Anti-PD-1/IL-7v immunocytokine (BICKI®IL7v) favors proliferation & survival of TCF1+ stem like memory T cells and a durable in vivo efficacy in monotherapy or using combinatorial strategies

Aurore Morello, Justine Durand, Margaux Seité, Isabelle Girault, Géraldine Teppaz, Virginie Thepenier, Cécile Batty, Ariane Desselle, Emmanuelle Wilhelm, Kevin Biteau, Marine Malloci, Stéphanie Neyton, Vanessa Gauttier, Caroline Mary, Nicolas Poirier

OSE IMMUNO THERAPEUTICS

BICKI®IL7, an optimized *design*
for selective cis-potentiation of PD-1+IL-7R+ T cells

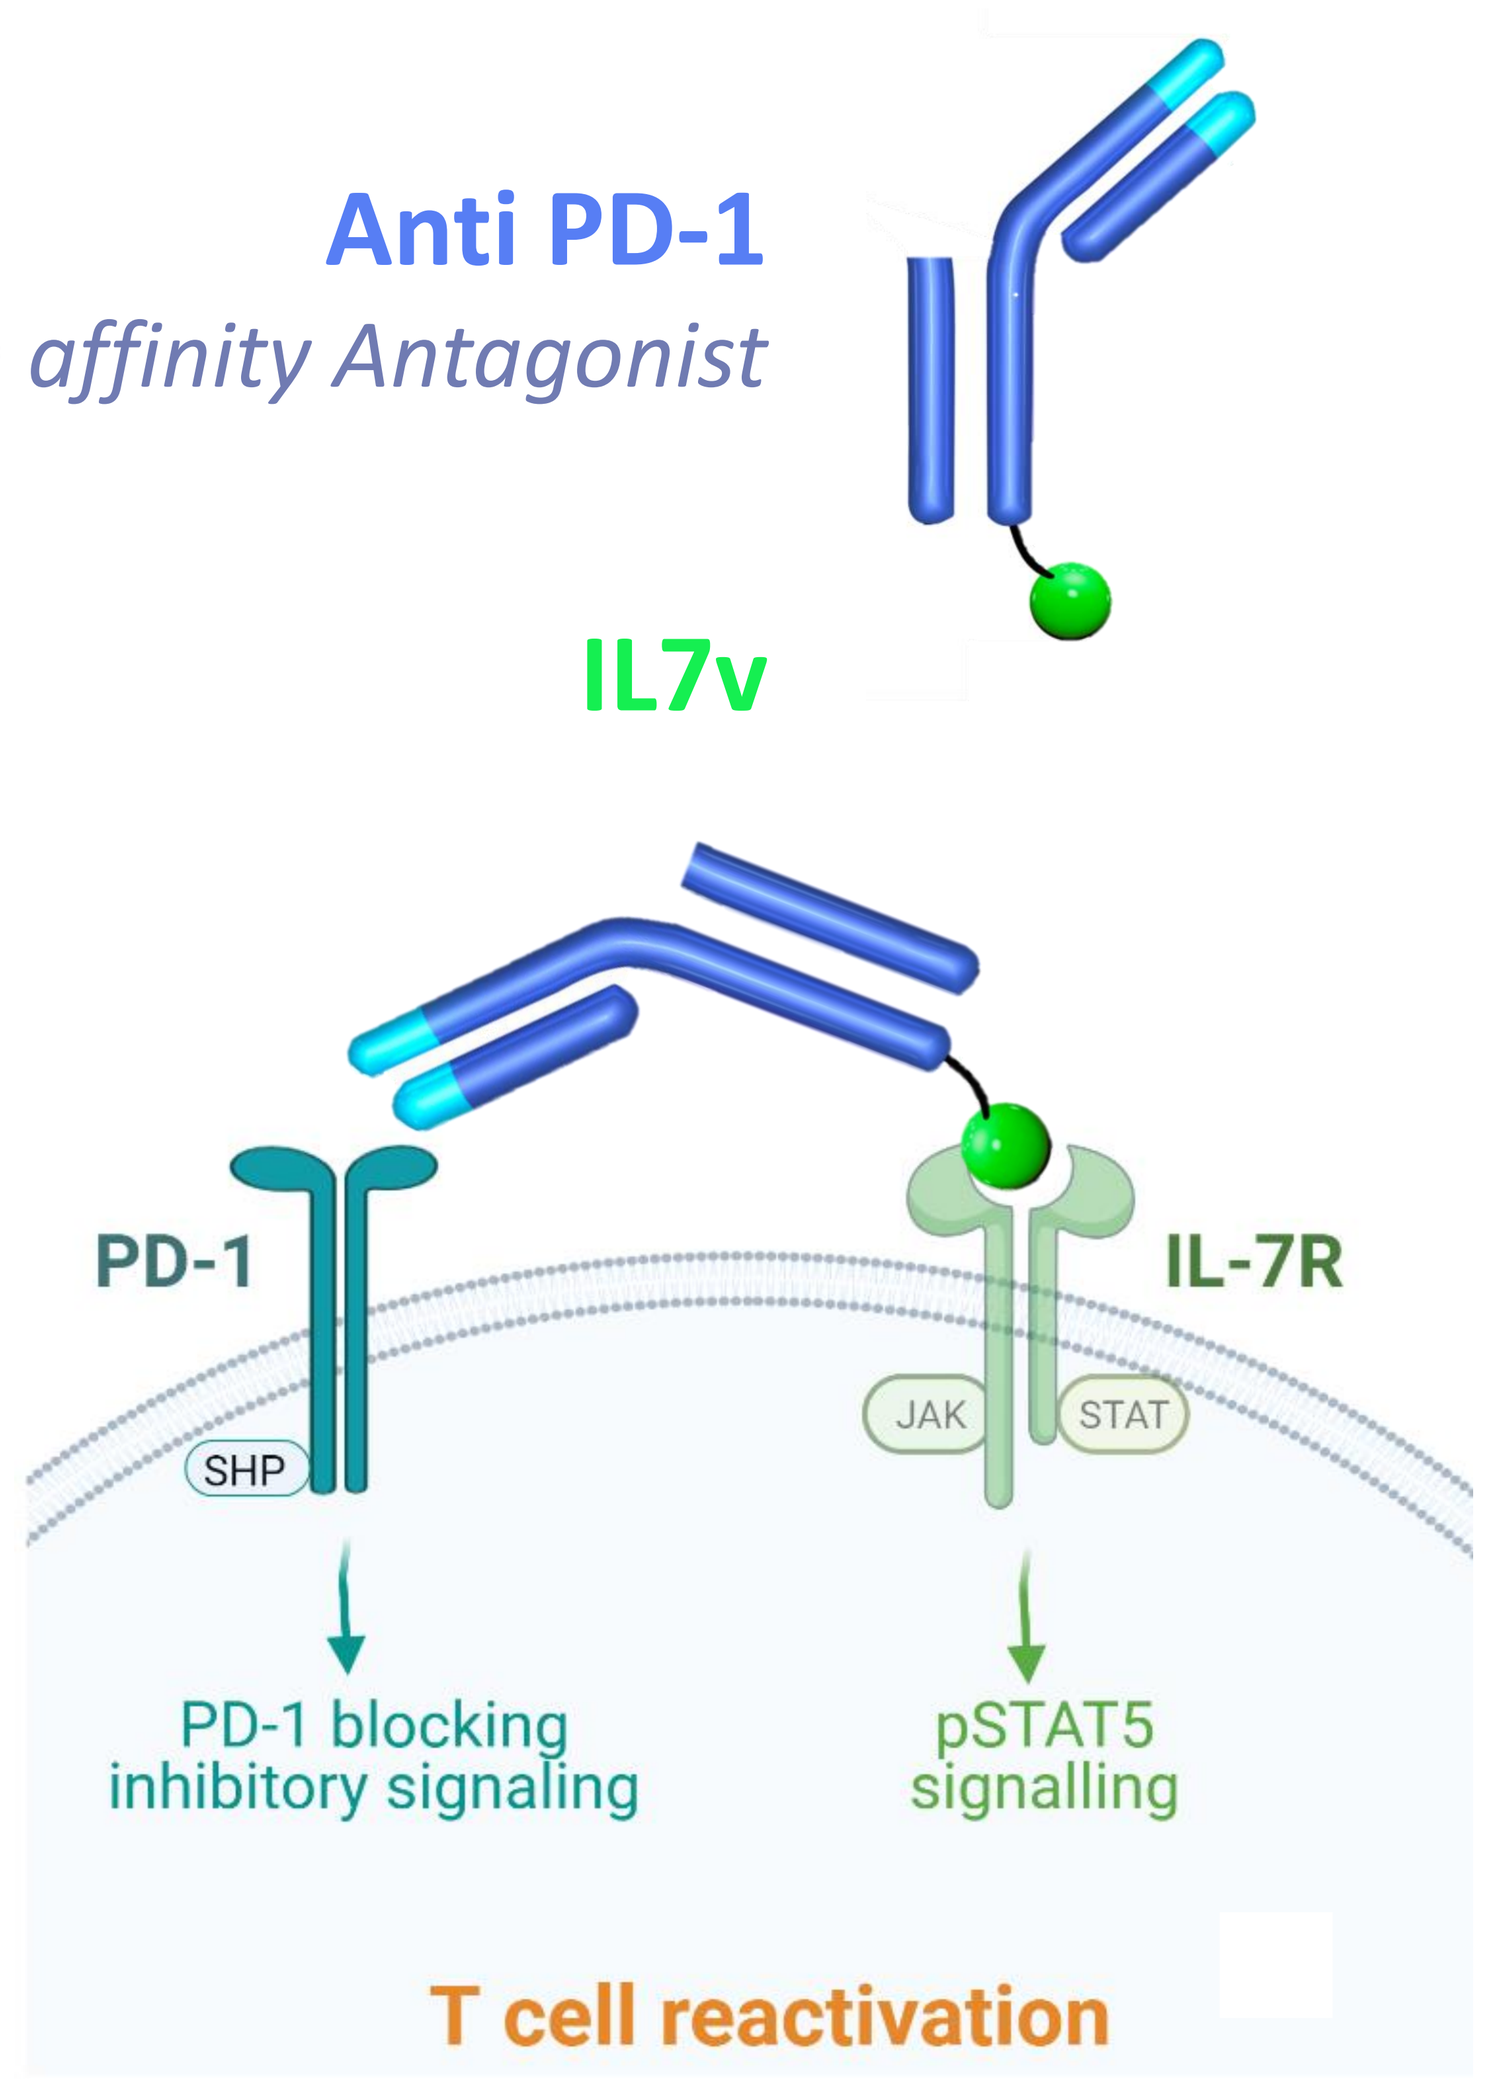
Anti PD-1
High affinity Antagonist
IL7v
PD-1
IL-7R
SHP
JAK
STAT
PD-1 blocking inhibitory signaling
pSTAT5 signalling
T cell reactivation

