AACR 2023

Anti-PD-1/IL-7v bispecific antibody (BICKI®IL7v) promotes TCF1+ stem like T cells expansion and long-lasting in vivo efficacy

Aurore Morello, Margaux Seité, Justine Durand, Isabelle Girault , Géraldine Teppaz, , Virginie Thepenier, Cécile Batty, Sabrina Pengam, Emmanuelle Wilhelm, Ariane Desselle, Kevin Biteau, Julien Taurelle, Caroline Mary, Nicolas Poirier

OSE IMMUNO THERAPEUTICS

aurore.morello@ose-immuno.com

1/ BICKI®IL7 *Optimized anti PD-1/IL7v immunocytokine design for selective cis-potentiation*

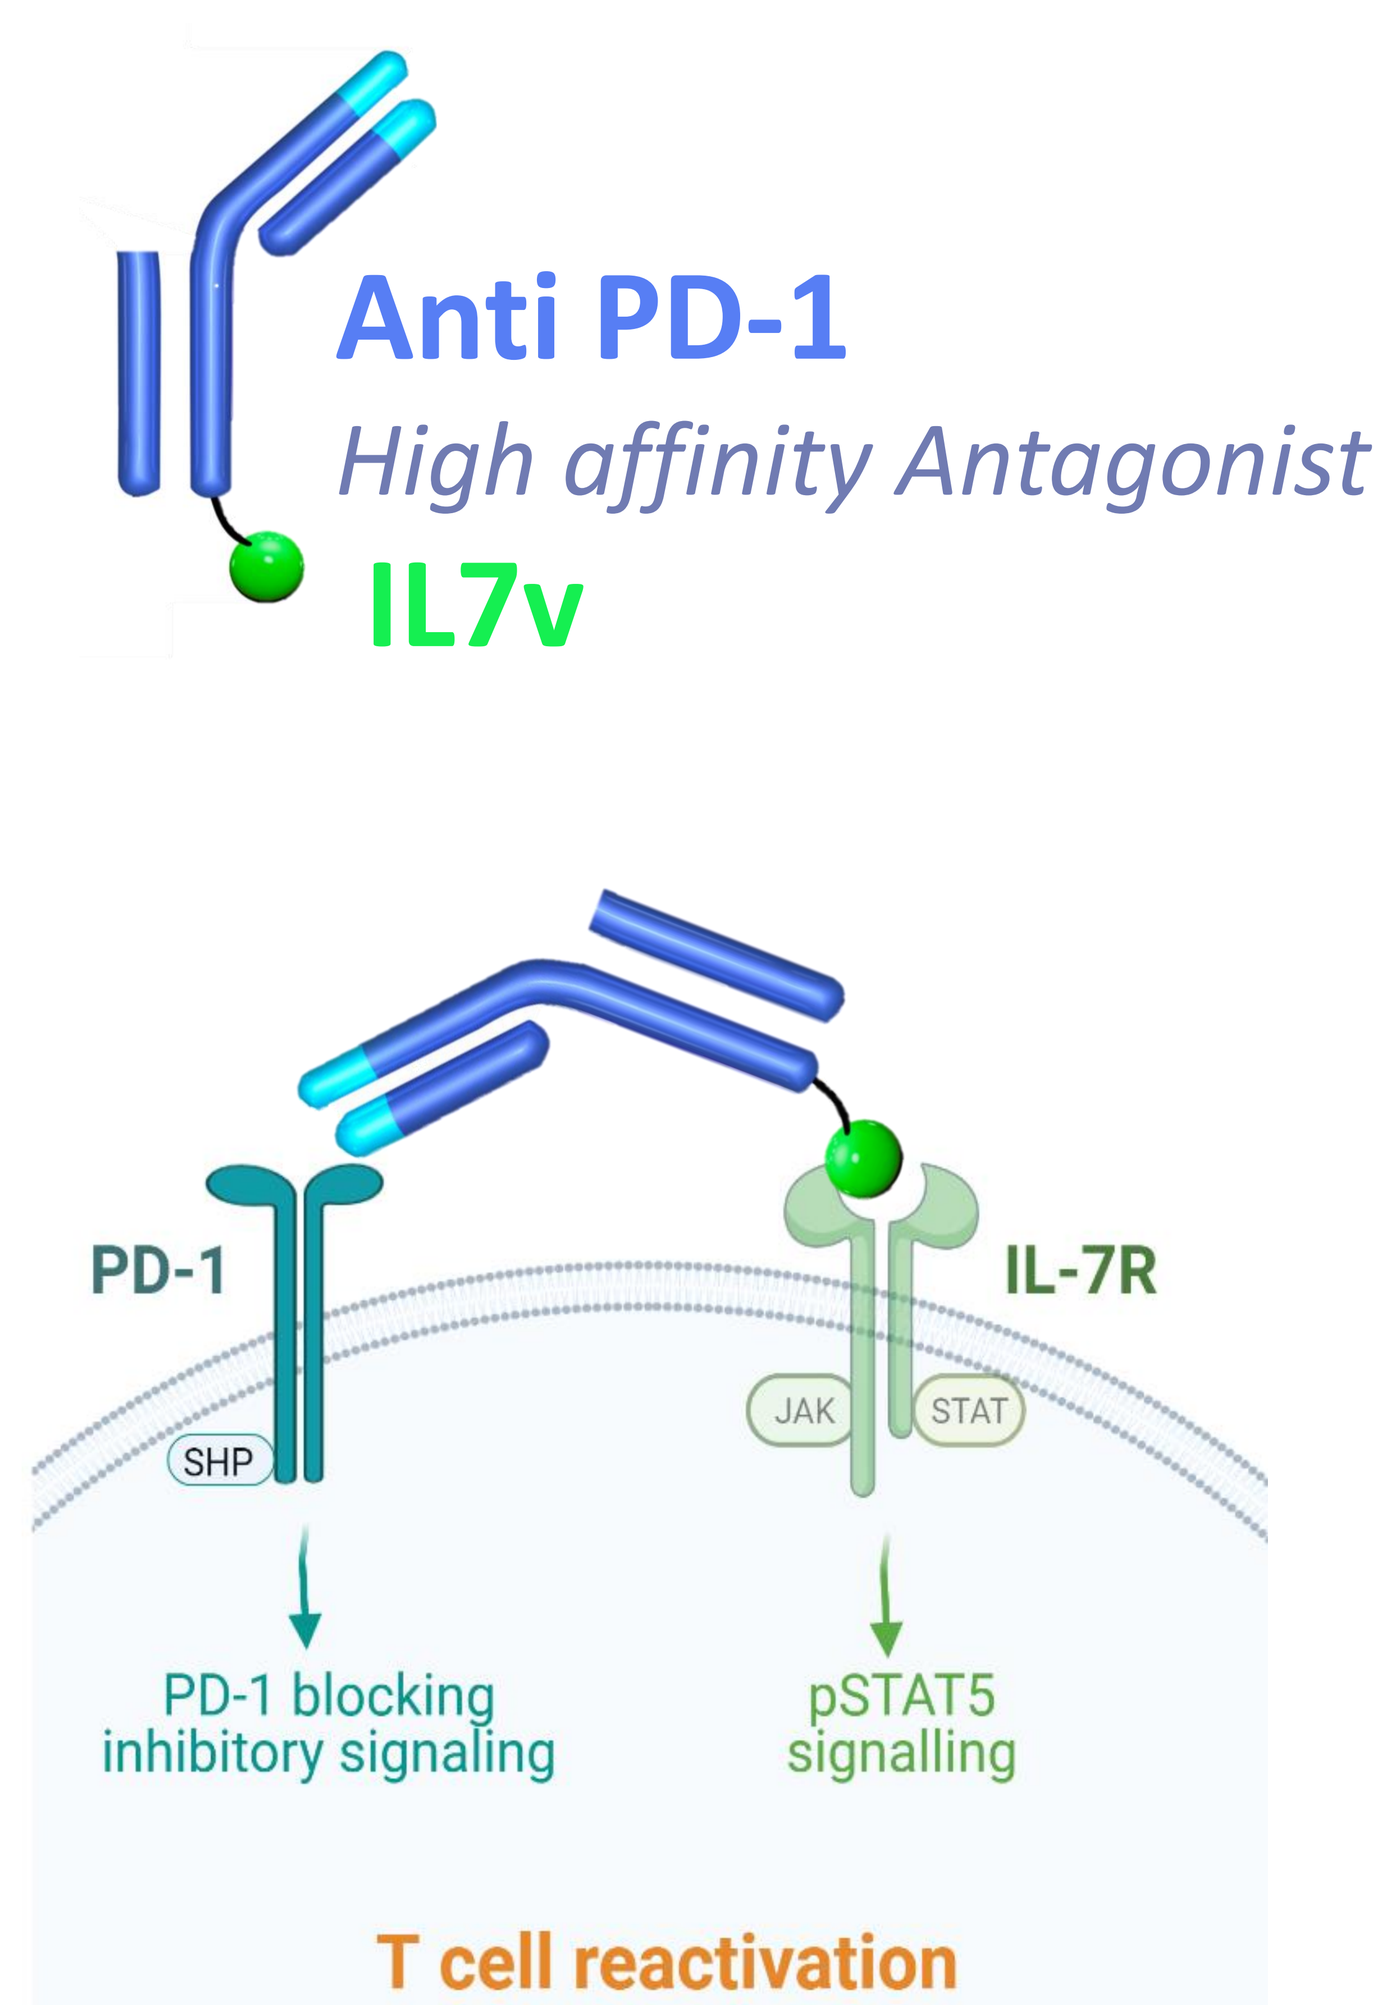

Anti PD-1
High affinity Antagonist
IL7v
PD-1
IL-7R
SHP
JAK
STAT
PD-1 blocking inhibitory signaling
pSTAT5 signalling
T cell reactivation

Enhanced in vivo drug exposure

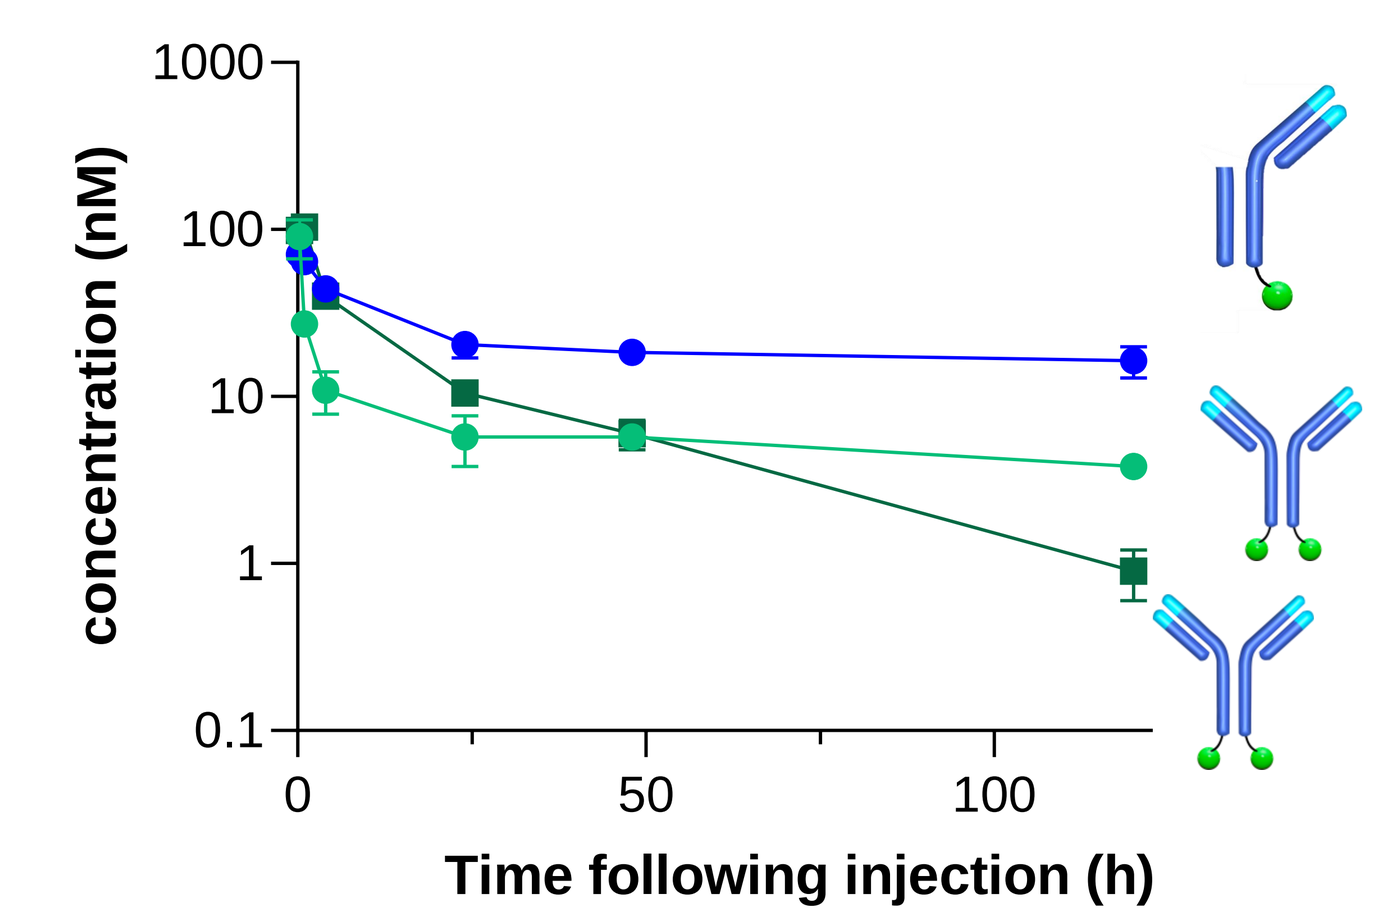

concentration (nM)
1000
100
10
1
0.1
0
50
100
Time following injection (h)

