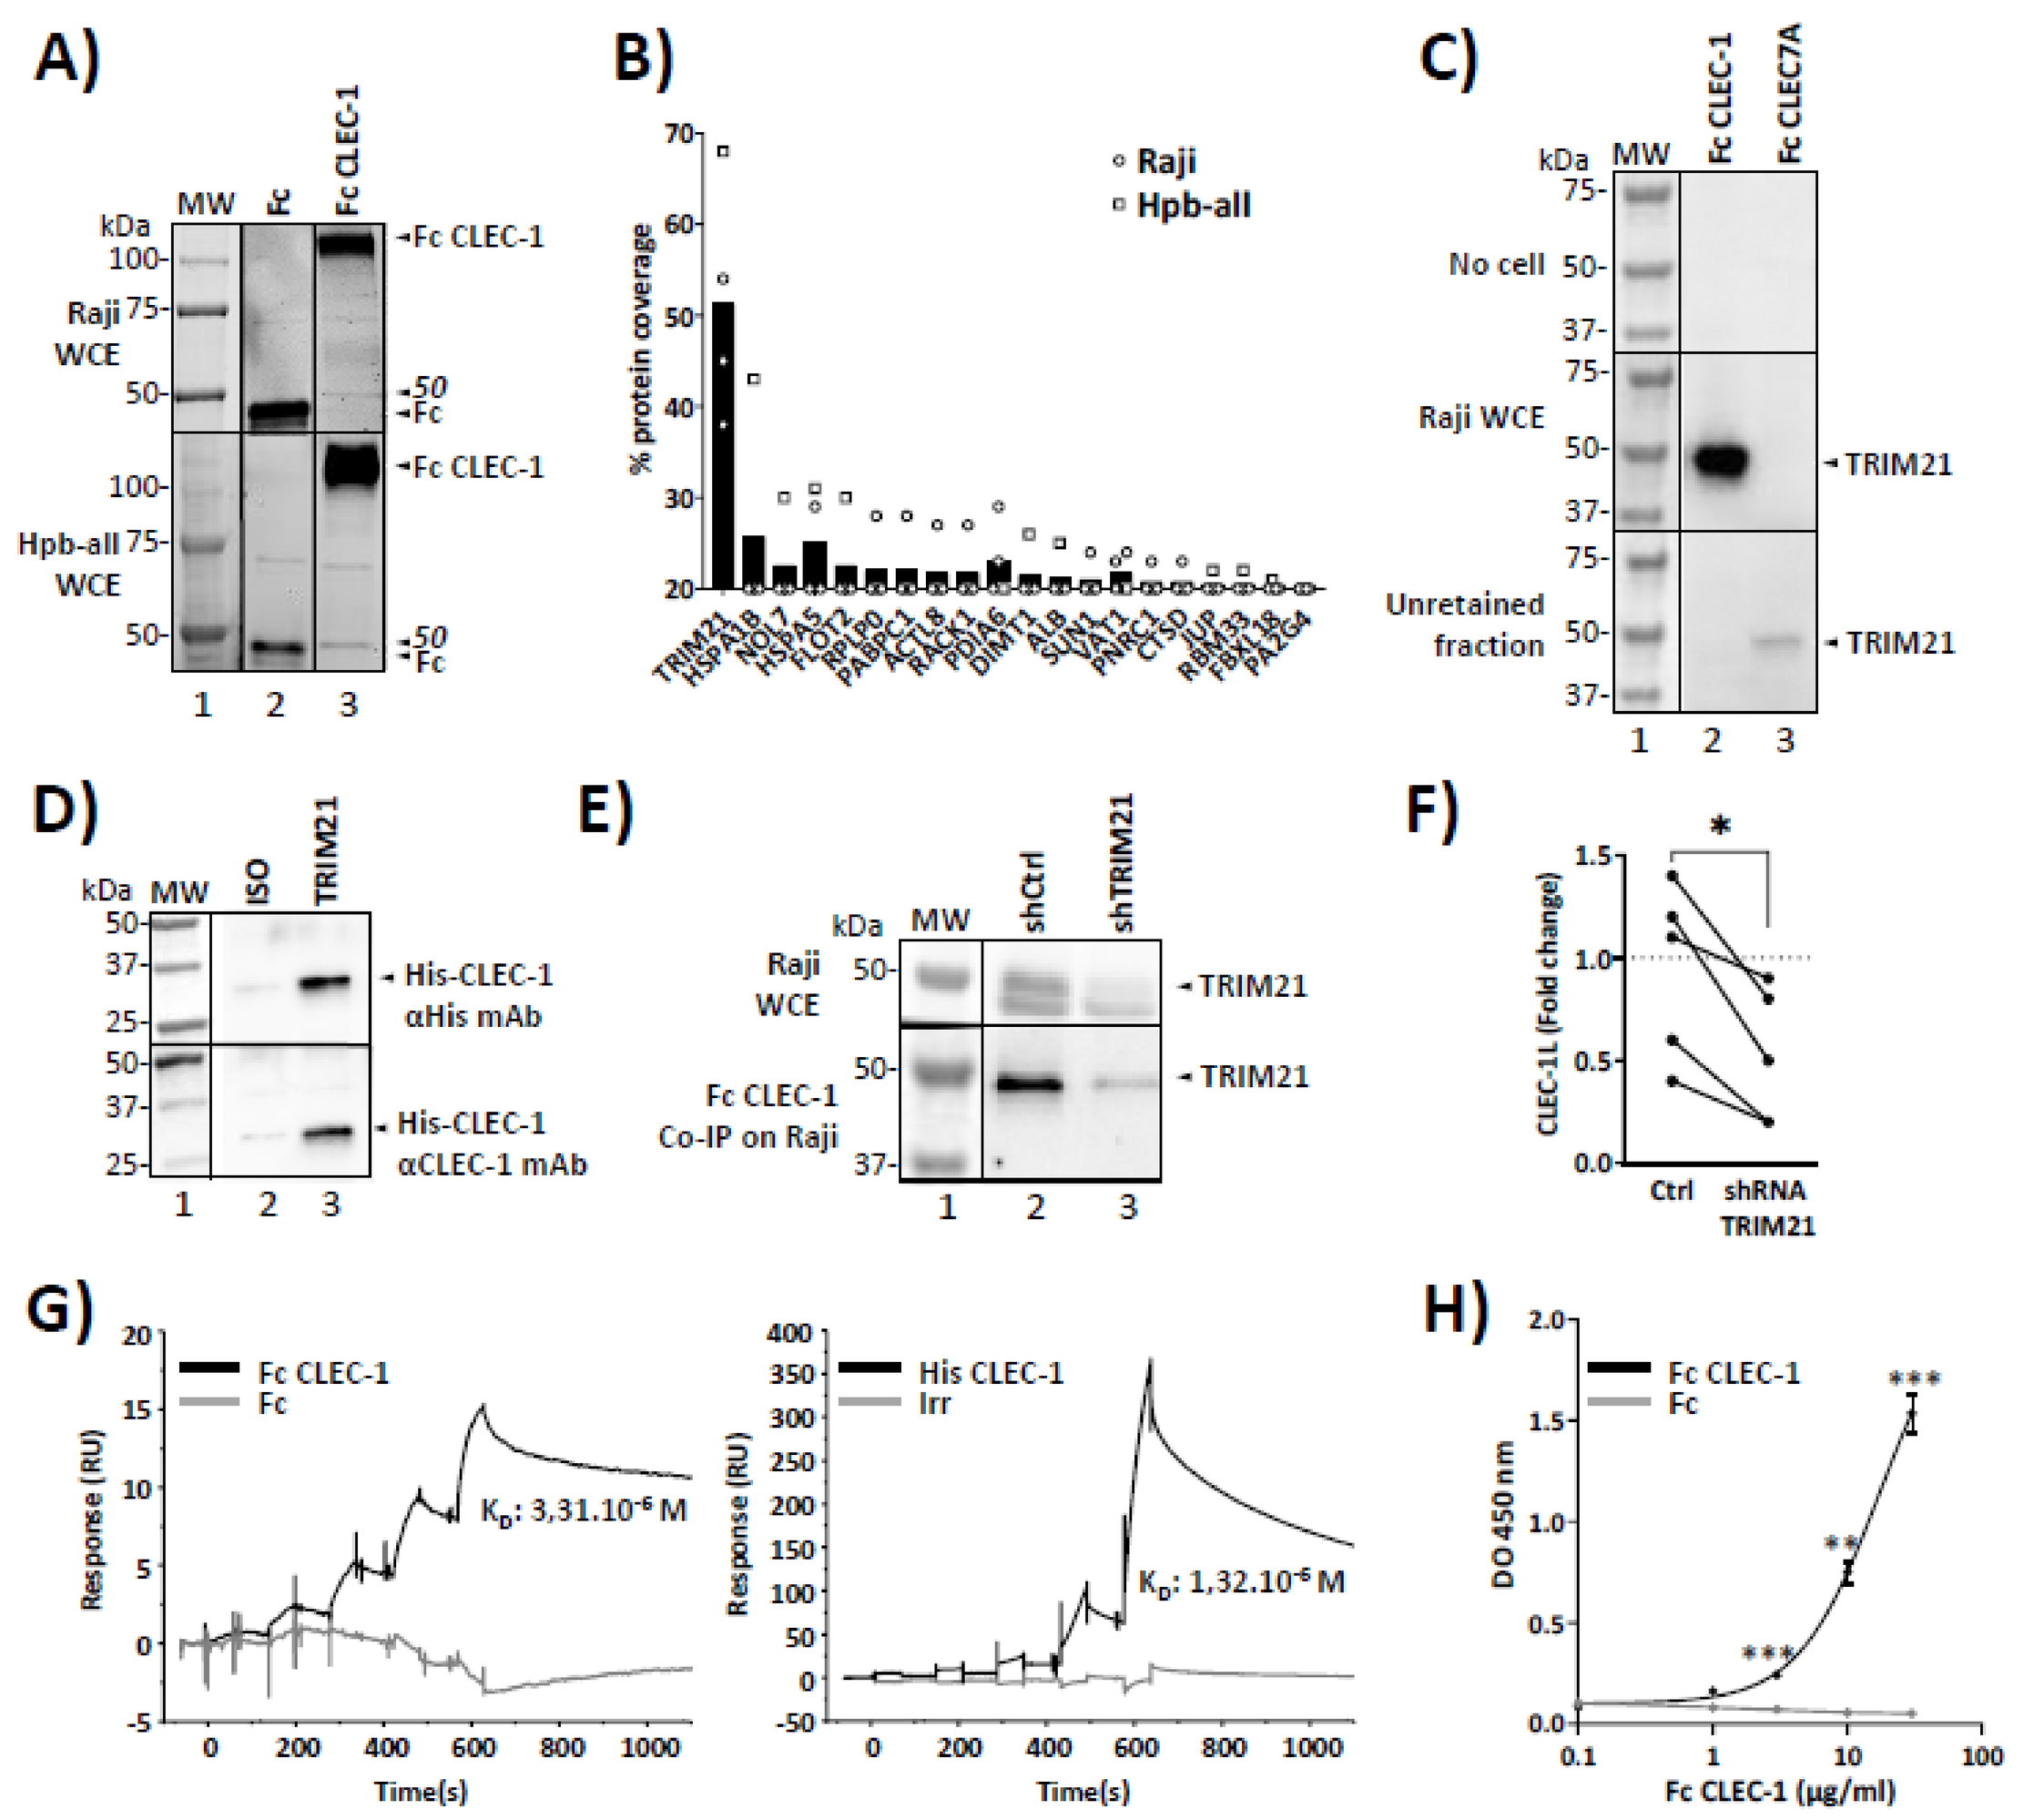
A)
B)
Raji
Hpb-all
% protein coverage
C)
No cell
Raji WCE
Unretained fraction
TRIM21
D)
His-CLEC-1
αHis mAb
His-CLEC-1
αCLEC-1 mAb
E)
Fc CLEC-1
Co-IP on Raji
F)
G)
Fc CLEC-1
Fc
His CLEC-1
Irr
Time(s)
H)
Fc CLEC-1 (µg/ml)