Enhanced Drug exposure

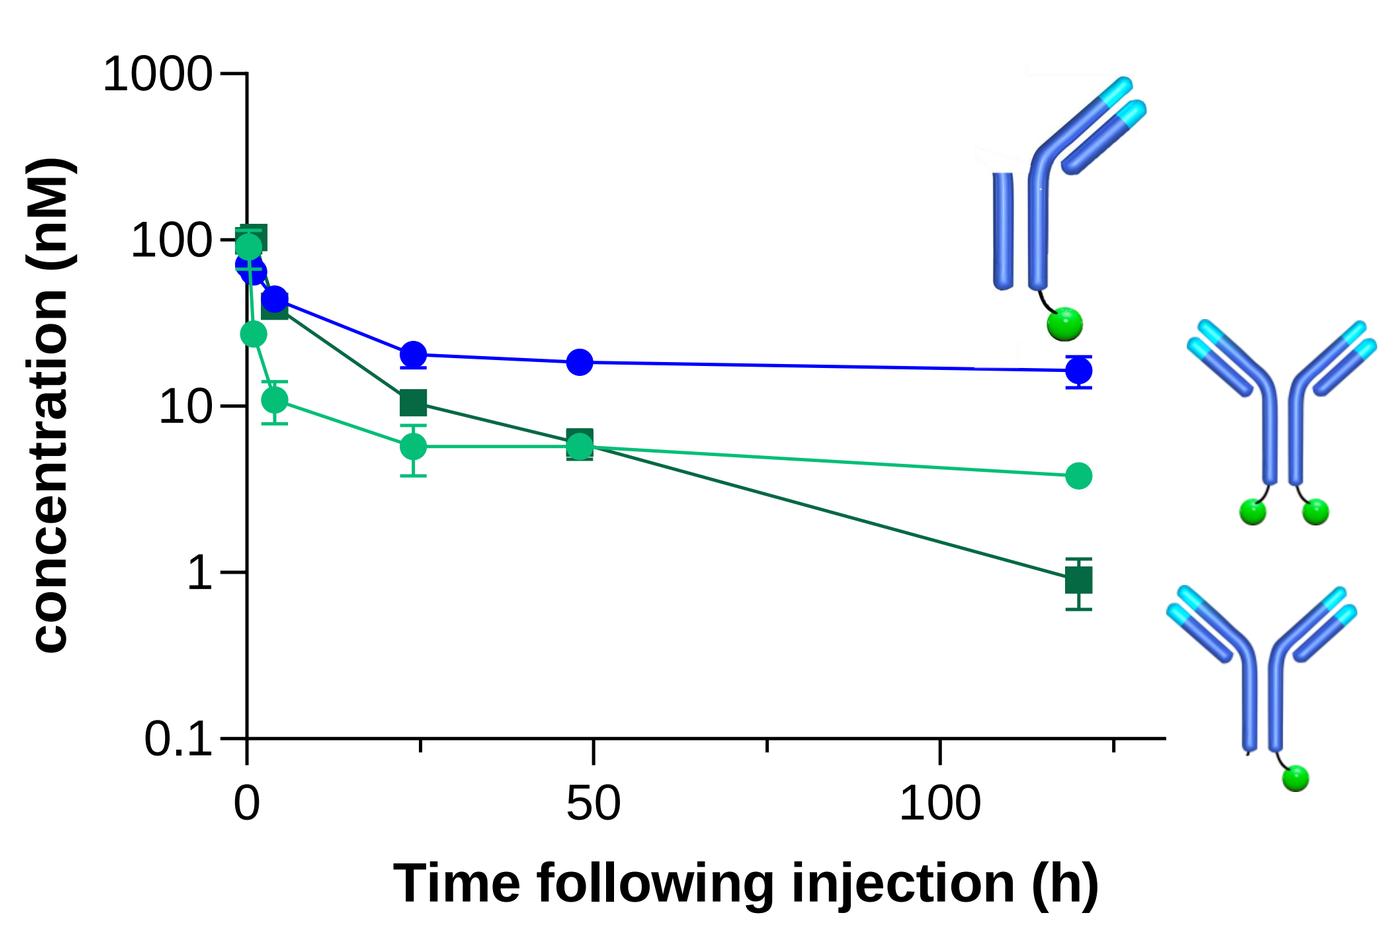
concentration (nM)
1000
100
10
1
0.1
0
50
100
Time following injection (h)

Improved Cis activity

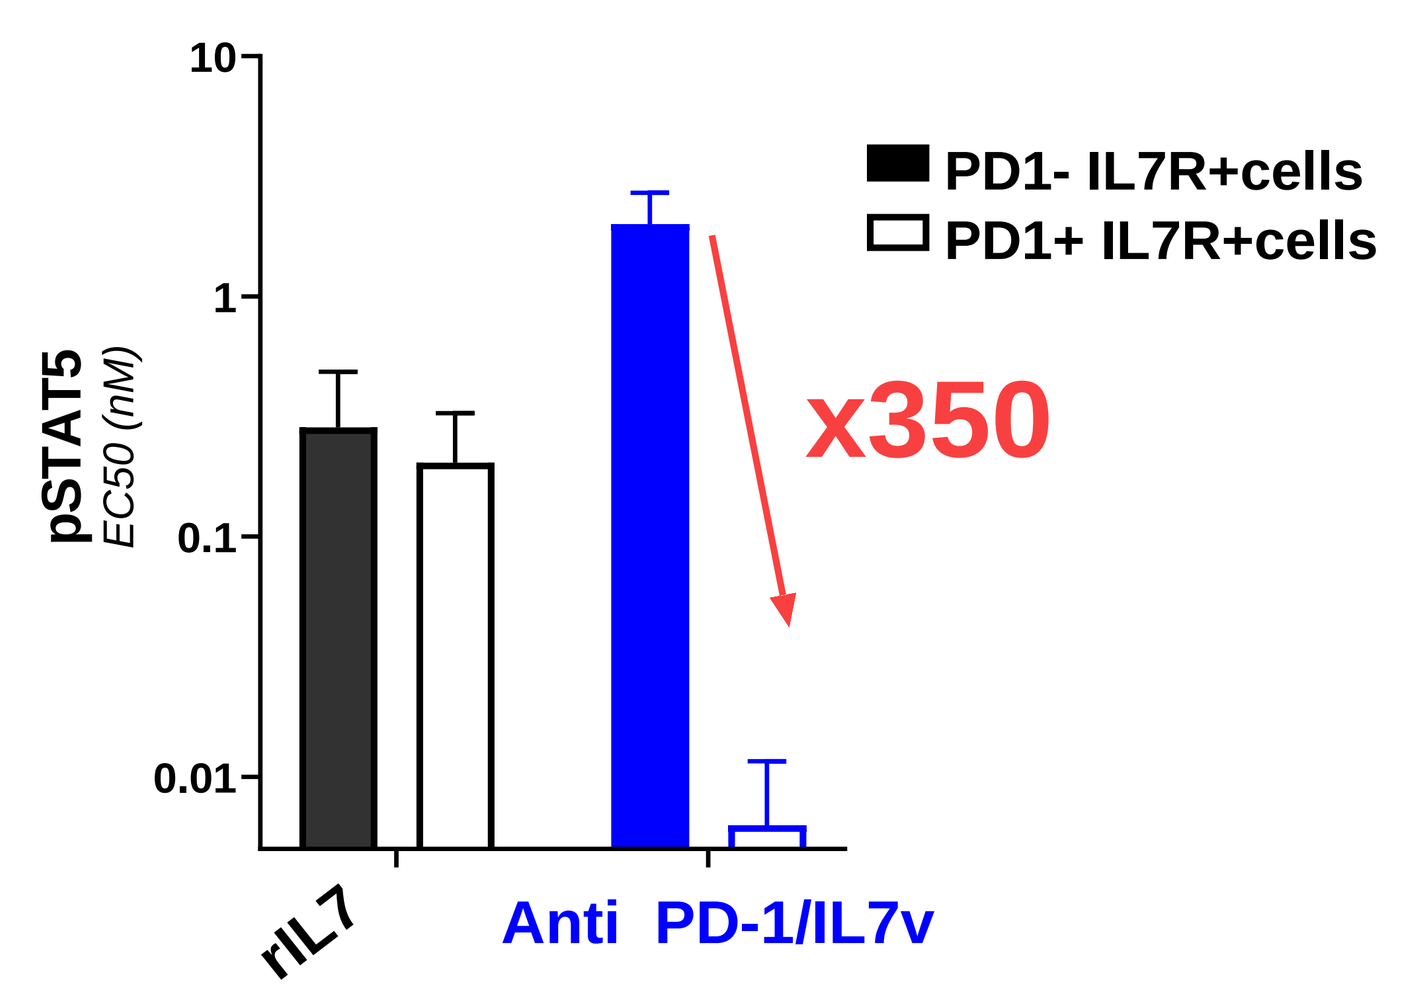
pSTAT5
EC50 (nM)
10
1
0.1
0.01
PD1- IL7R+cells
PD1+ IL7R+cells
x350
rIL7
Anti PD-1/IL7v