Cis activity

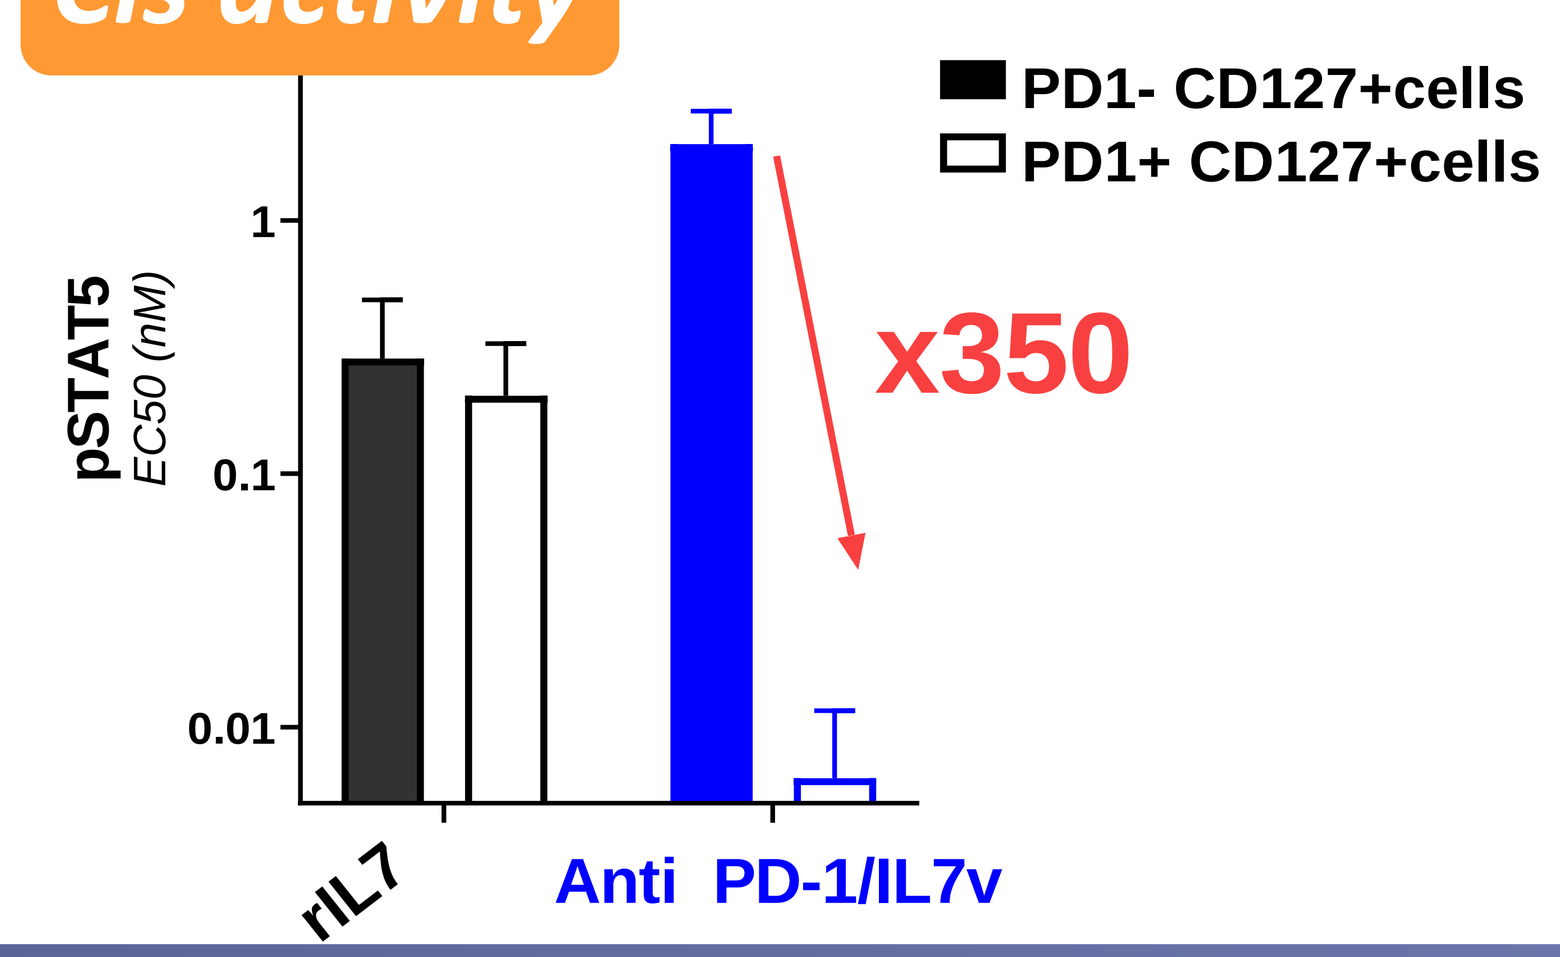

PD1- CD127+cells
PD1+ CD127+cells
pSTAT5
EC50 (nM)
1
0.1
0.01
x350
rIL7
Anti PD-1/IL7v

3/ BICKI®IL7v revives TILs & promotes long-term proliferation stem-like TCF1+ CD8 T cells

Enhanced IFNg secretion, proliferation & survival of human TILs

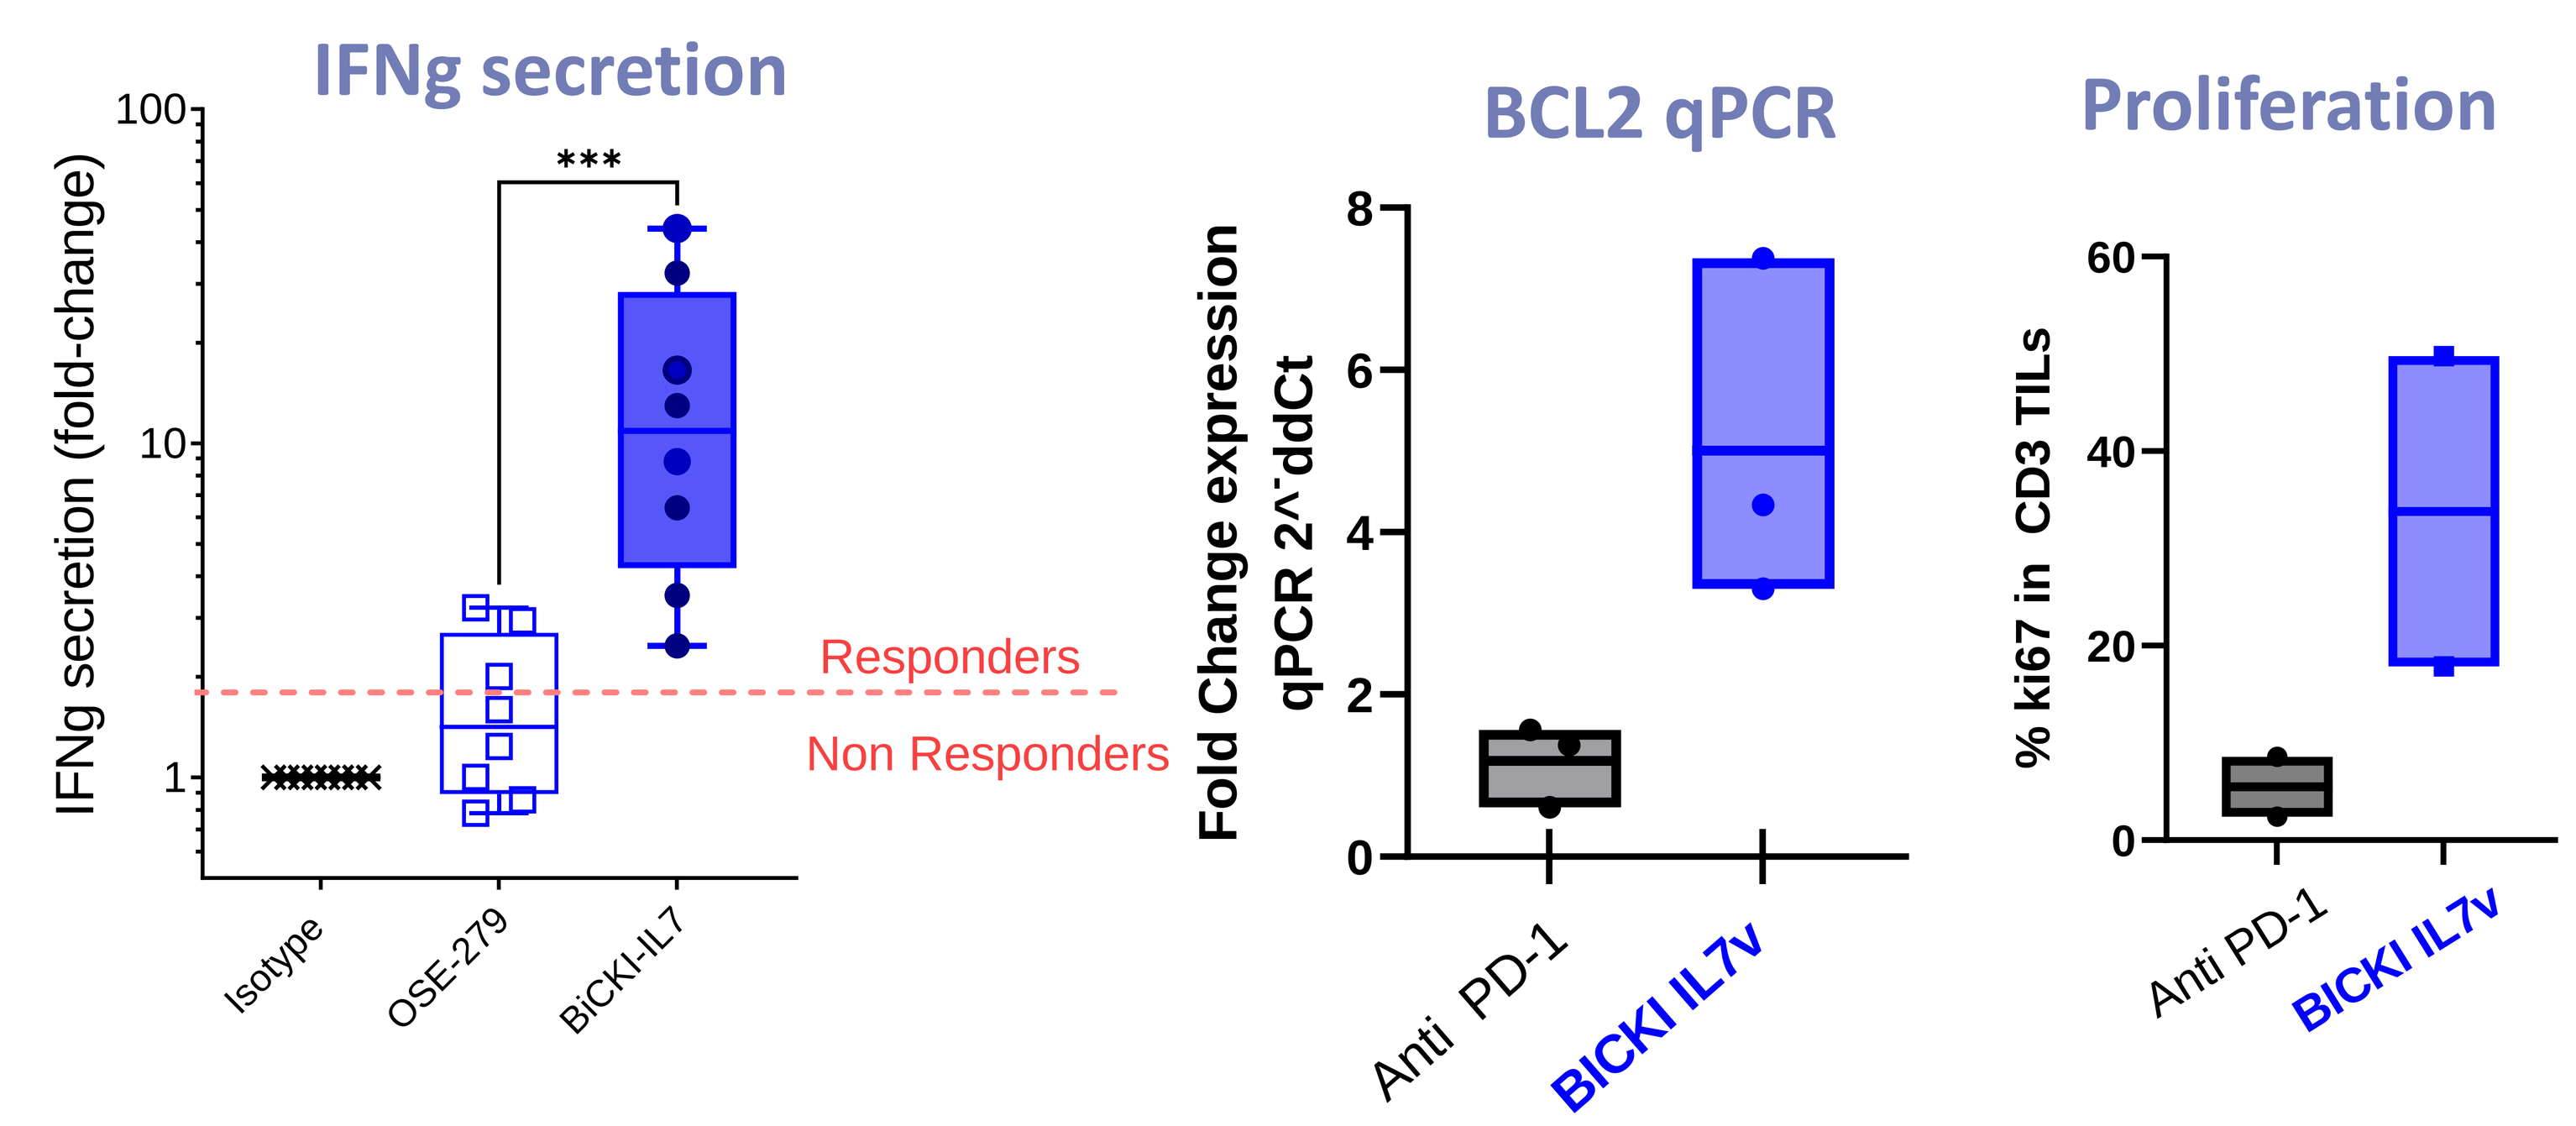

IFNg secretion
IFNg secretion (fold-change)
100
10
1

Responders
Non Responders
Isotype
OSE-279
BiCKI-IL7
BCL2 qPCR
Fold Change expression qPCR 2^ddCt
8
6
4
2
0
Anti PD-1
BICKI IL7v
Proliferation
% ki67 in CD3 TILs
60
40
20
0
Anti PD-1
BICKI IL7v