After several affinity purifications coupled, eluted proteins of human Fc CLEC-1 with protein extracts from necrotic human cells revealed one specific band around 50 kDa (**A**). LC-MS identification performed on this band produced a short list of 20 possible candidates which TRIM21 harboring the highest mean coverage (50%), and whose molecular weight of 52 kDa corresponds to the main band observed (**B**). Specificity of the interaction of TRIM21 with Fc CLEC-1 was confirmed by Western blot (**C**). We excluded the possibility of an interaction with the Fc fragment by use the His CLEC-1 recombinant protein (**D**). Interaction of TRIM21 with CLEC-1 was also confirmed by knocking down TRIM21 via shRNA in necrotic cells, leading to a subsequent lower enrichment of TRIM21 following the co-IP of Fc CLEC-1 and to a decrease in the Fc CLEC-1 binding by flow cytometry (**E and F**). To finish, we validated the recognition of TRIM21 by Biacore and enzyme-linked immunosorbent assay (**G and H**).

7. TRIM21 is induced at the cell surface of necrotic cells

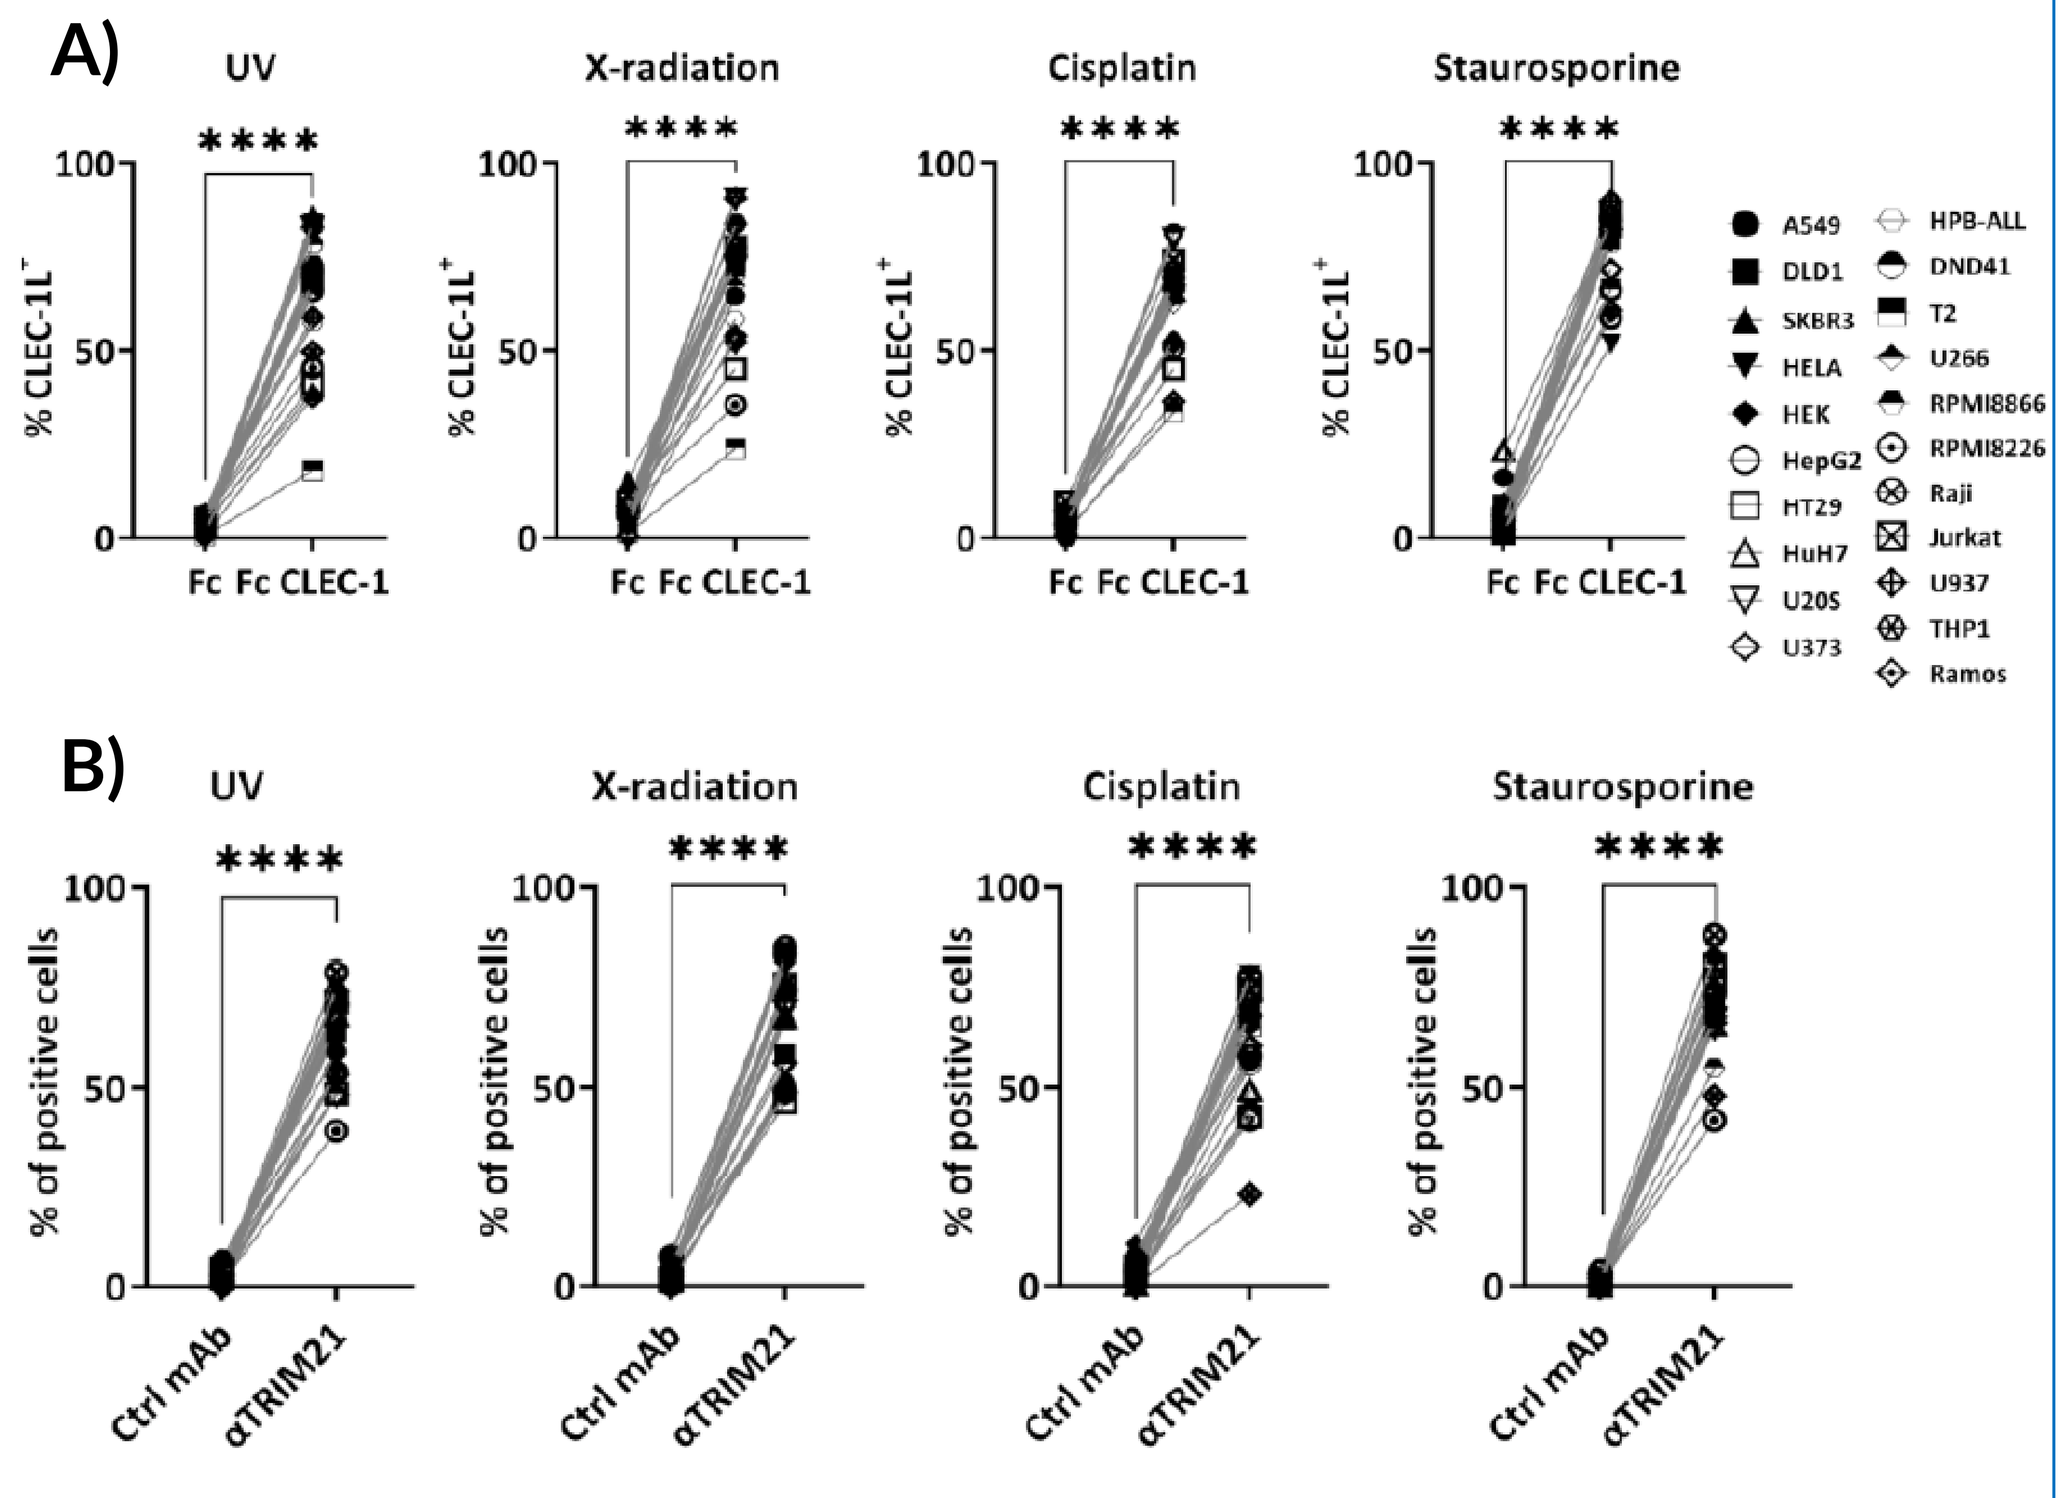
A)
UV
X-radiation
Cisplatin
Staurosporine
B)
% of positive cells
Ctrl mAb
αTRIM21