1/ BICKI®IL-7v promotes long-term proliferation of stem-like TCF1+ CD8 T cells

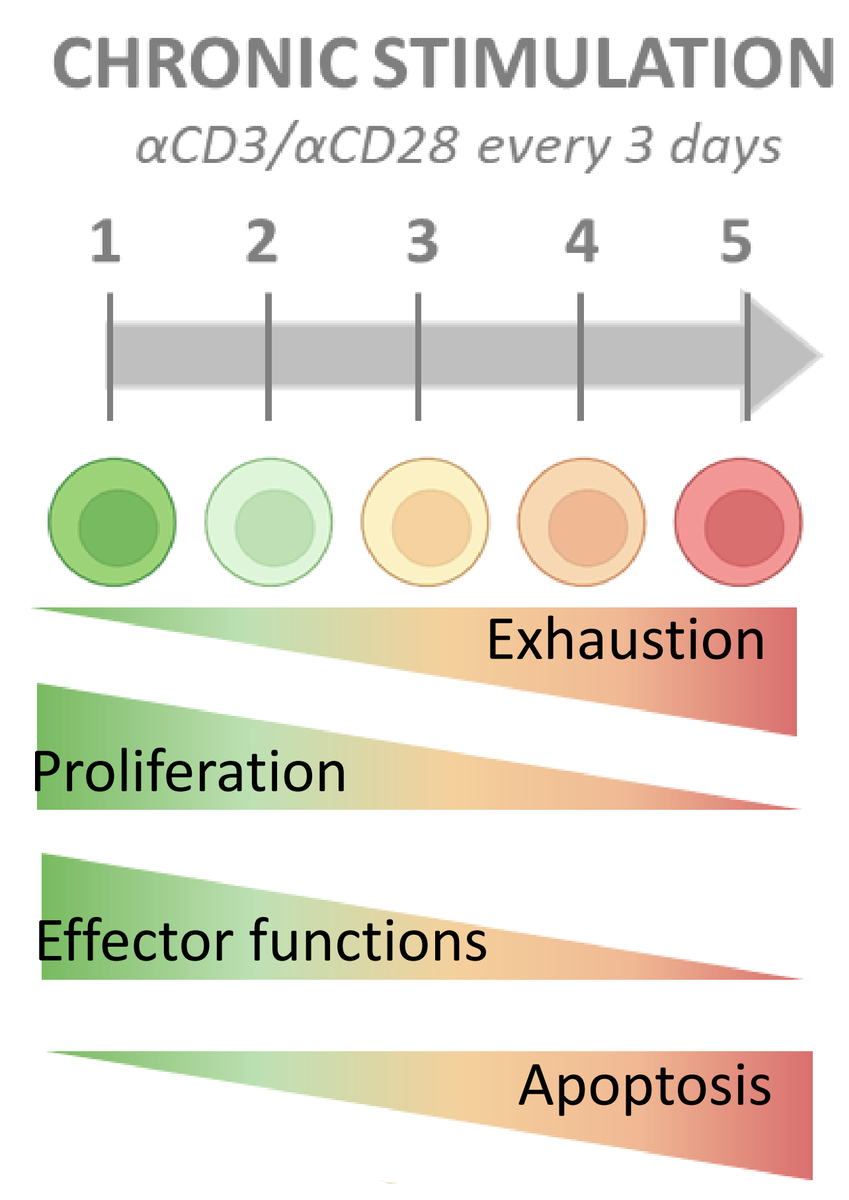
CHRONIC STIMULATION
αCD3/αCD28 every 3 days
1
2
3
4
5
Exhaustion
Proliferation
Effector functions
Apoptosis