Proliferation Chronically stimulated T cells

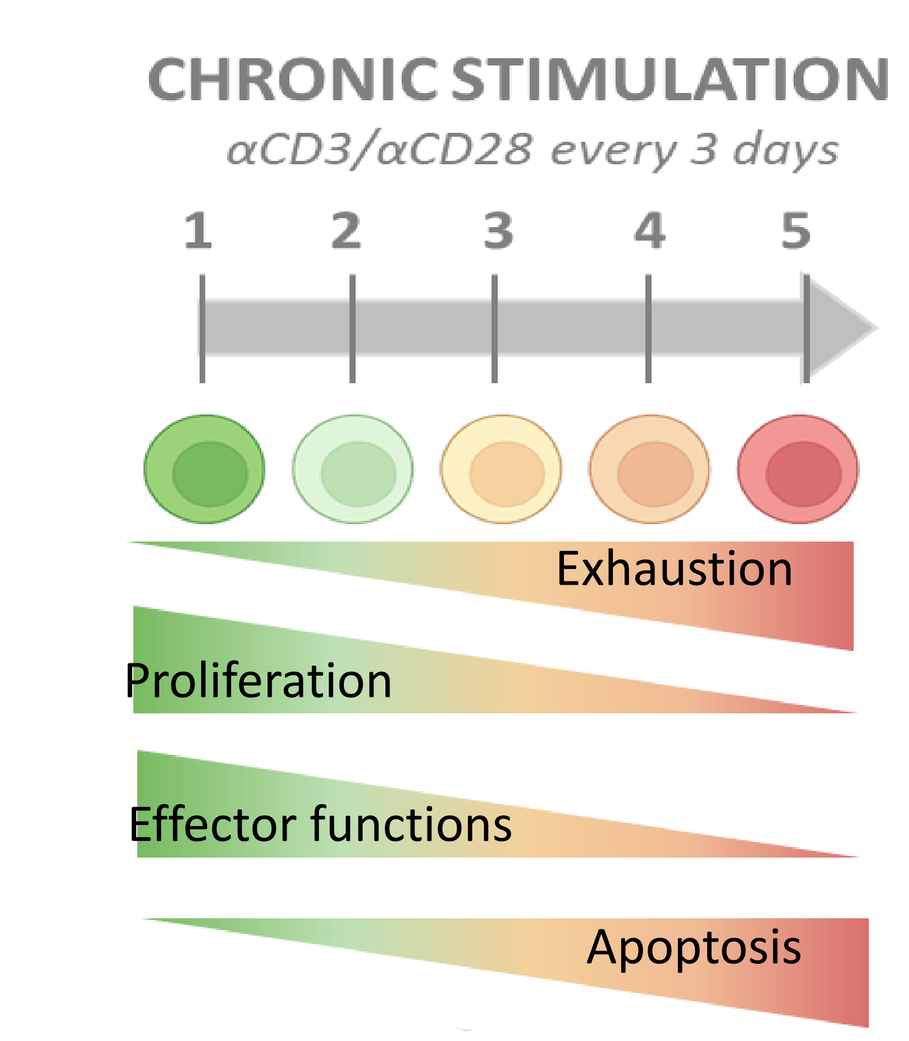

CHRONIC STIMULATION
αCD3/αCD28 every 3 days
1
2
3
4
5
Exhaustion
Proliferation
Effector functions
Apoptosis