Human tumor cell lines treated with 150mJ/cm² UV radiation, 10Gy X–radiation, 20µM cisplatin or 1µM of staurosporin for 18h in culture expressed strongly the CLEC-1 ligands (**A**) and TRIM21 (**B**).
(NSCLC : non-small cells lung cancer A549; HCC : hepatocellular carcinoma Huh7, HepG2; OS : osteosarcoma U2OS; GB : glioblastoma U373; CRC : colorectal cancer HT-29, DLD1; triple negative breast cancer SK-BR3; ovarian cancer HeLa; human embryonic kidney HEK; T-ALL : T acute lymphoblastic leukemia HPBALL, DND41, Jurkat ; B-ALL Ramos, RPMI8866; myeloma U266, RPMI8226, lymphoma T2; acute myeloid leukemia THP-1, U937)

8. TRIM21 is overexpressed in cancer

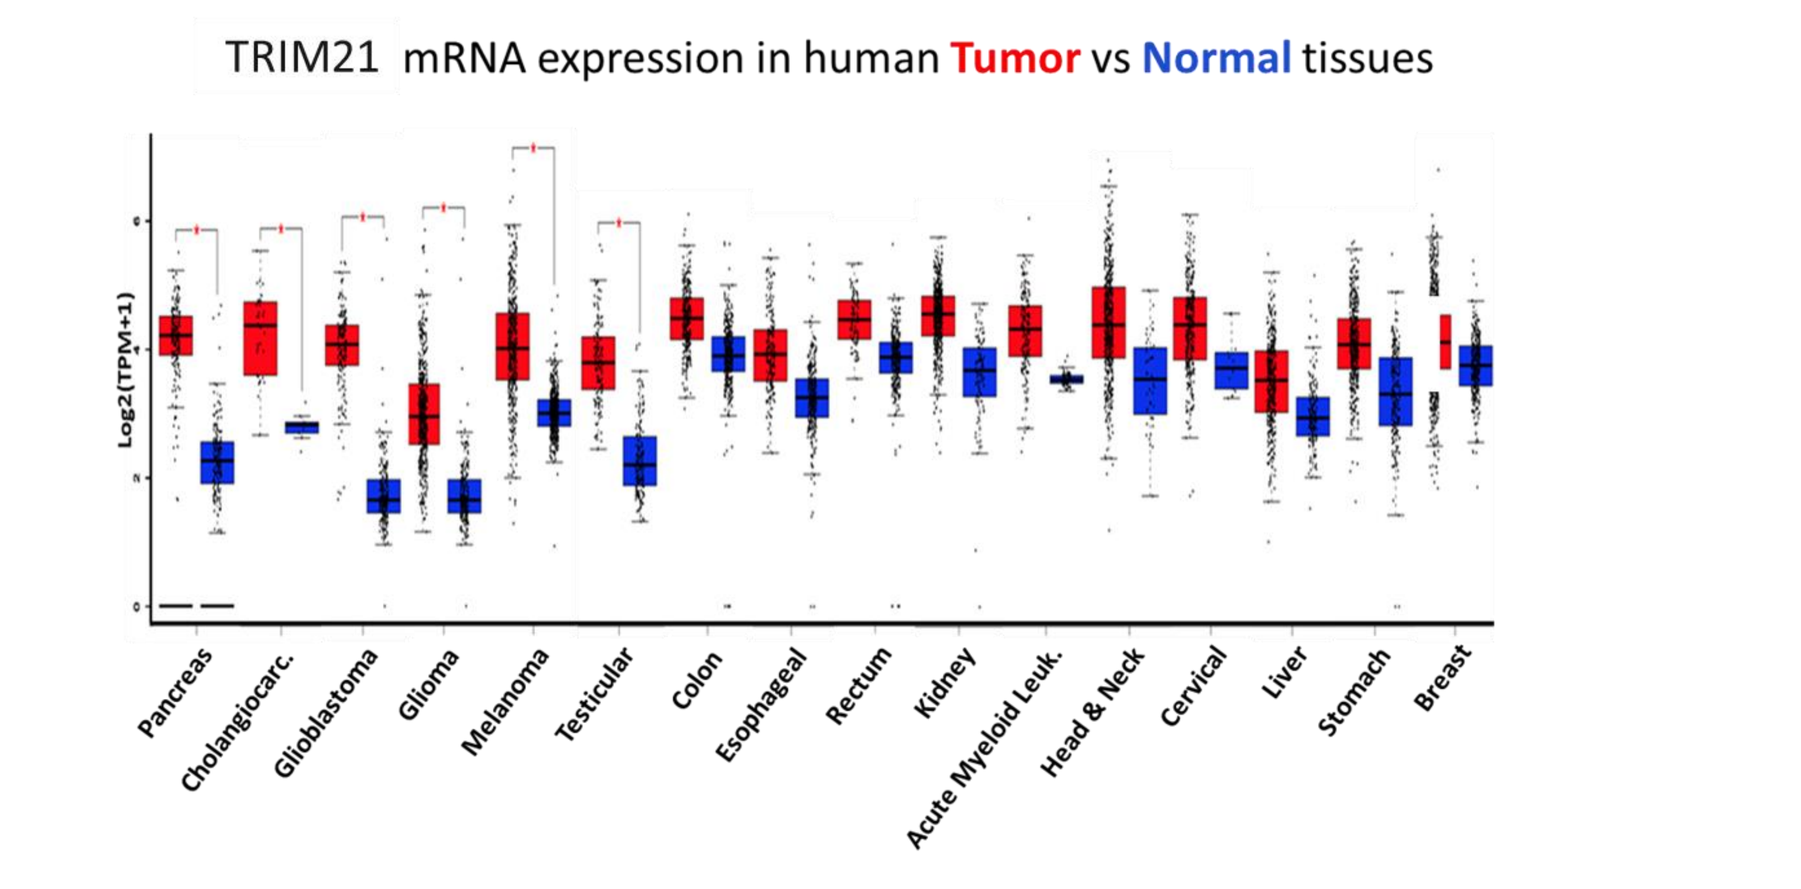
TRIM21 mRNA expression in human Tumor vs Normal tissues
Pancreas
Cholangiocarc.
Glioblastoma
Glioma
Melanoma
Testicular
Colon
Esophageal
Rectum
Kidney
Acute Myeloid Leuk.
Head & Neck
Cervical
Liver
Stomach
Breast

Analysis of datasets from The Cancer Genome Atlas (TCGA) with GEPIA tool indicated that human cancers overexpress TRIM21 compared to healthy tissues.