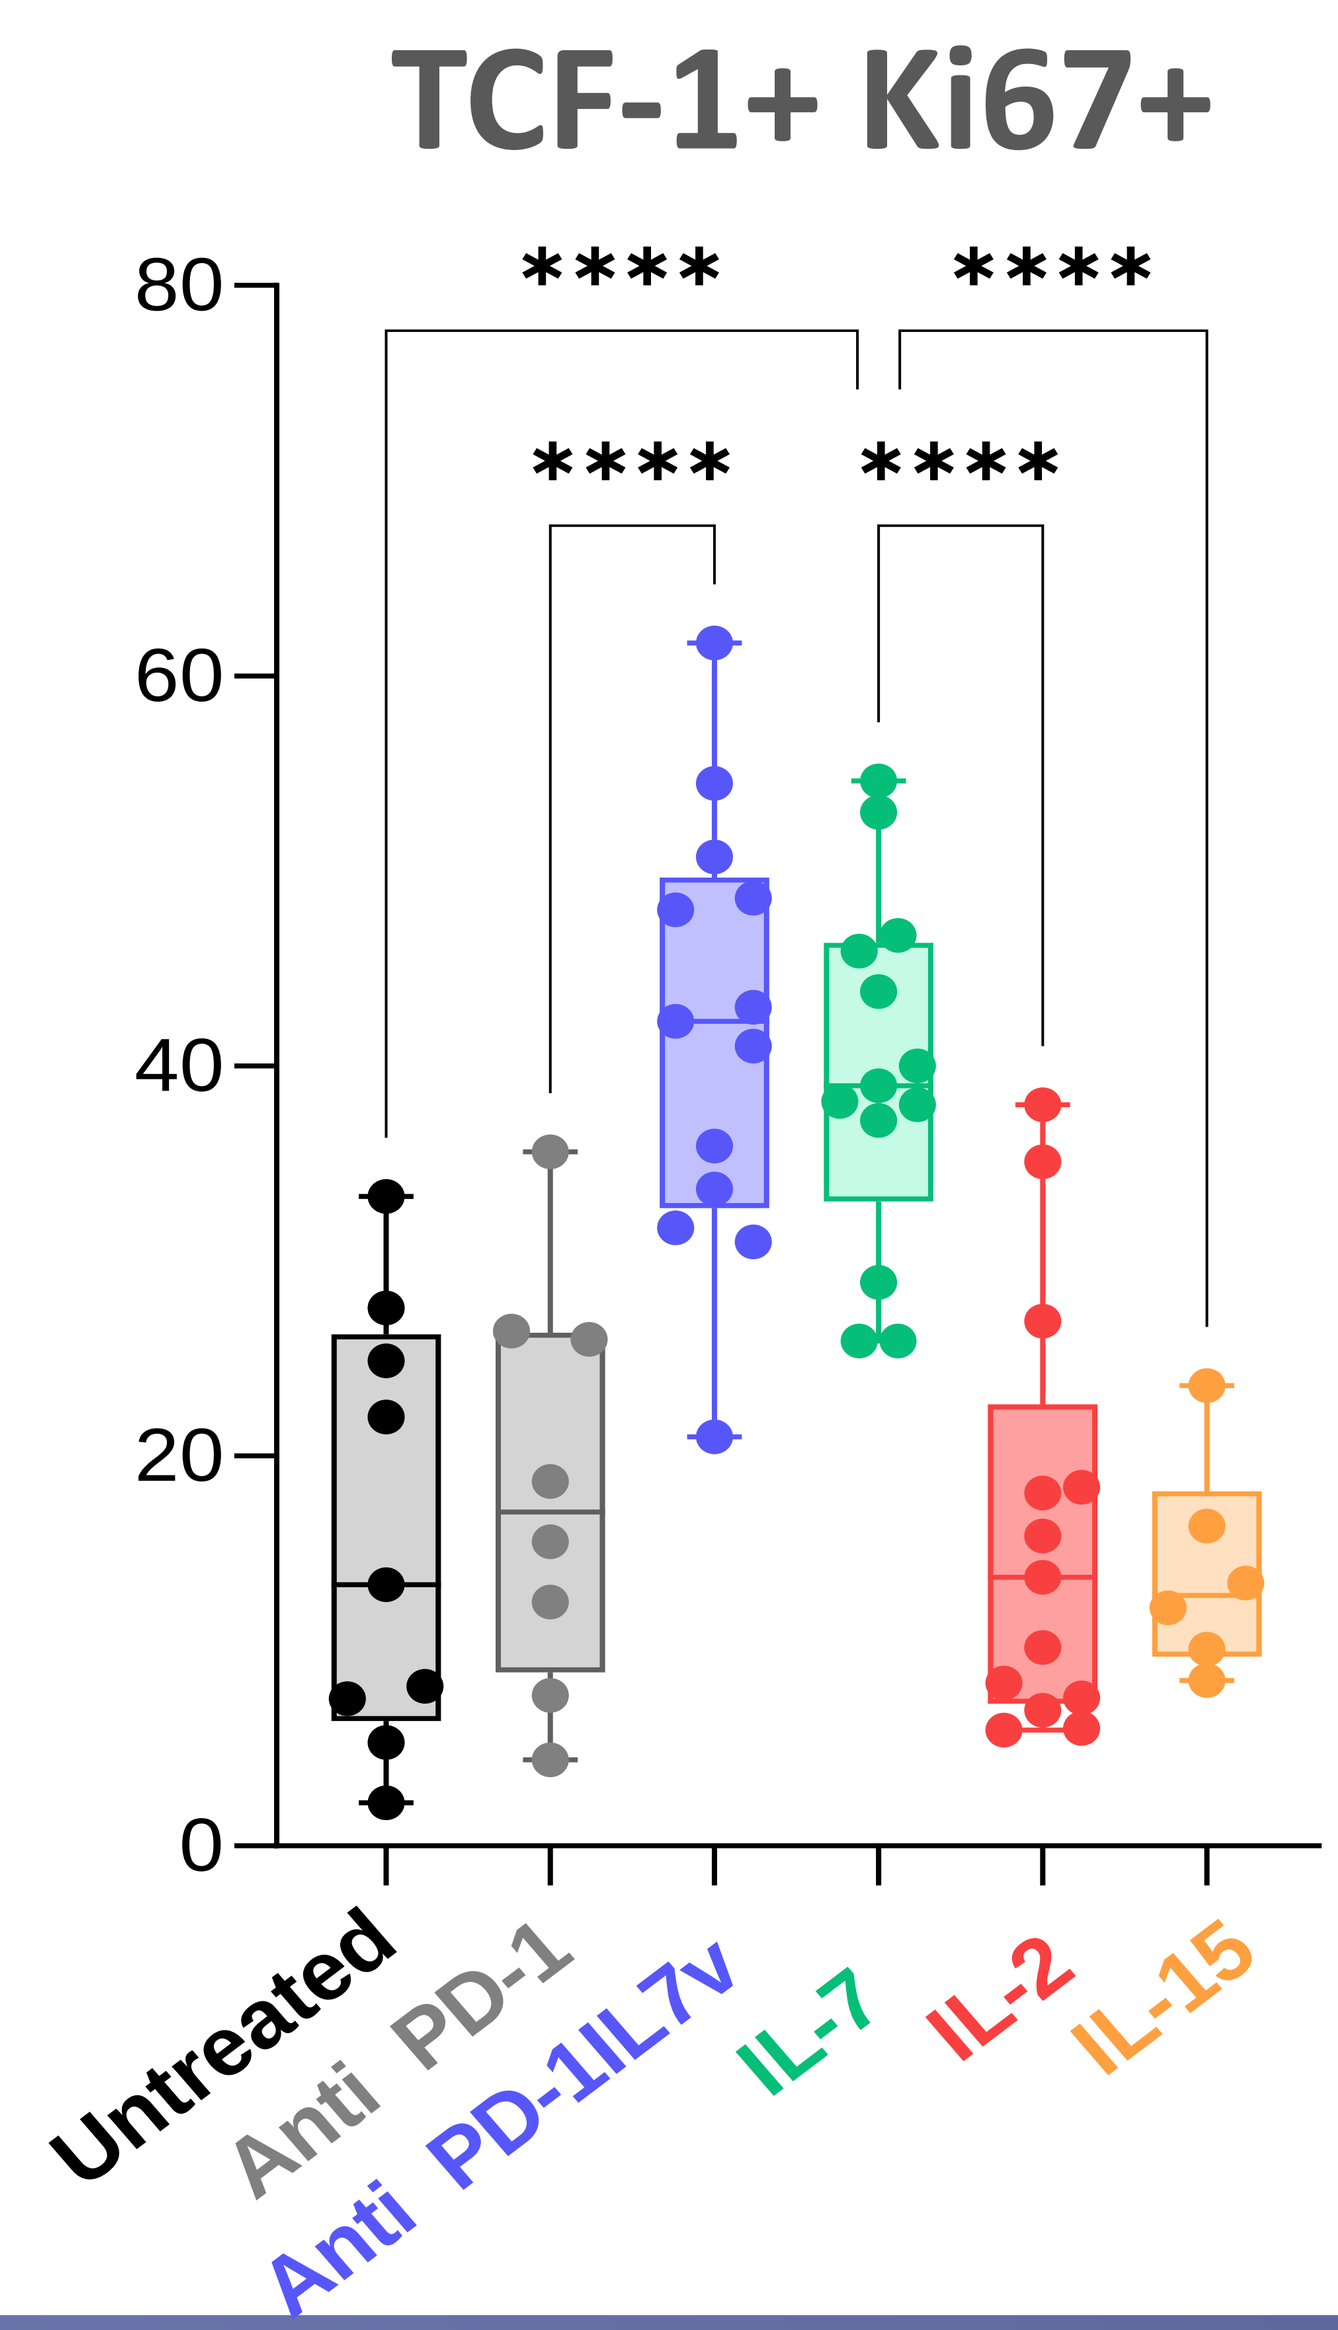
TCF-1+ Ki67+
80
60
40
20
0

Untreated
Anti PD-1
Anti PD-1IL7v
IL-7
IL-2
IL-15

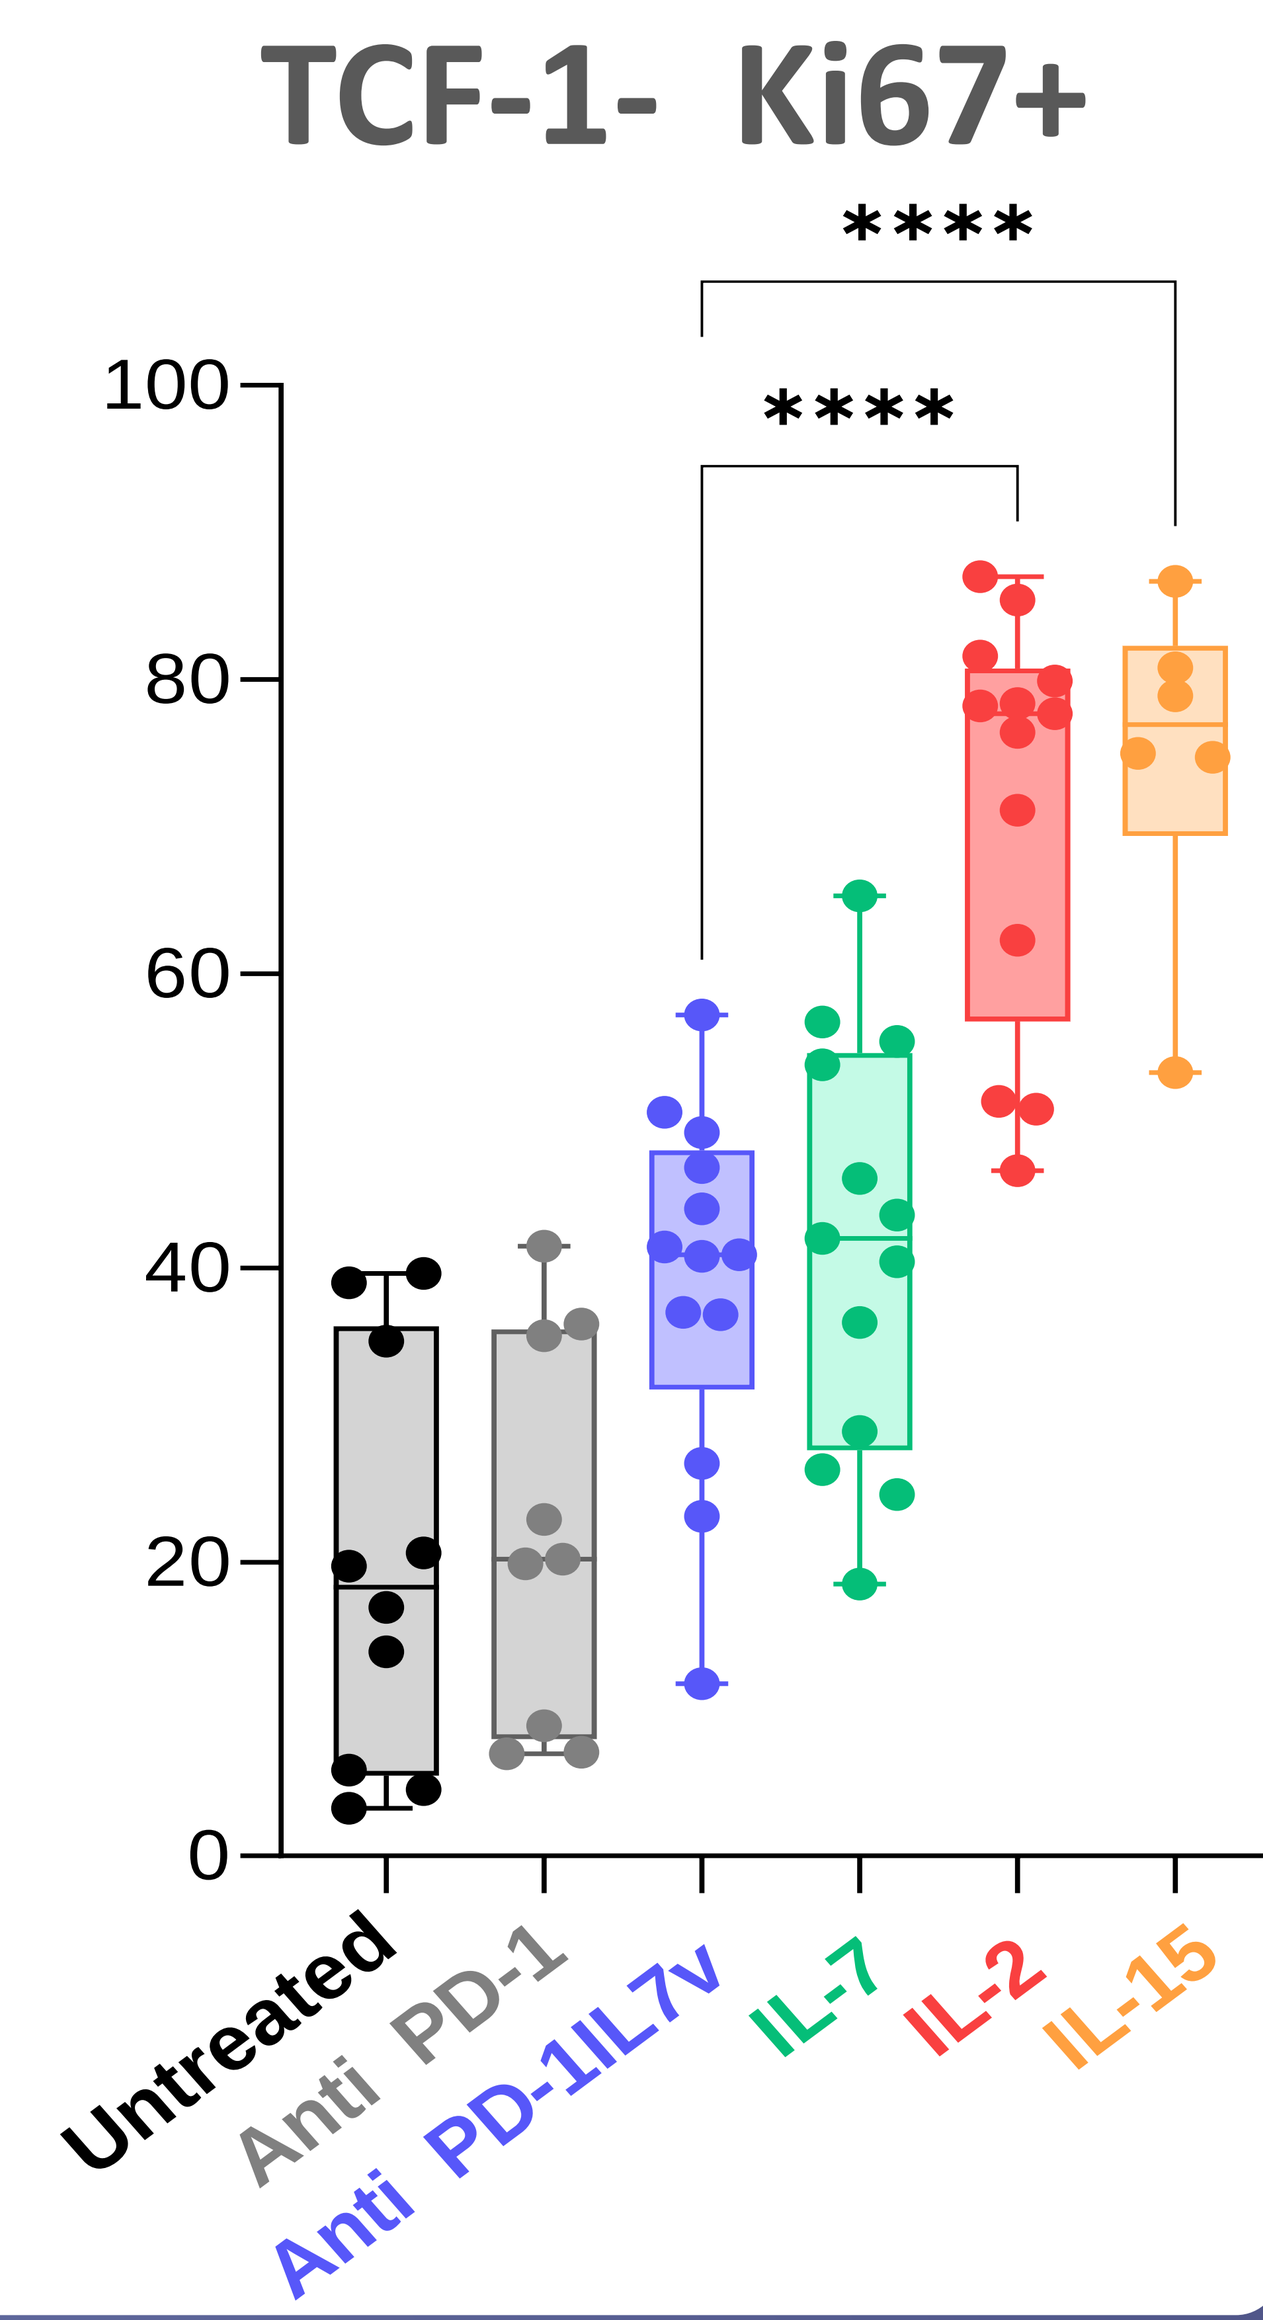
TCF-1- Ki67+
100
80
60
40
20
0

Untreated
Anti PD-1
Anti PD-1IL7v
IL-7
IL-2
IL-15

2/ Efficient monotherapy efficacy in vivo
by promoting infiltration and proliferation of TCF1+ memory stem-like CD8 T cells