TCF-1+ Ki67+

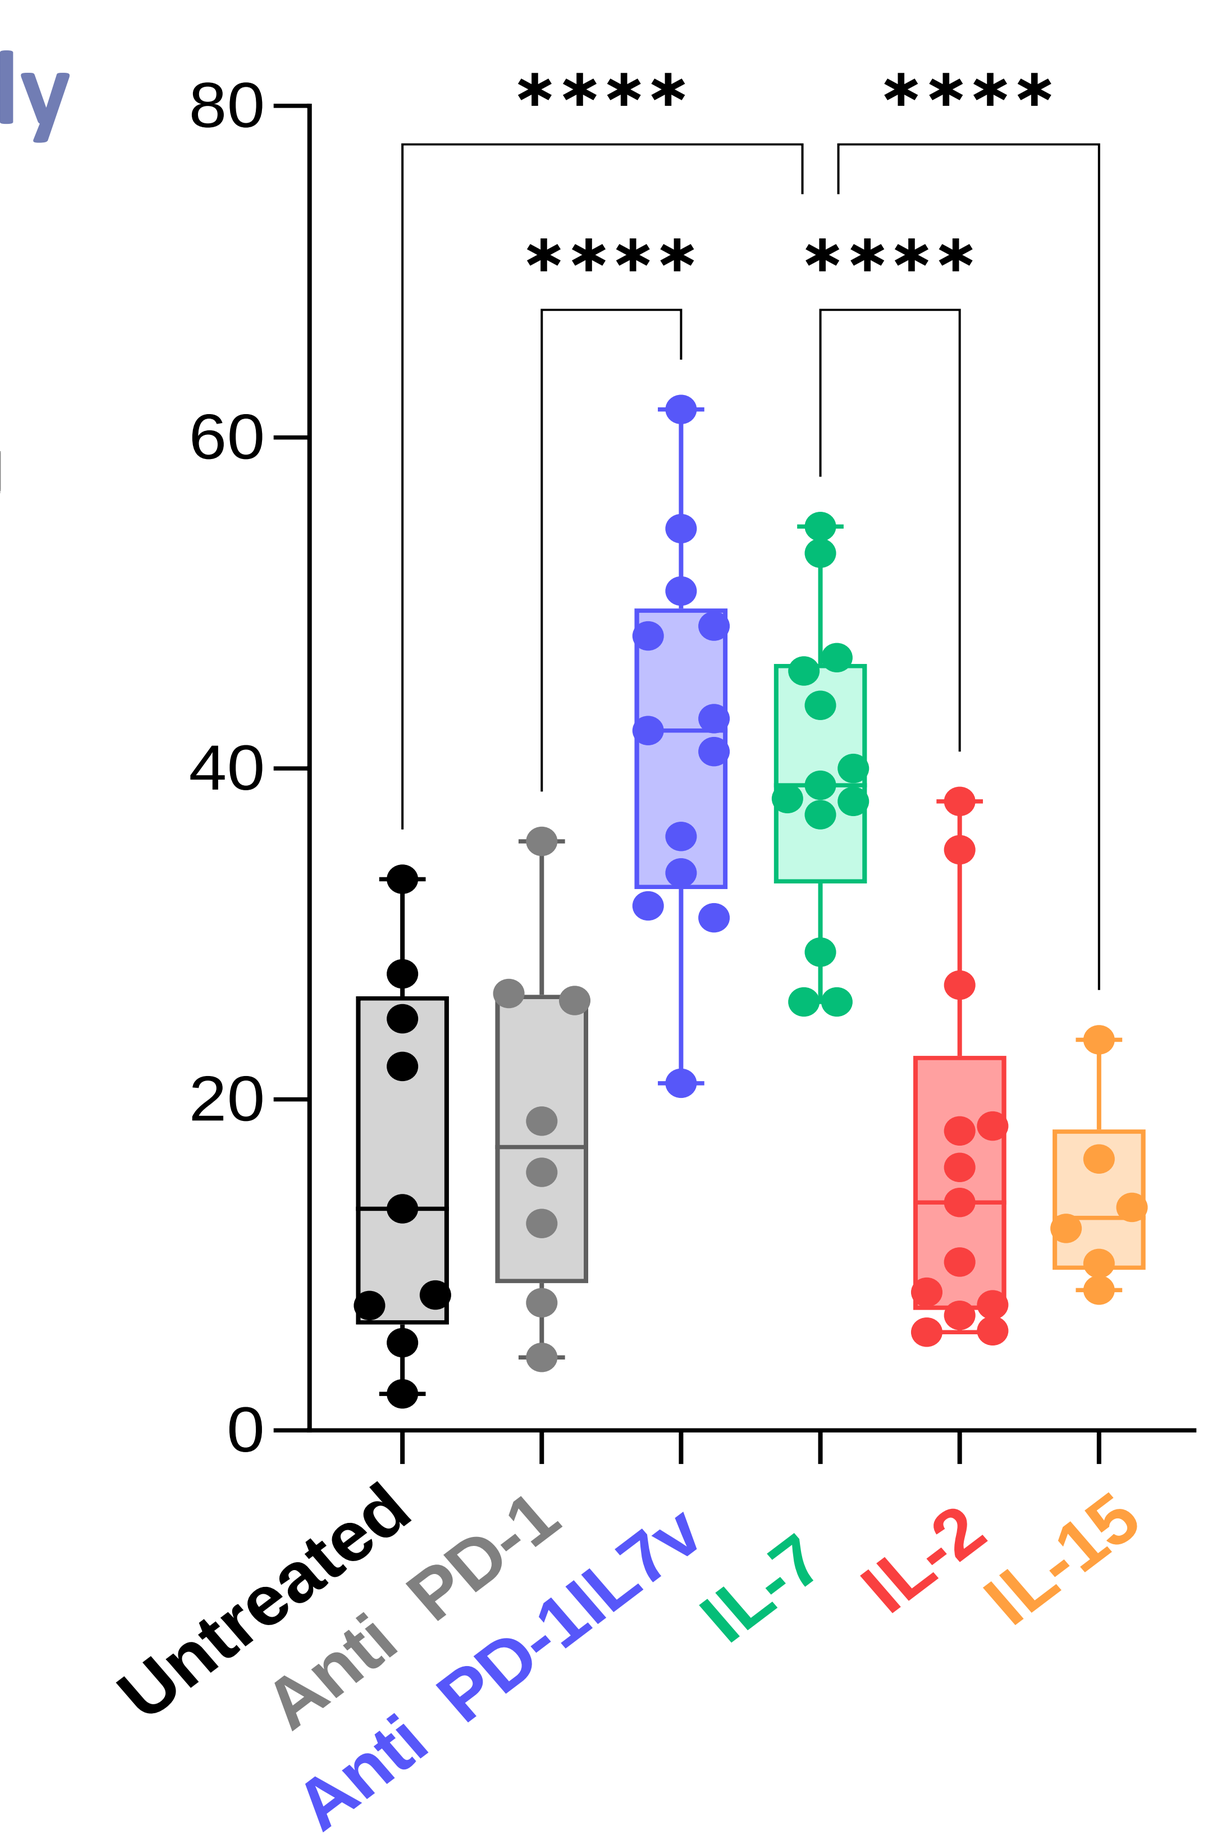

80
60
40
20
0

Untreated
Anti PD-1
Anti PD-1IL7v
IL-7
IL-2
IL-15

TCF-1- Ki67+

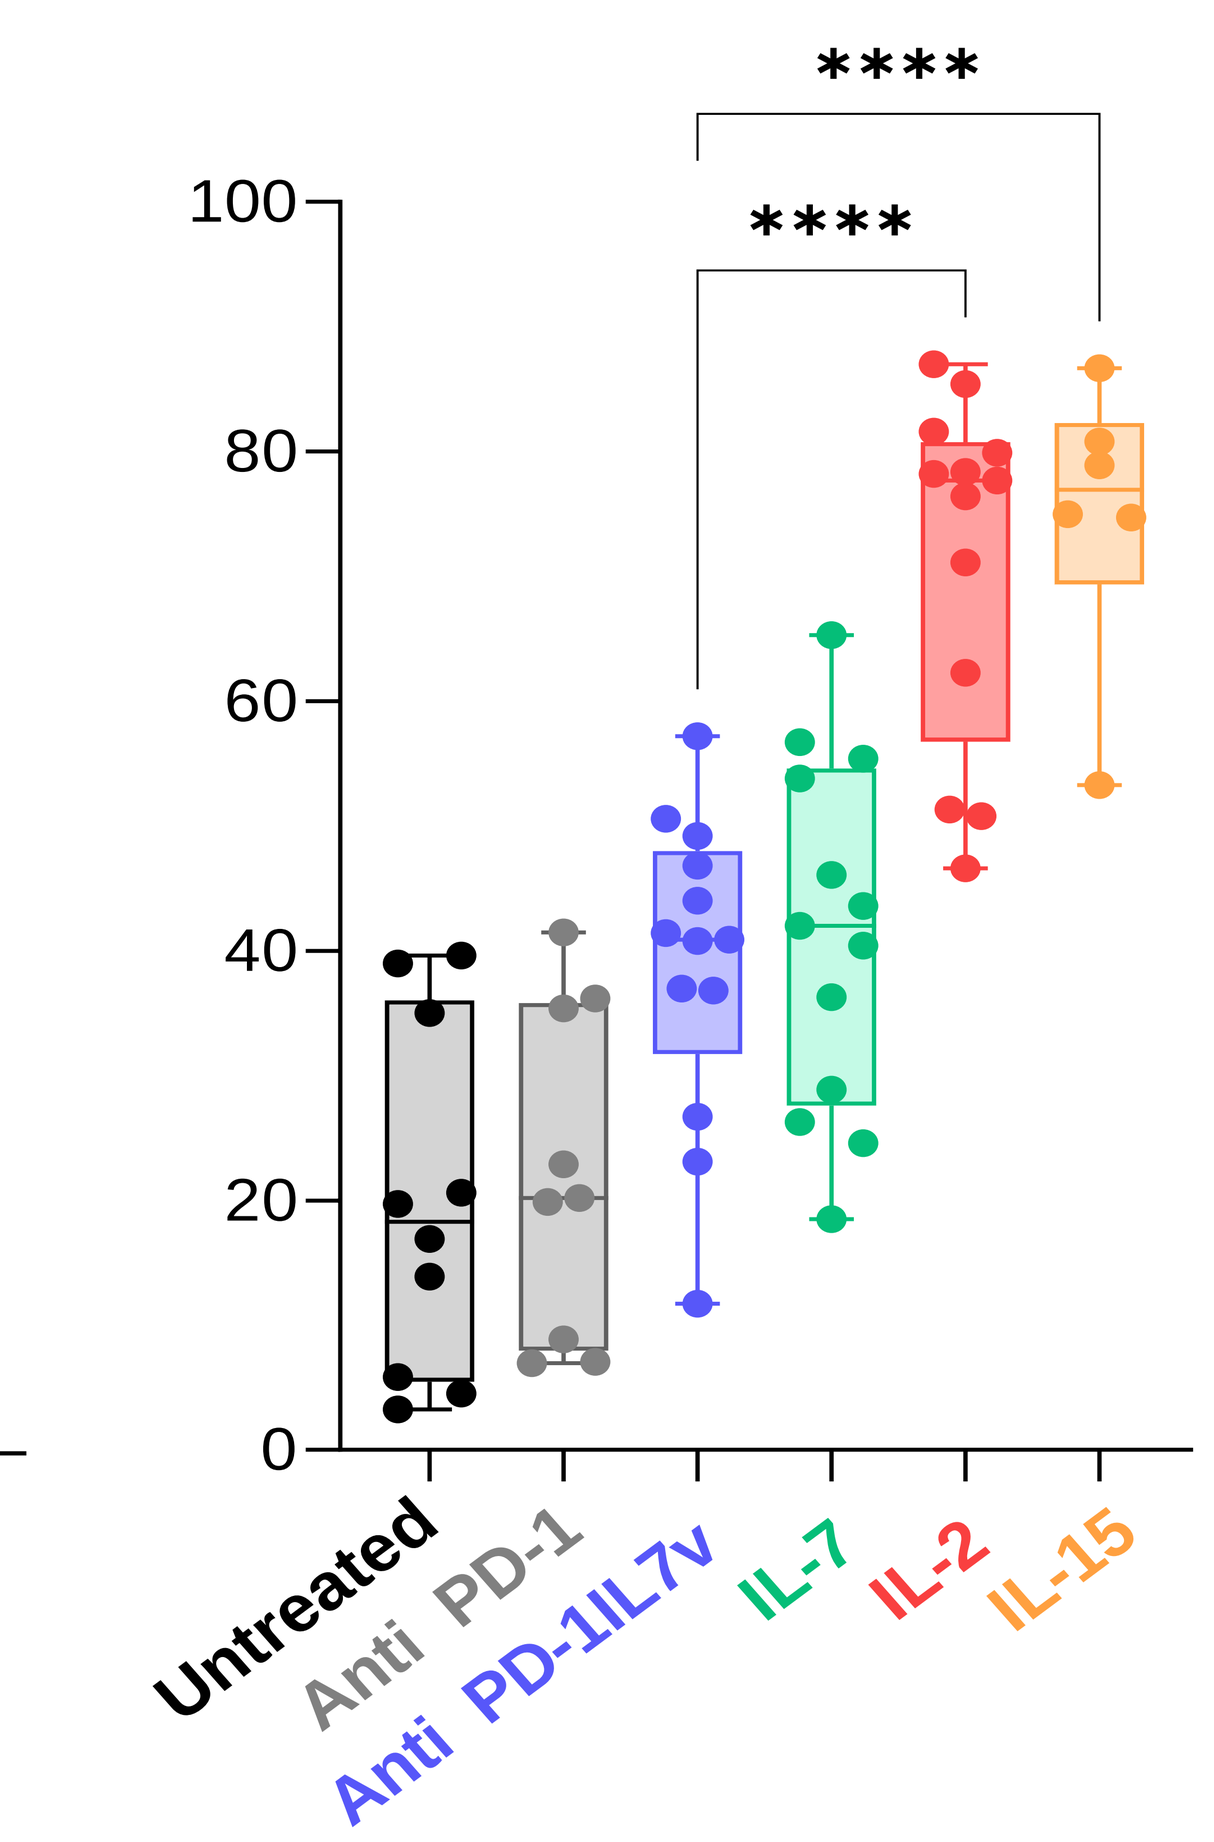

100
80
60
40
20
0

Untreated
Anti PD-1
Anti PD-1IL7v
IL-7
IL-2
IL-15

2/ Rationale for co-targeting PD-1 and IL-7 in TILs: *IL7R expression correlates with better ICI response/survival and T-cell stemness*