9. TRIM21 associates with worse survival

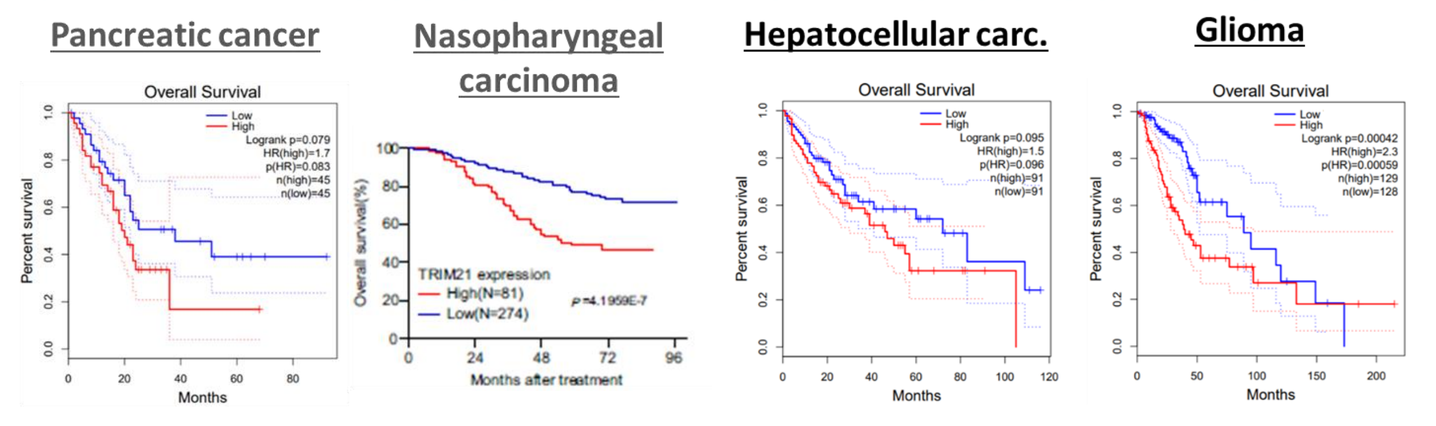
Pancreatic cancer
Nasopharyngeal carcinoma
Hepatocellular carc.
Glioma

In several cancers, such as Pancreatic cancer, Nasopharyngeal carcinoma, Hepatocellular carcinoma and glioma, TRIM21 expression associates with decreased overall survival.

CONCLUSIONS & OUTLOOK

- CLEC-1 is expressed by myeloid cells and by tumor-associated macrophages in human and mouse tumors.
- CLEC-1 expression inhibits anti-tumoral response and CD8+ T-cell cross-priming by dendritic cells.
- CLEC-1 recognizes an endogenous ligand that is exposed by necrotic cells following stress (UV, X-ray, chemotherapies).
- CLEC-1/CLEC1-L interaction on necrotic cells is functional and promotes anti-tumor response *in vivo* in combination with chemotherapy treatment.
- TRIM21, a ubiquitously expressed FcR and E3 ubiquitin ligase, overexpressed in tumors and associating with worse overall survival is a novel endogenous ligand of CLEC-1.
- Targeting of the axis CLEC-1 /TRIM21 with antagonist anti-CLEC-1 or anti-TRIM21 antibodies are being evaluated to characterize the involvement of the newly identified CLEC-1 interaction with TRIM21 in the regulation of CLEC-1's function as an inhibitory myeloid checkpoint.
- Altogether, the CLEC-1/TRIM21 axis is of high interest in immune desert and to fight Radiotherapy and Chemotherapy resistance.

References:

1. Drouin et al. Science Advances 2022

Contact information : nicolas.poirier@ose-immuno.com ; irene.baccelli@ose-immuno.com