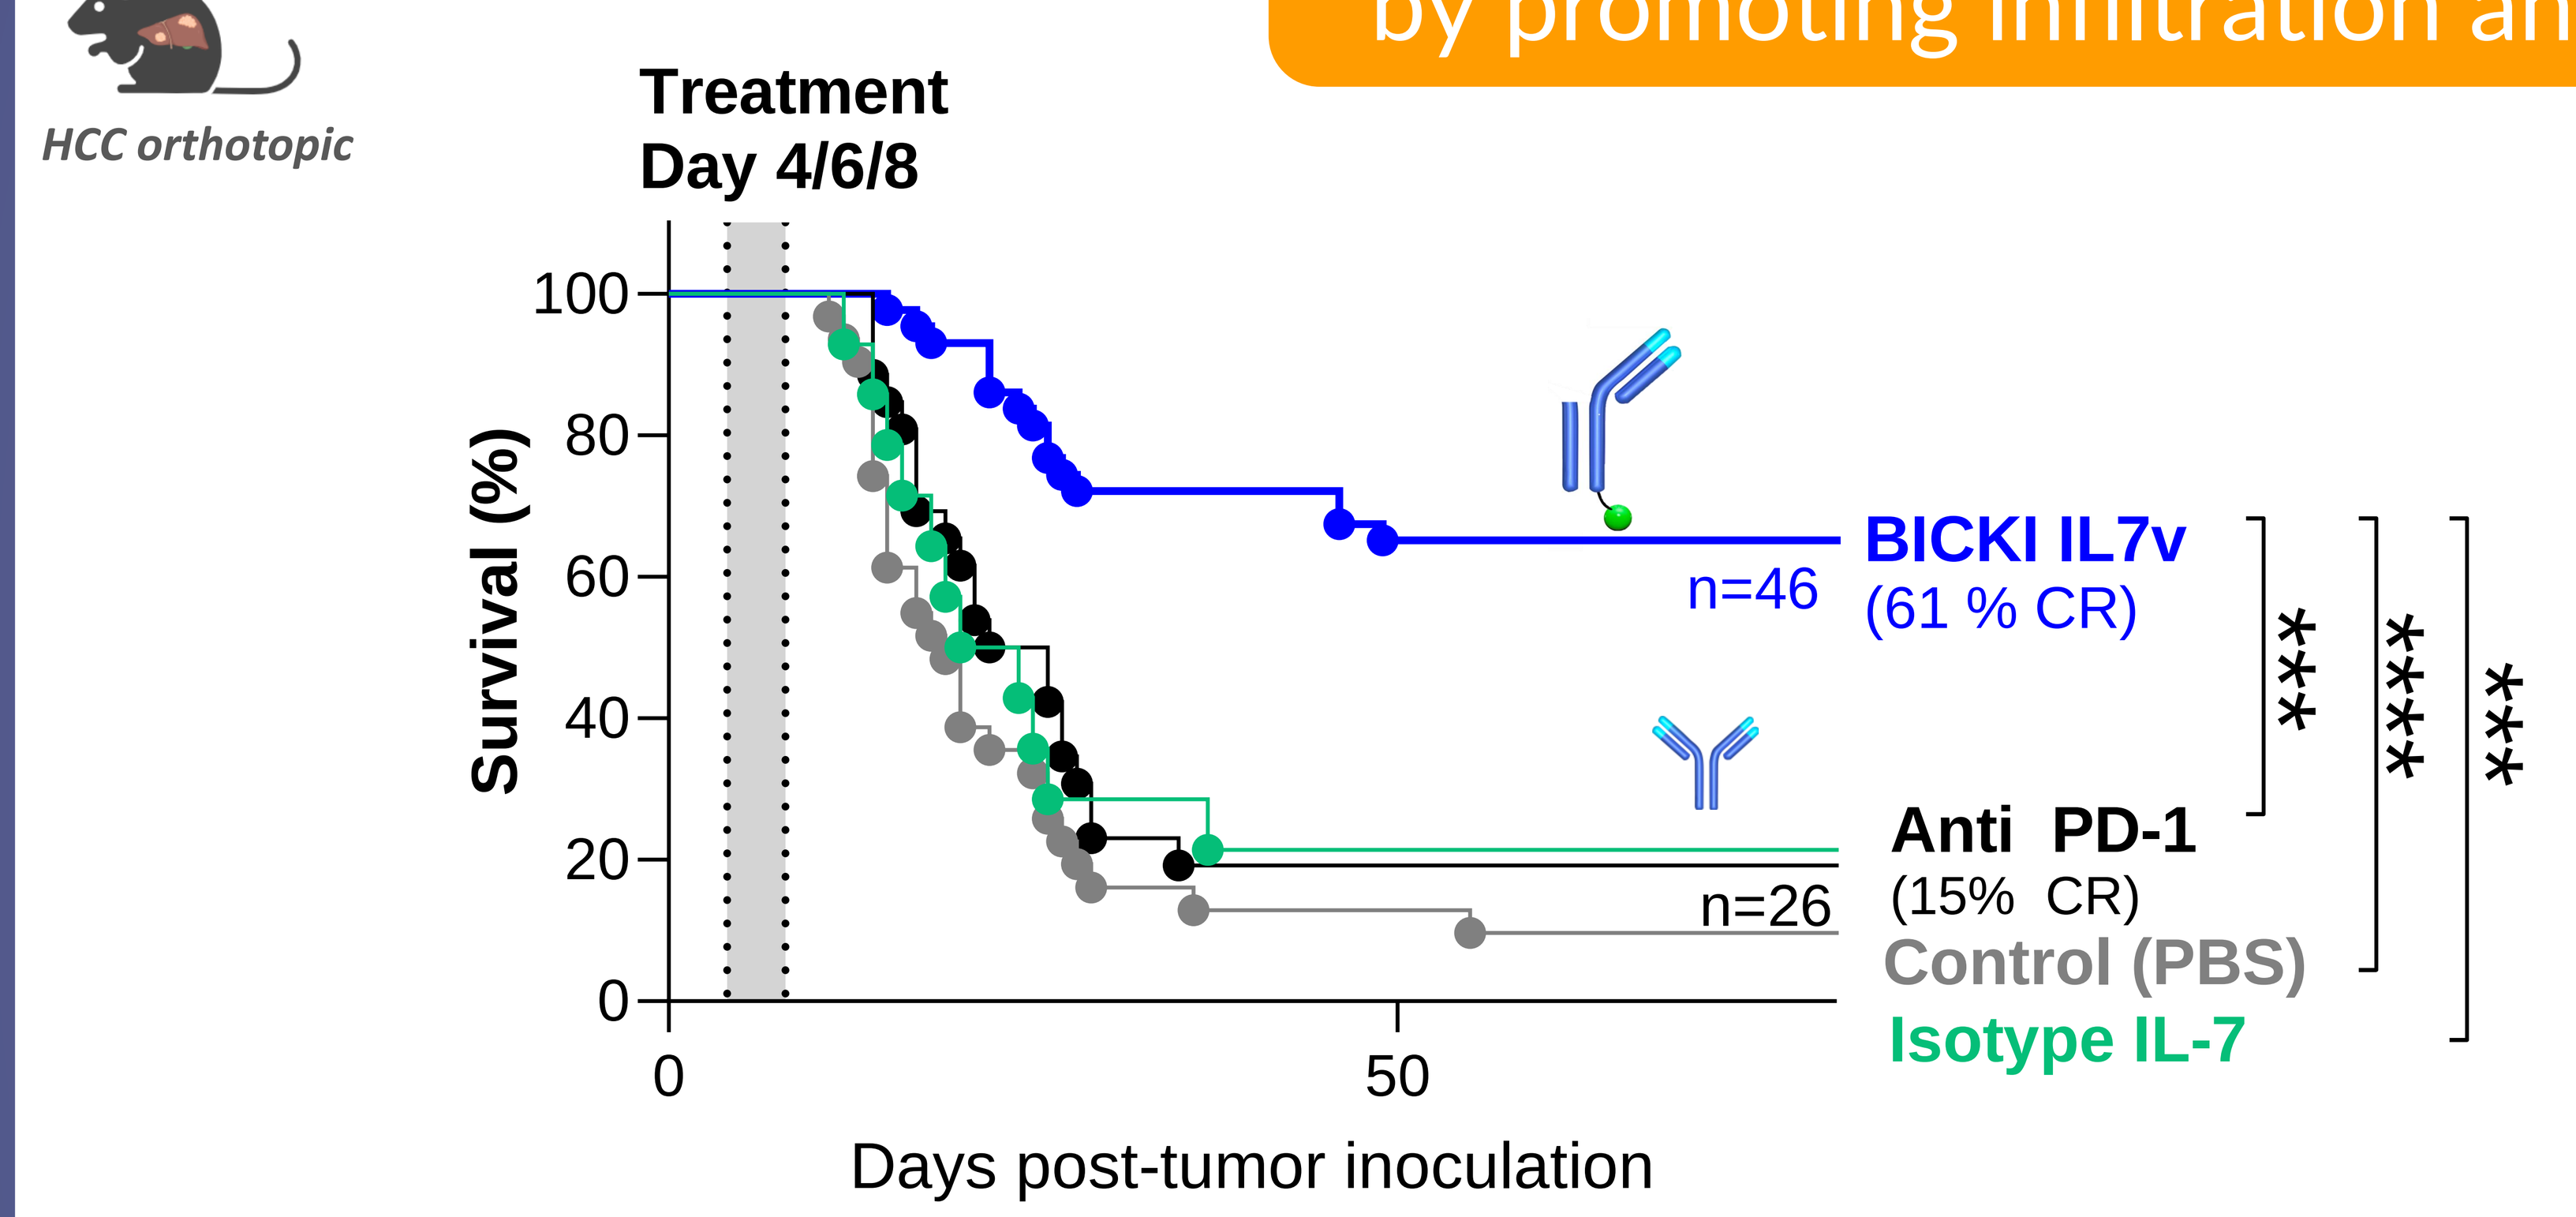
HCC orthotopic
Treatment
Day 4/6/8
Survival (%)
100
80
60
40
20
0
0
50
Days post-tumor inoculation
BICKI IL7v
n=46
(61 % CR)
Anti PD-1
(15% CR)
n=26
Control (PBS)
Isotype IL-7

BICKI®IL7 efficacy relies on proliferation of Infiltrating T cells but also induces recruitment of fresh T cells into the TME