scRNAseq

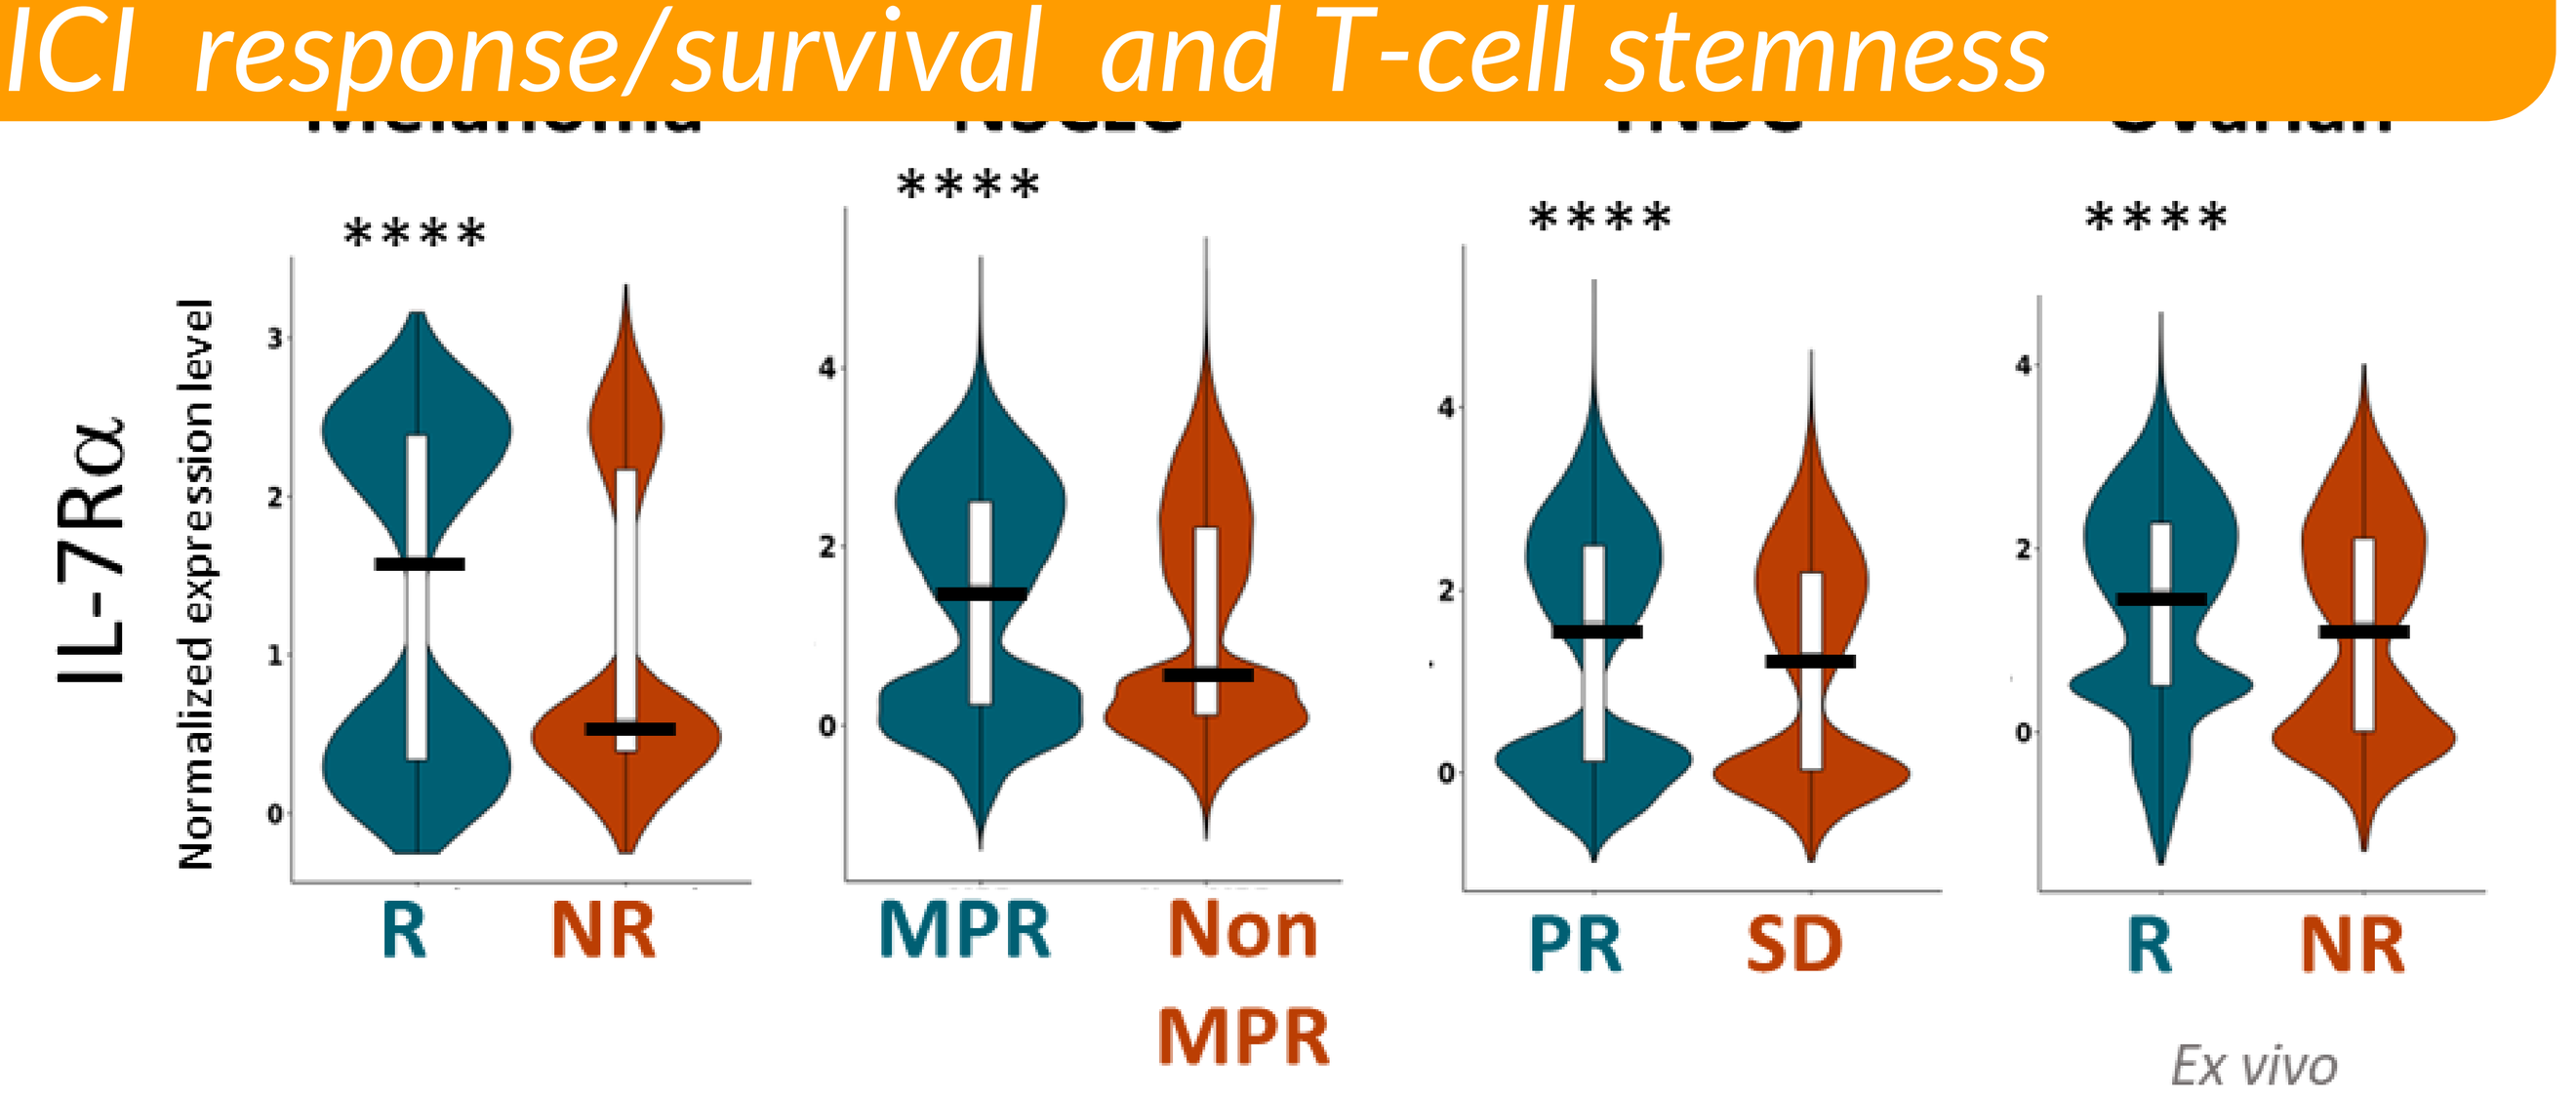

IL-7Rα
Normalized expression level

R
NR
MPR
Non MPR
PR
SD
Ex vivo

Anti PD(L)1 responders

Anti PD(L)1 Non responders

Melanoma scRNAseq

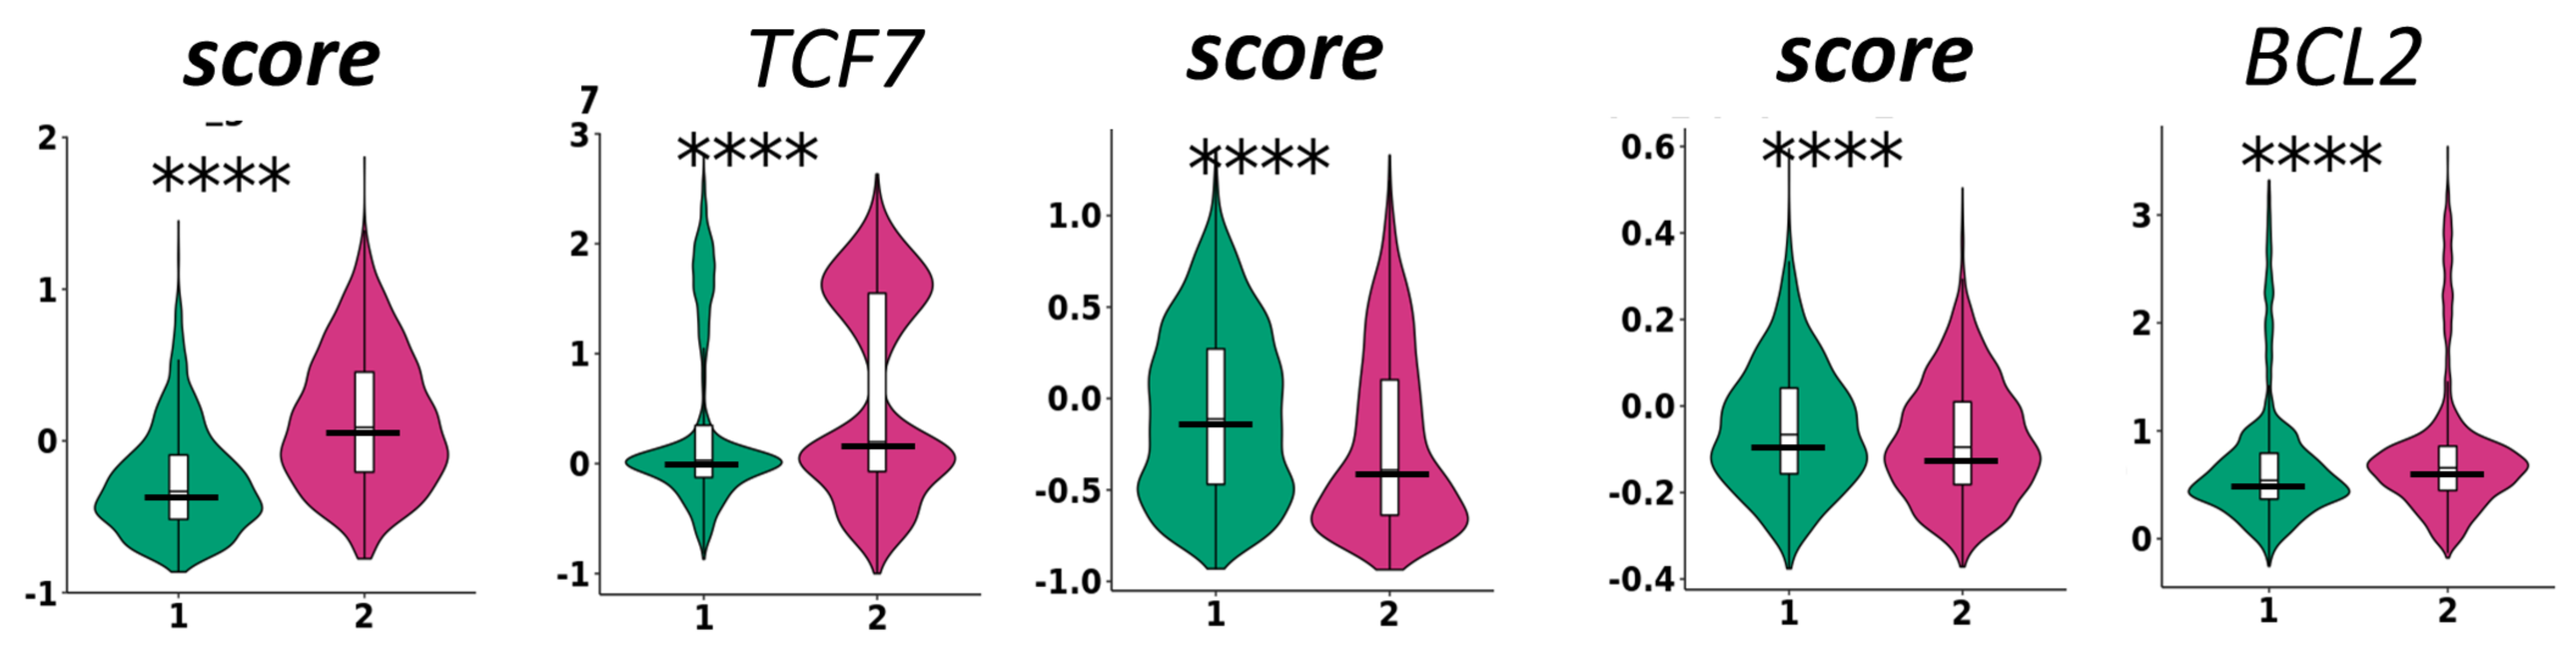

Stemness score
TCF7
Exhaustion score
Pro-apoptosis score
BCL2

1
2

(1) IL7R expressionLow

(2)IL7R expression High

Melanoma *RNAseq*

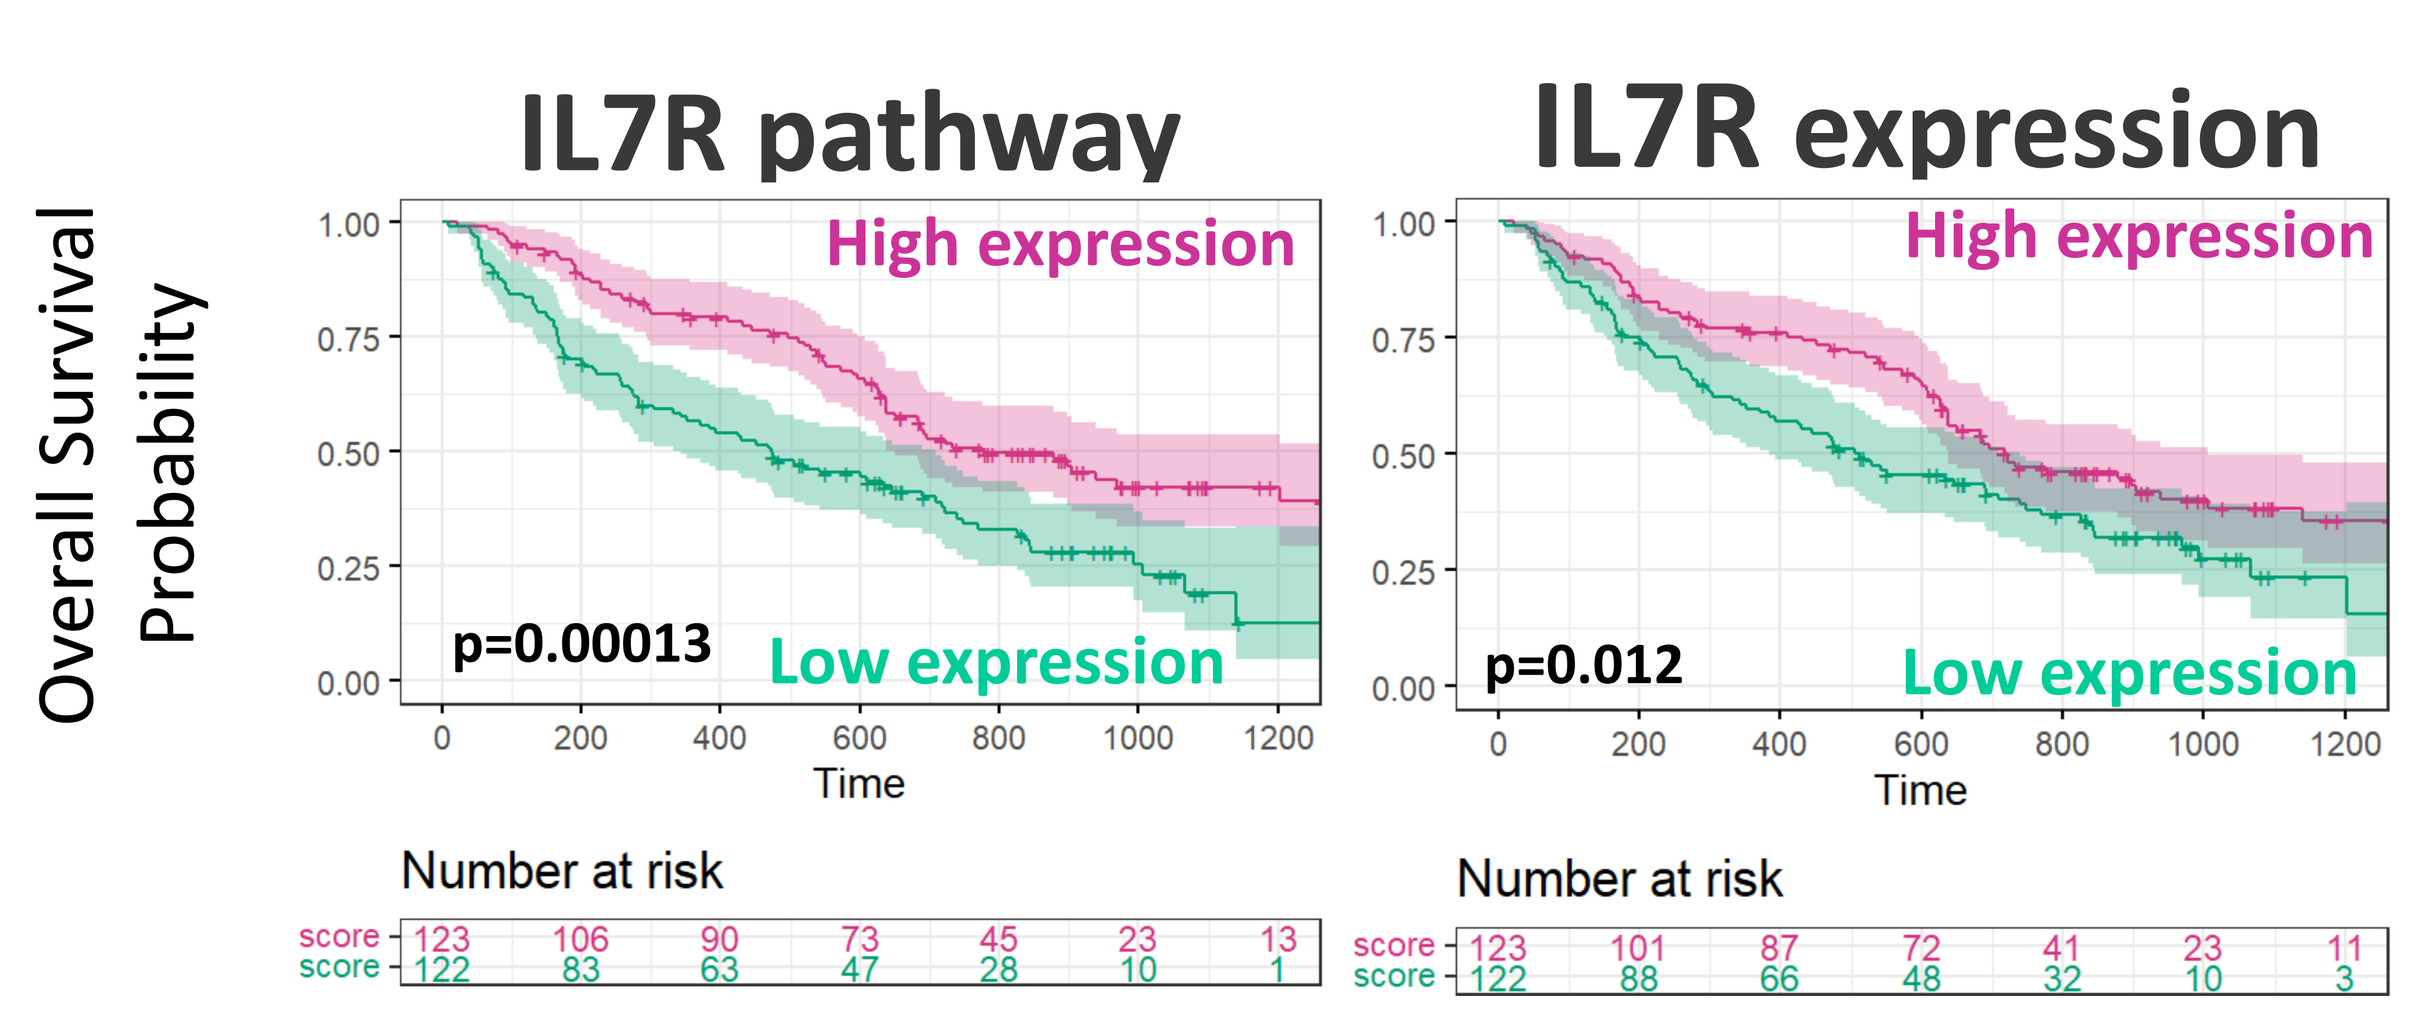

IL7R pathway
IL7R expression
Overall Survival Probability
High expression
Low expression
p=0.00013
p=0.012
Time
Number at risk
score
123
106
90
73
45
23
13
122
83
63
47
28
10
1
123
101
87
72
41
23
11
122
88
66
48
32
10
3