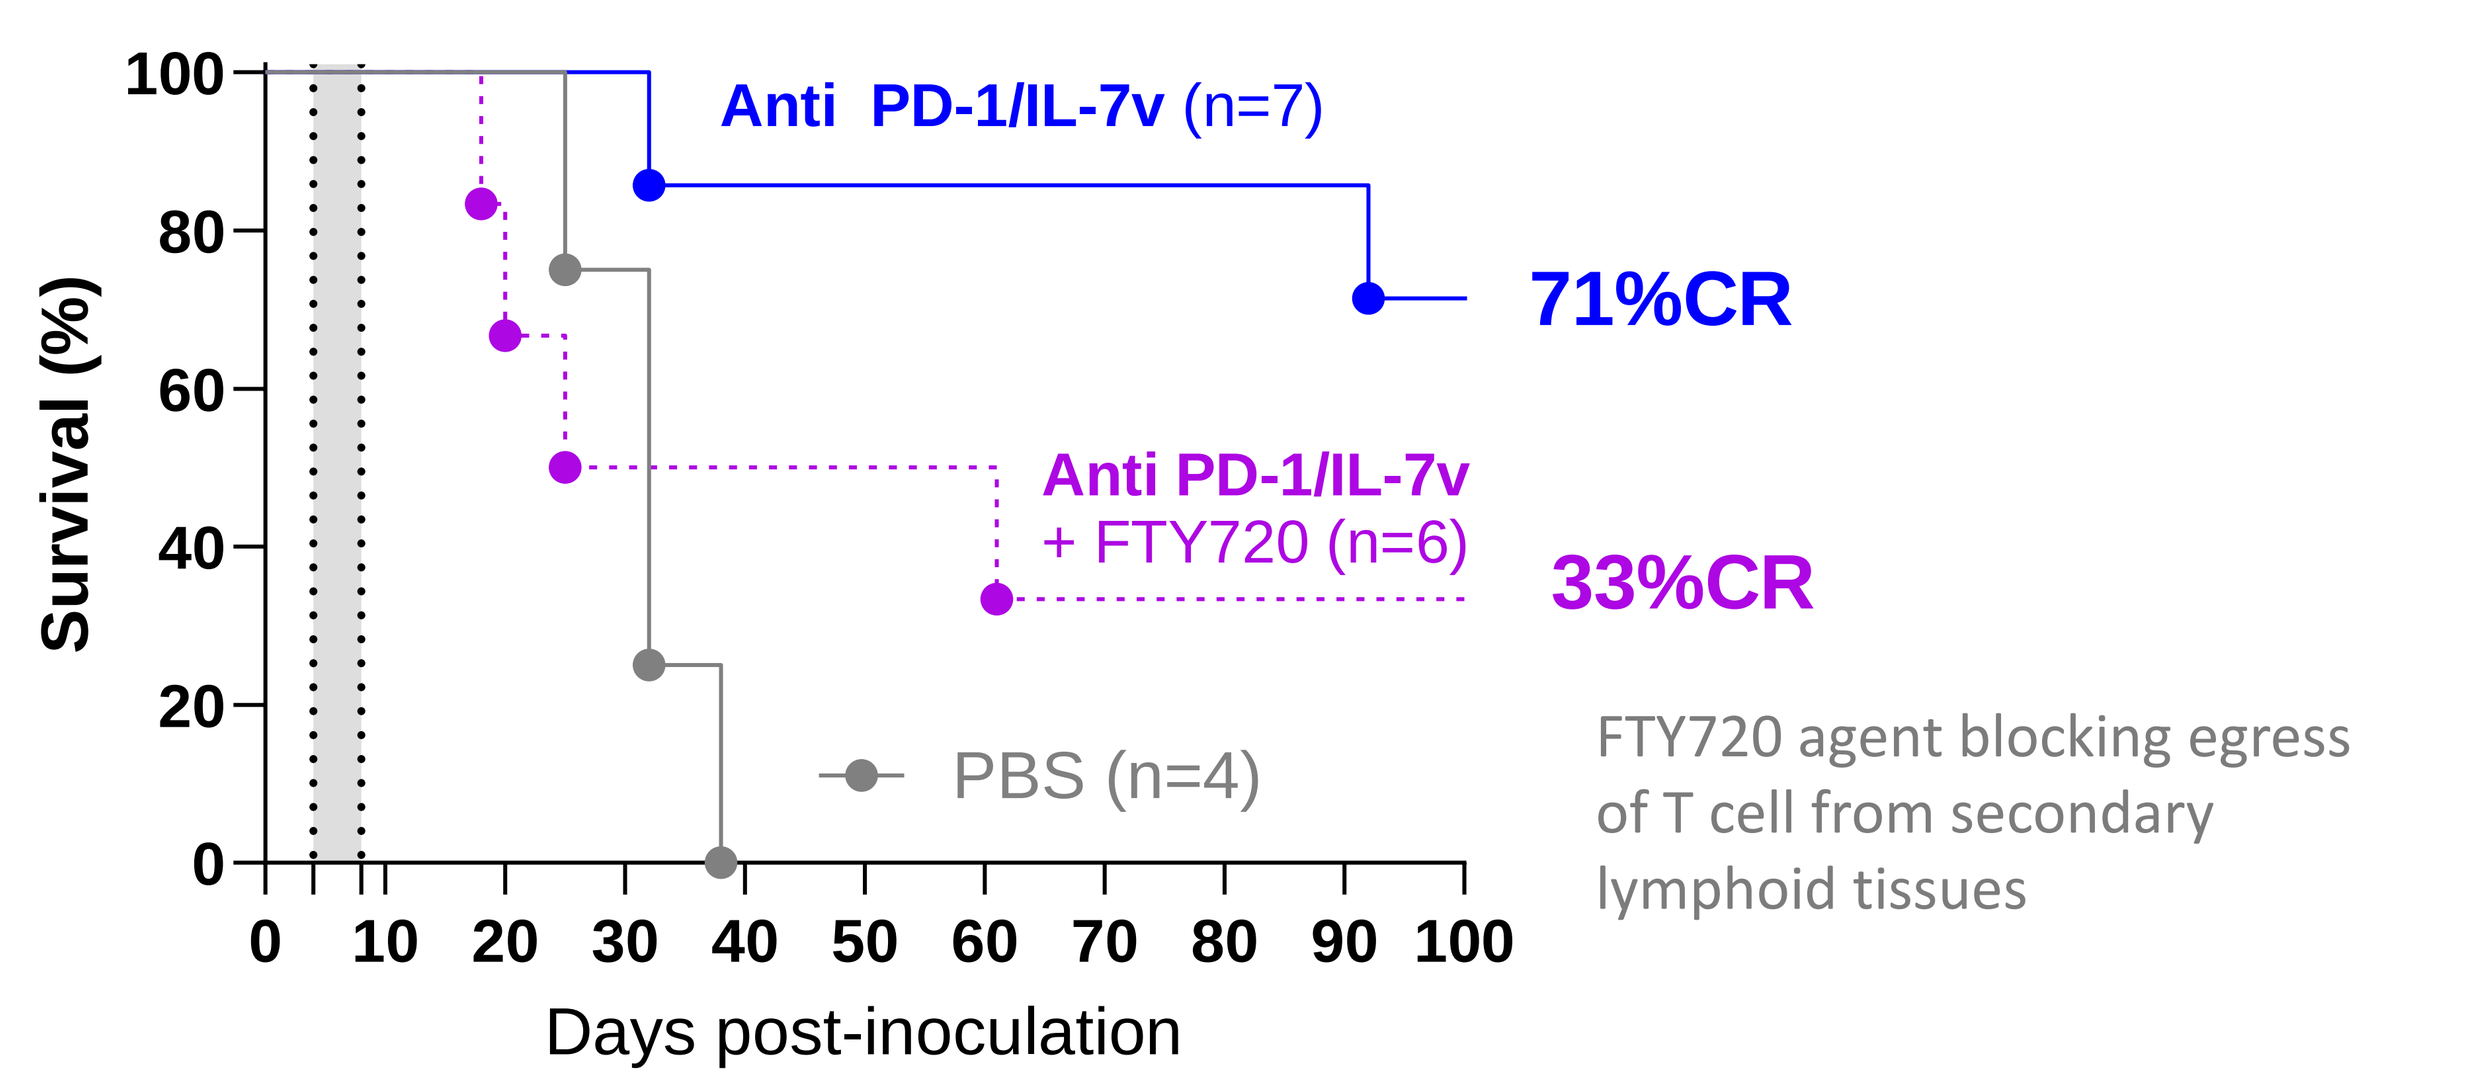
Survival (%)
100
80
60
40
20
0
0
10
20
30
40
50
60
70
80
90
100
Days post-inoculation
Anti PD-1/IL-7v (n=7)
71%CR
Anti PD-1/IL-7v
+ FTY720 (n=6)
33%CR
PBS (n=4)
FTY720 agent blocking egress
of T cell from secondary
lymphoid tissues

BICKI®IL7v favors specific infiltration and proliferation of TCF1+ stem-like T cells

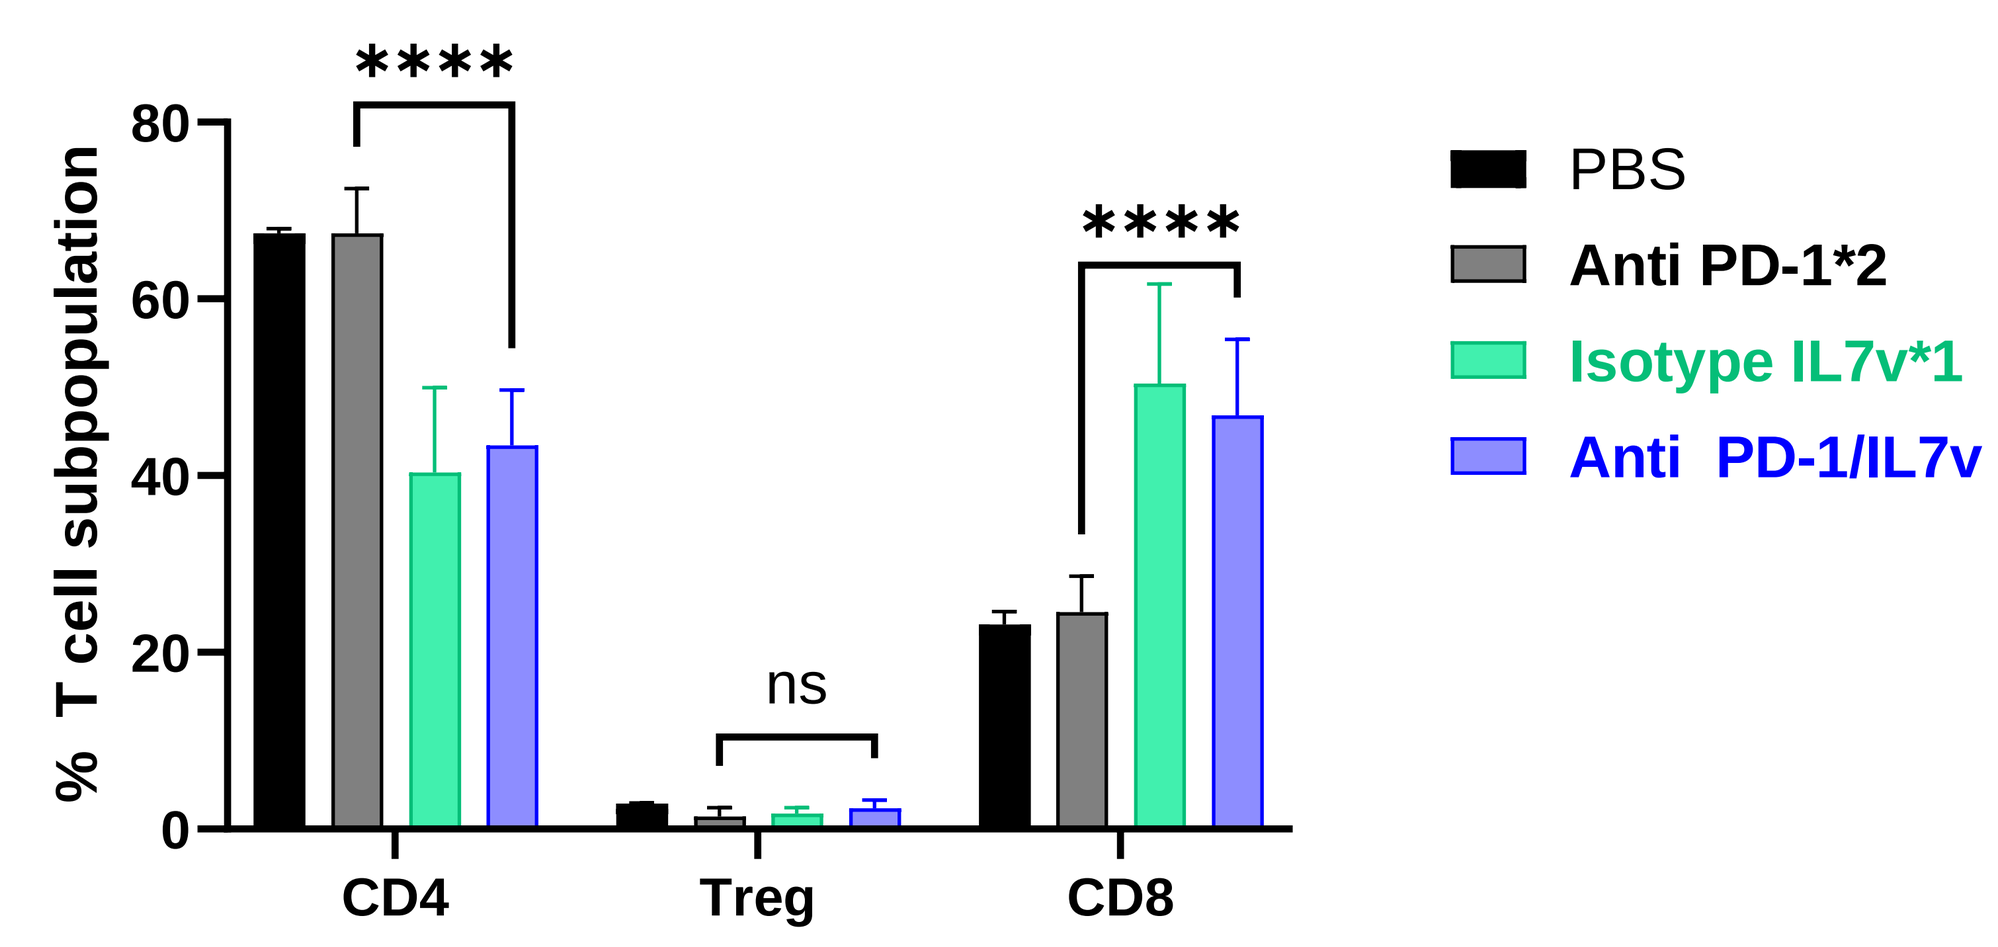
% T cell subpopulation
80
60
40
20
0

ns

CD4
Treg
CD8
PBS
Anti PD-1*2
Isotype IL7v*1
Anti PD-1/IL7v

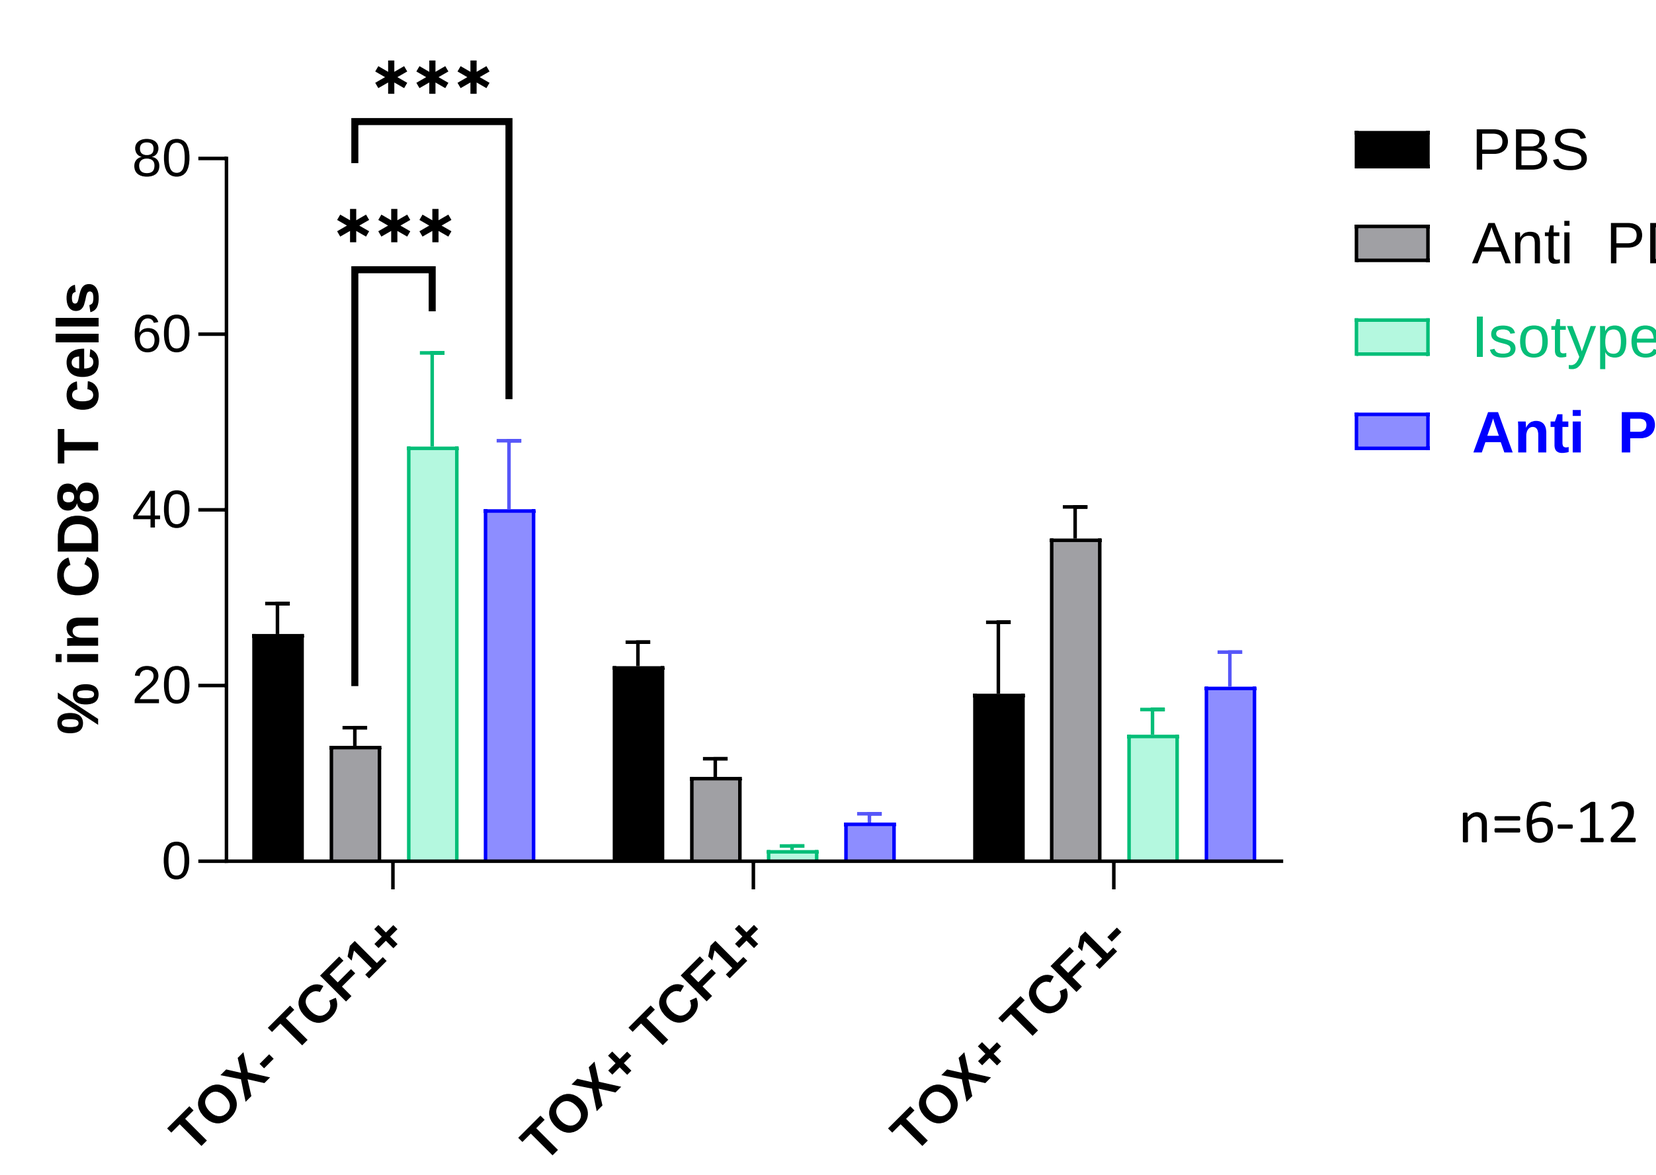
% in CD8 T cells
80
60
40
20
0

TOX- TCF1+
TOX+ TCF1+
TOX+ TCF1-
PBS
Anti PD-1
Isotype IL7v*1
Anti PD-1/IL-7v
n=6-12