Sade-Feldman et al. 2018, Caushi et al. 2021, Zhang Y et al. 2021, Duraisway al.2021, *Gide et al. 2019, Liu et al.,2019, Riaz et al., 2017*

4/ Anti PD-1 IL7v monotherapy *in vivo* efficacy by promoting infiltration and proliferation of TCF1+ stem-like CD8 Tcells

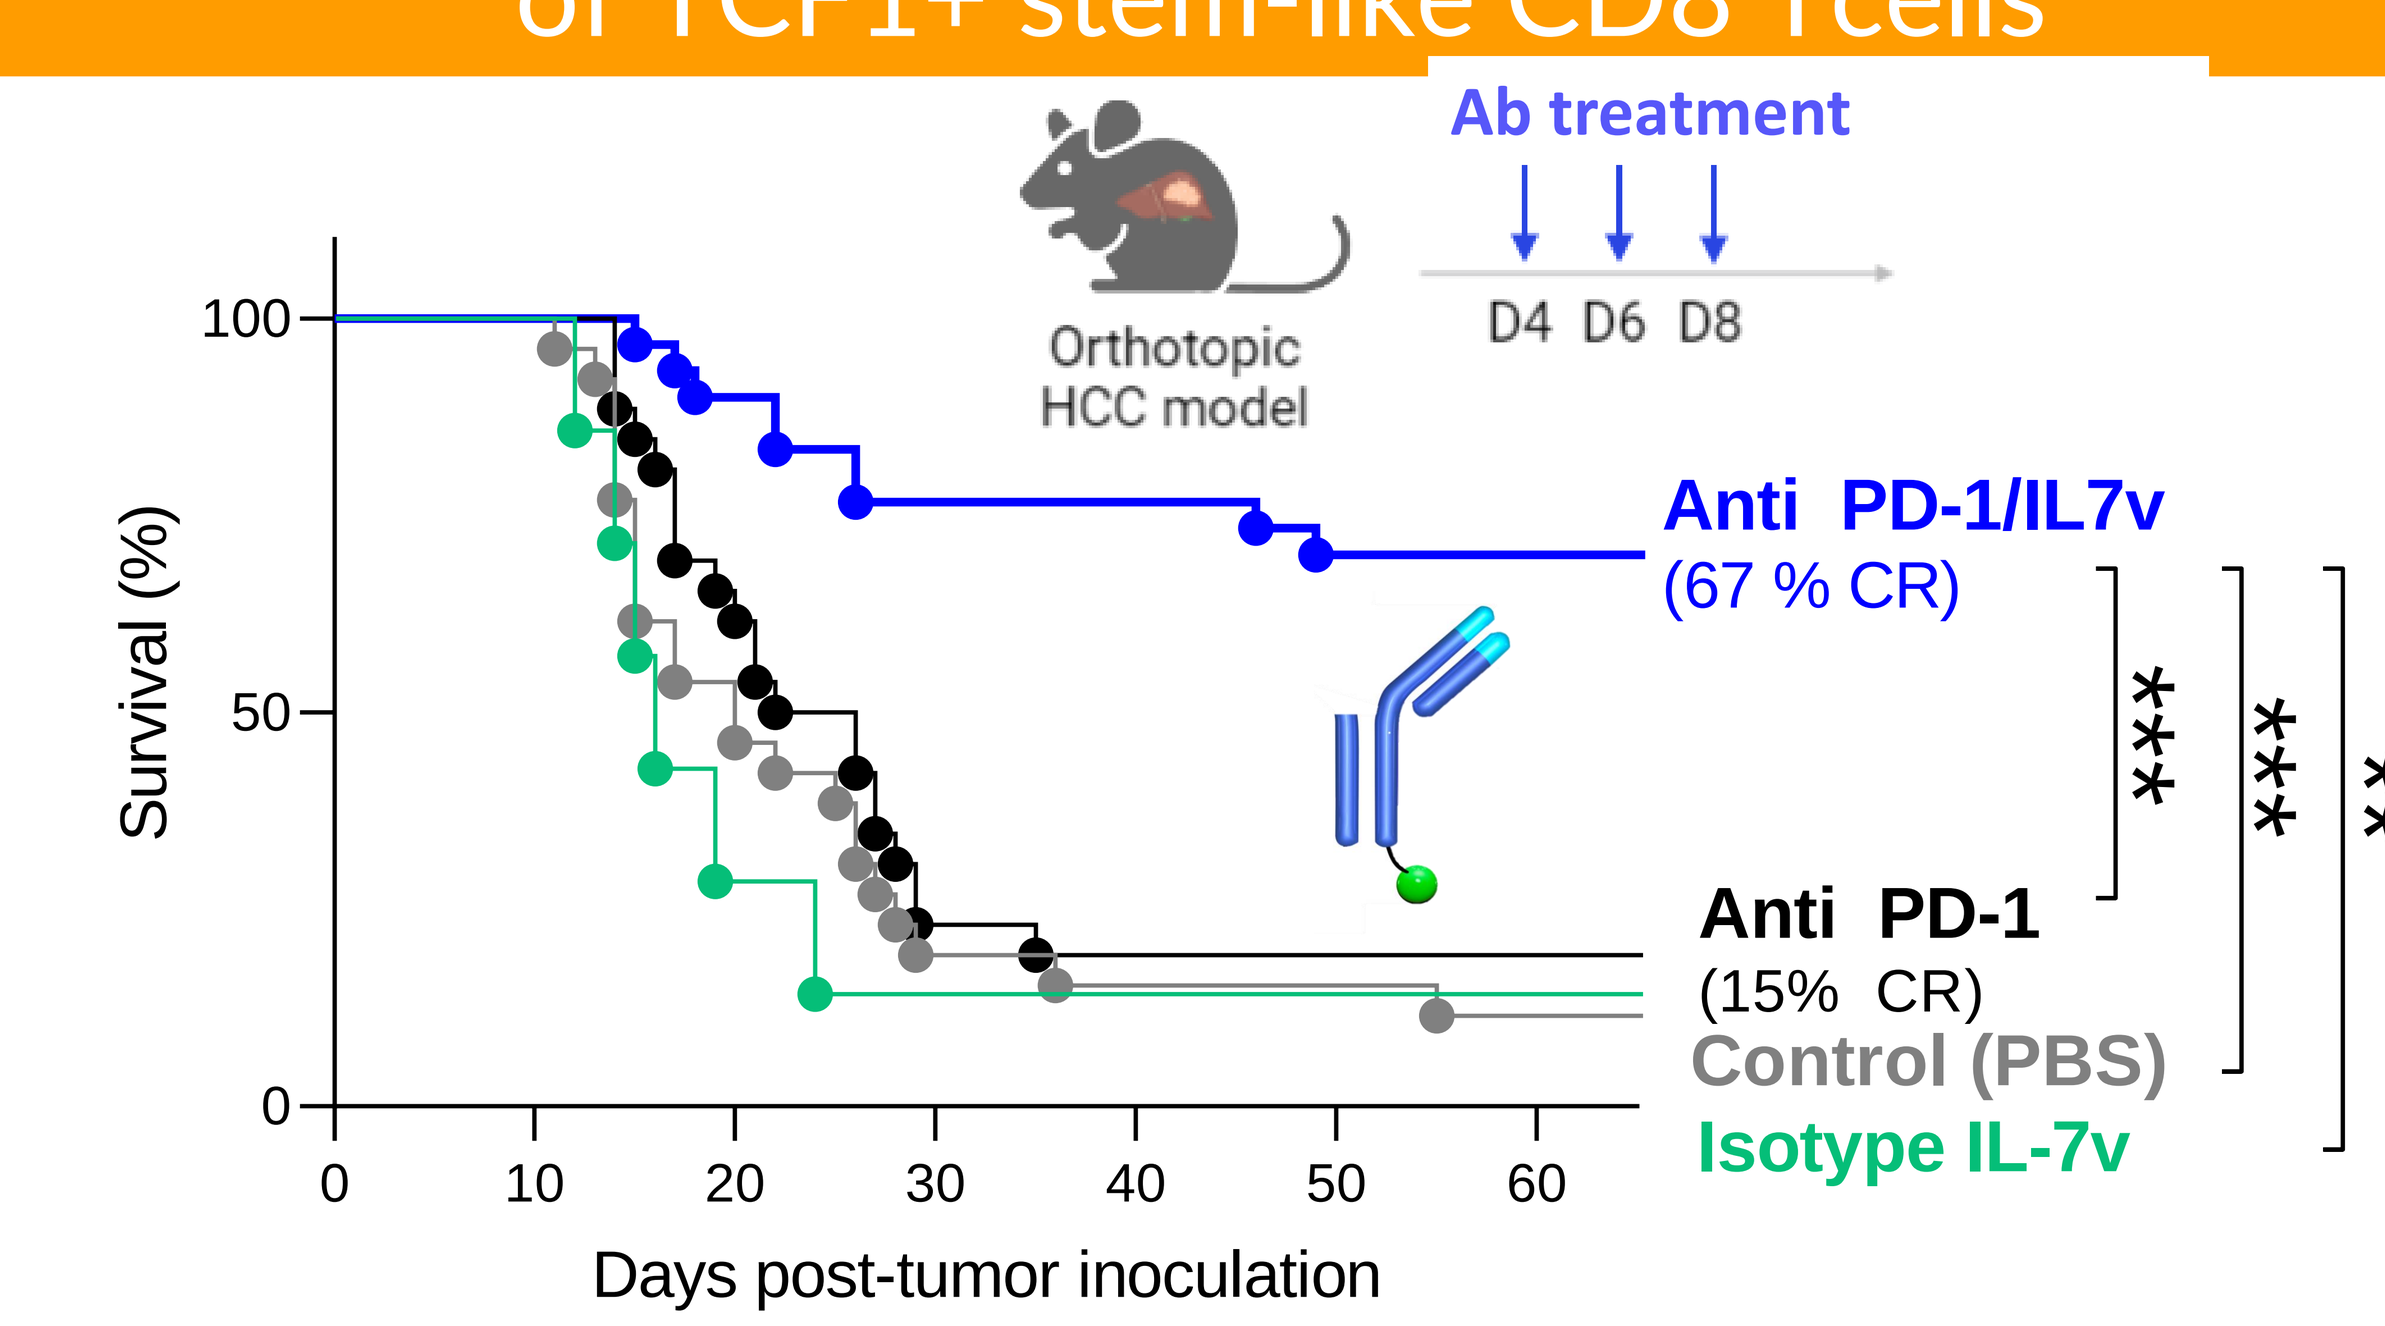

Orthotopic HCC model
Ab treatment
D4 D6 D8
Survival (%)
100
50
0
Days post-tumor inoculation
Anti PD-1/IL7v
(67 % CR)
Anti PD-1
(15% CR)
Control (PBS)
Isotype IL-7v

**

Anti PD-1 IL7v overcomes T cell exclusion & induces high proliferation of stem-like T cells

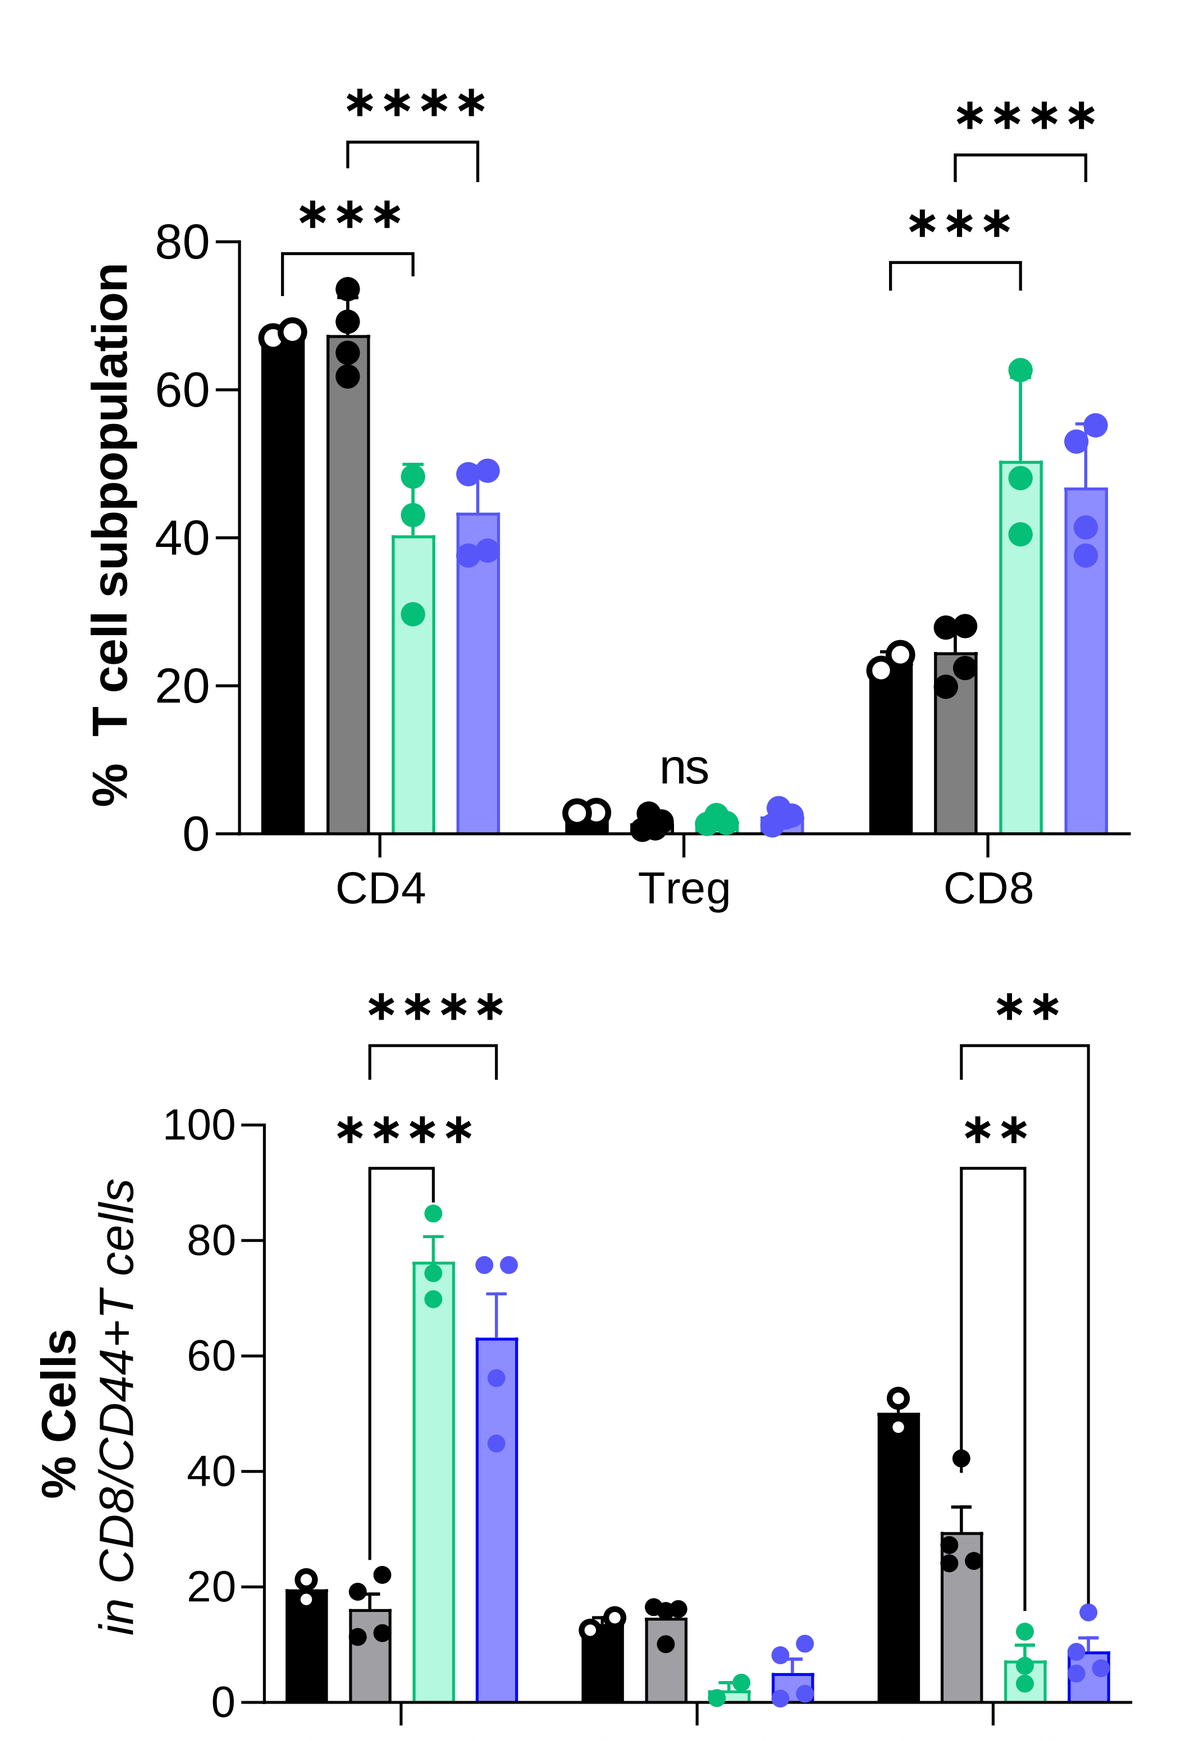

% T cell subpopulation
CD4
Treg
CD8
ns

% Cells in CD8/CD44+T cells
TCF1+ TOX-
TCF1+ TOX+
TCF1- TOX+
**

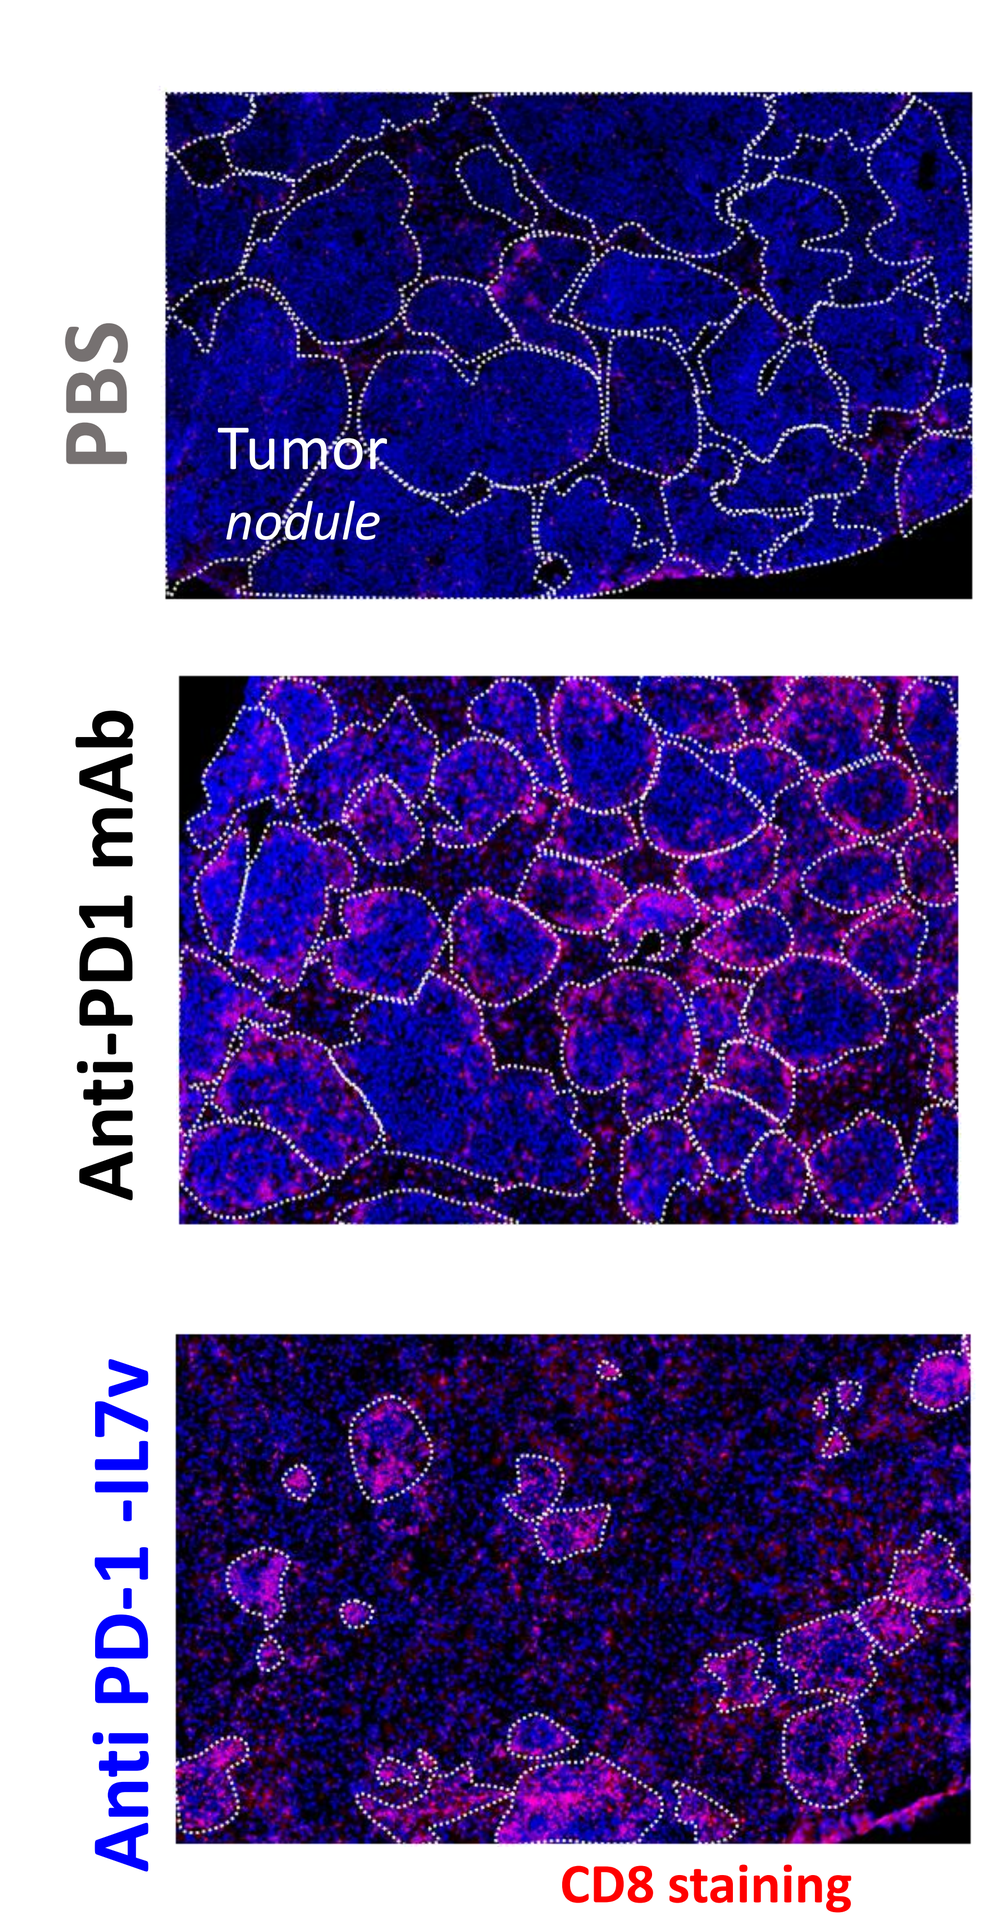

PBS
Tumor nodule
Anti-PD1 mAb
Anti PD-1 -IL7v
CD8 staining
nucleus

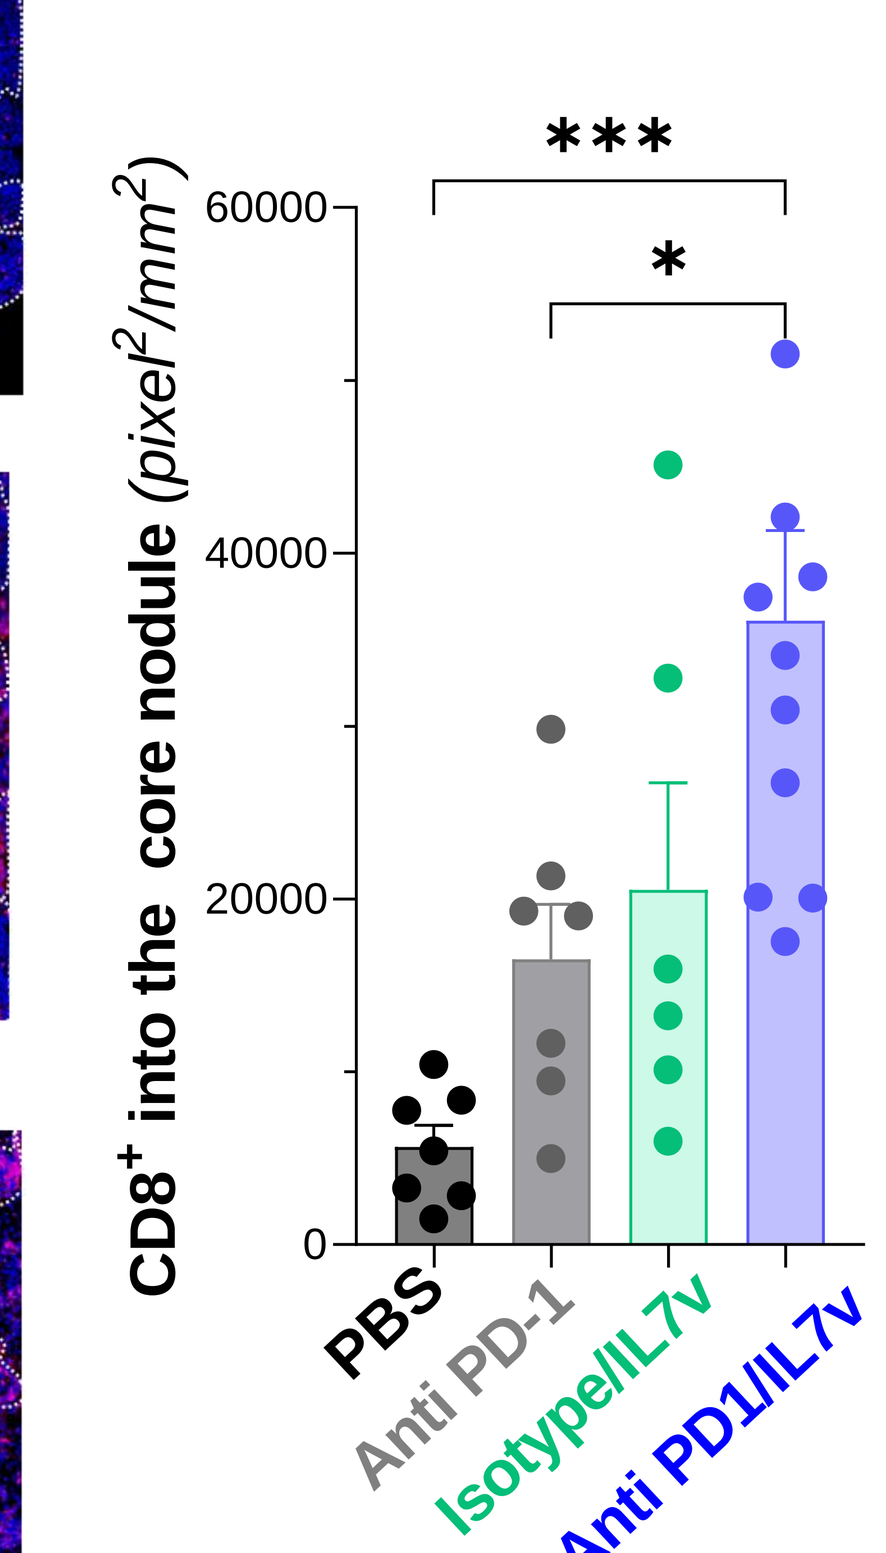

CD8+ into the core nodule (pixel²/mm²)
60000
40000
20000
0

*
PBS
Anti PD-1
Isotype/IL7v
Anti PD1/IL7v

5/ Anti PD-1/IL7v efficacy post α-PD(L)1 relapse model

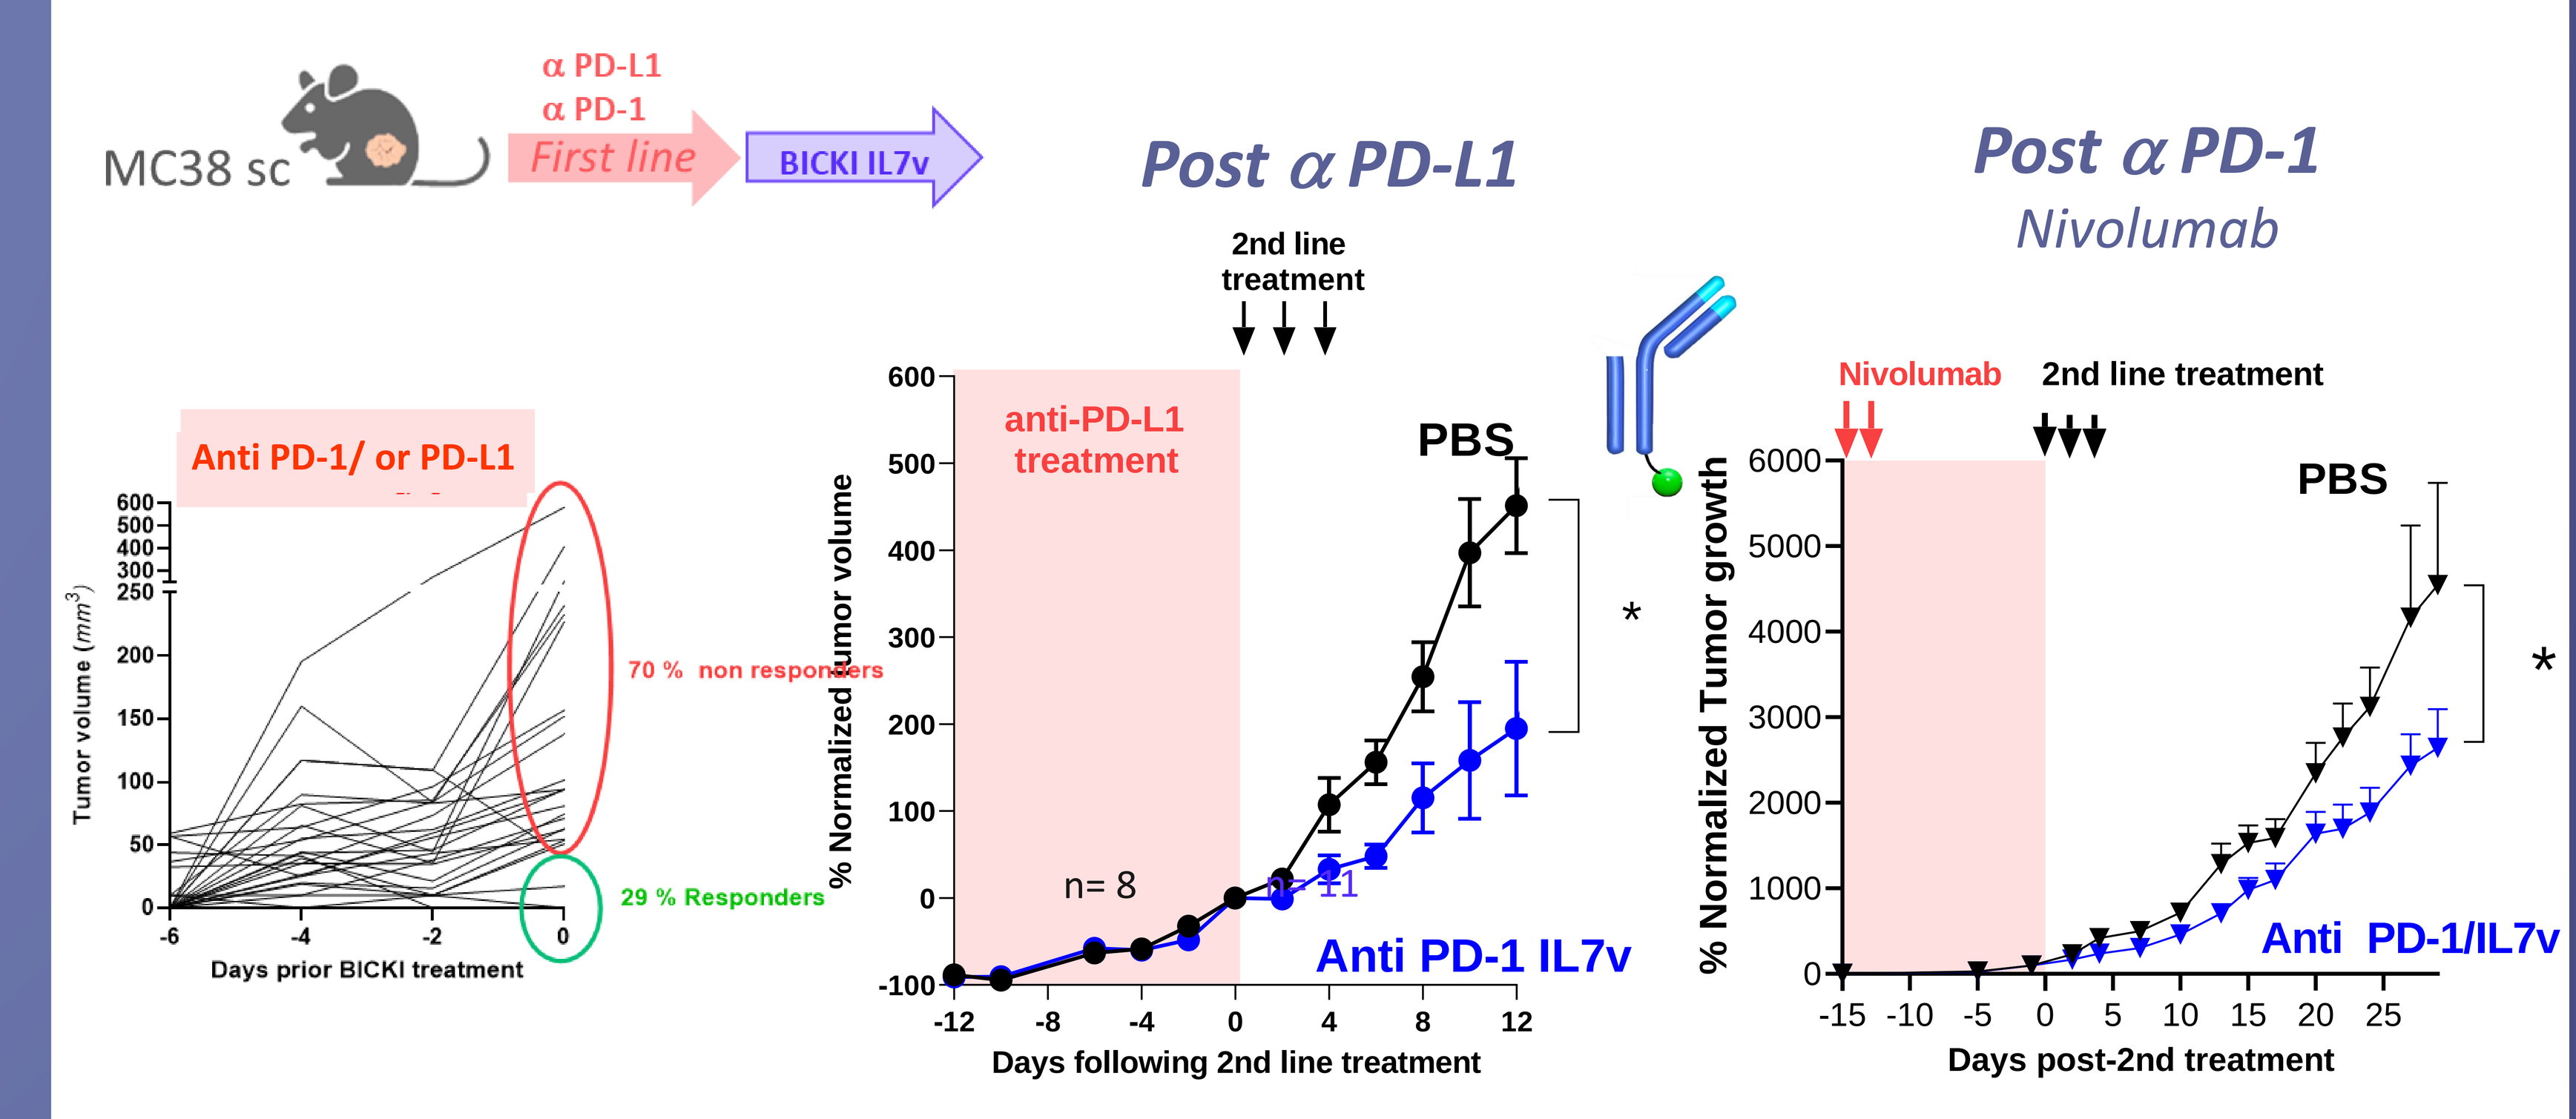

MC38 sc
α PD-L1
α PD-1
First line
BICKI IL7v
Anti PD-1/ or PD-L1
Tumor volume (mm³)
70 % non responders
29 % Responders
Days prior BICKI treatment
Post α PD-L1
2nd line treatment
anti-PD-L1 treatment
% Normalized tumor volume
PBS
n= 8
Anti PD-1 IL7v
Days following 2nd line treatment
*
Post α PD-1
Nivolumab
Nivolumab
2nd line treatment
% Normalized Tumor growth
PBS
Anti PD-1/IL7v
Days post-2nd treatment
*

Conclusion

To sustain a durable PD-1+ stem like CD8 T cell response

- Optimized molecule for Cis targeting activity
- Conserved high PD-1 binding and PD-1/PD-L1 antagonist activity
- Allows a selective delivery of IL-7 on PD-1+ cells
- Promotes proliferation of PD-1+ CD127+ TCF1+ stemness T cells
- Significant *in vivo* anti tumor efficacy
- in PD-1 sensitive, resistant syngeneic and humanized models
- *In vivo* efficacy post αPD-1 or α-PD-L1 refractory tumor