BICKI®IL7v overcomes T cell exclusion

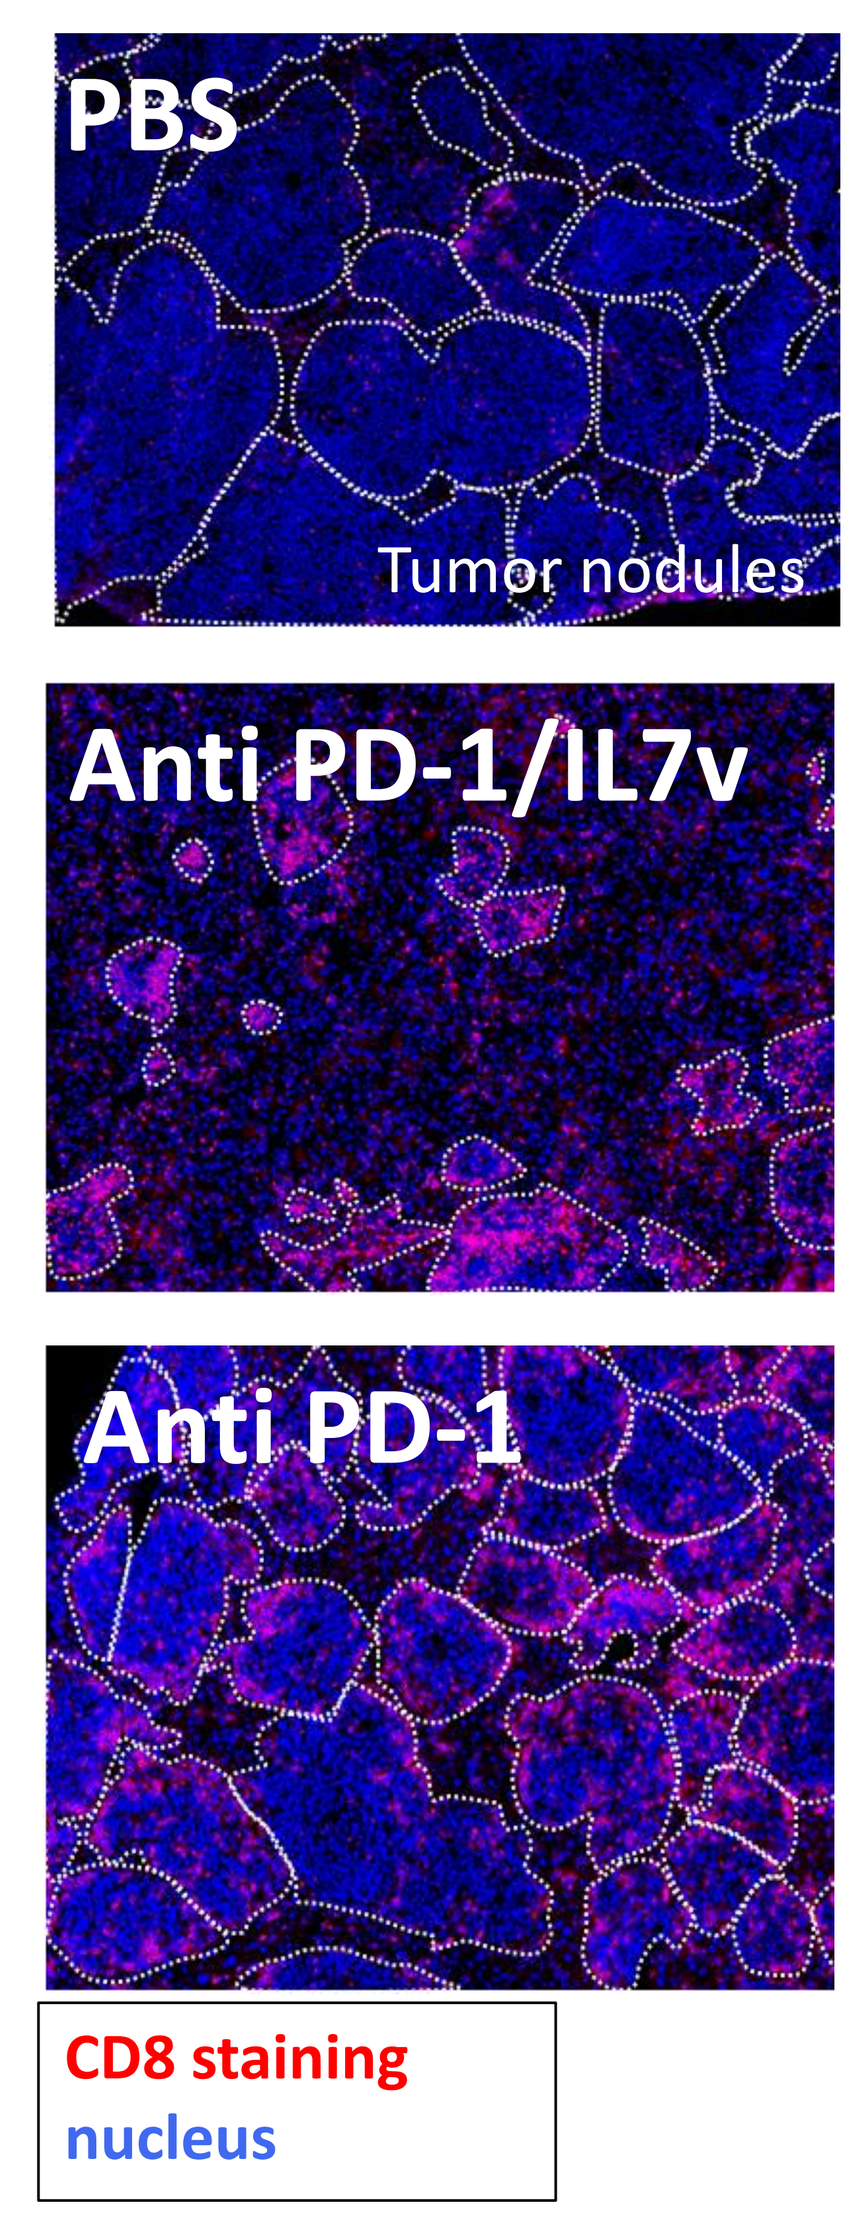
PBS
Tumor nodules
Anti PD-1/IL7v
Anti PD-1
CD8 staining
nucleus

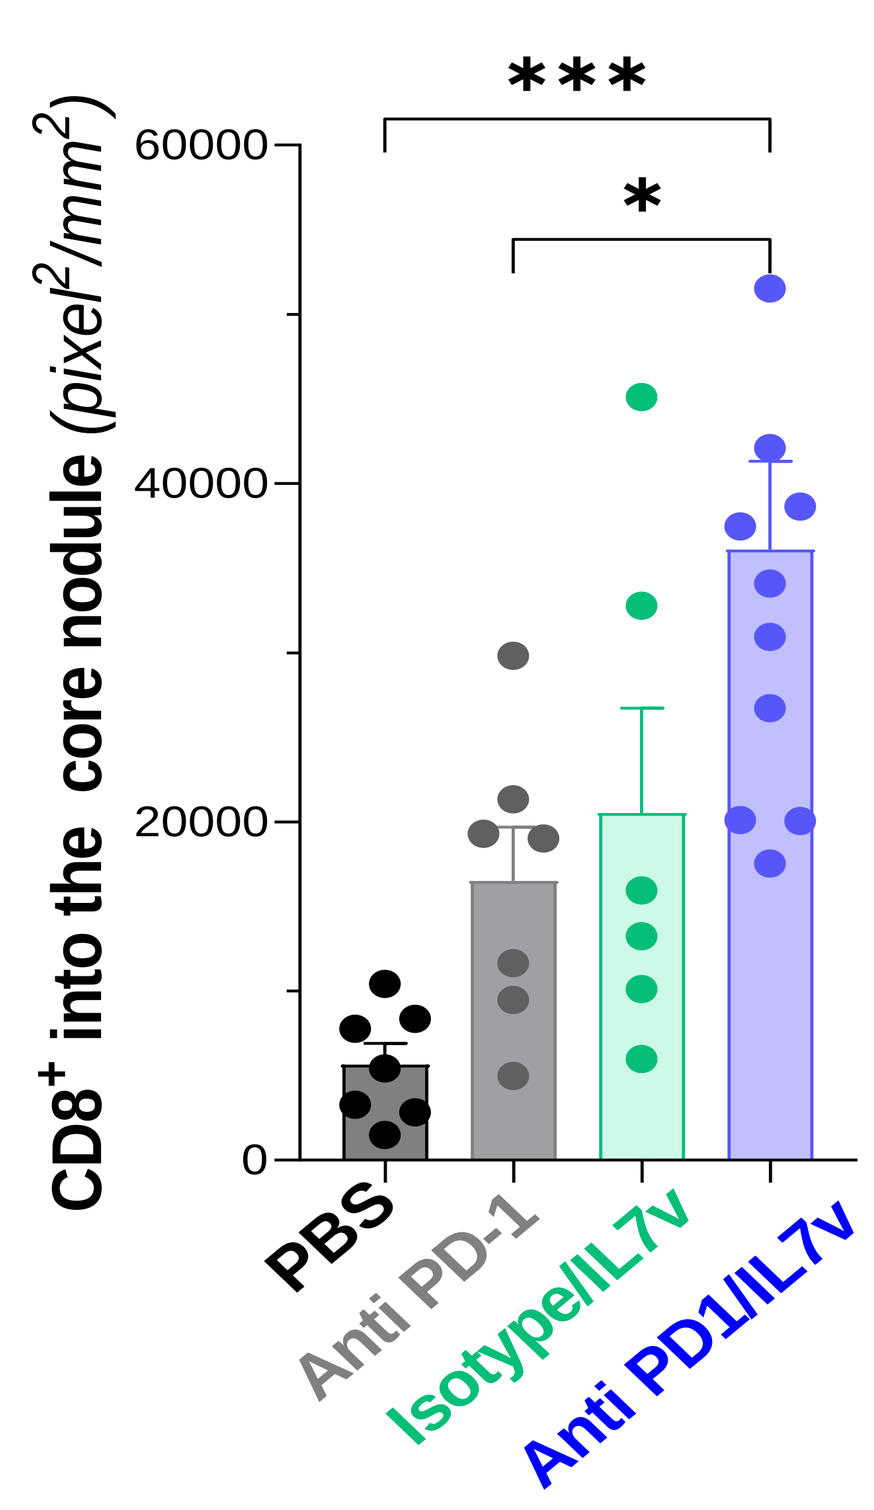
CD8+ into the core nodule (pixel2/mm2)
60000
40000
20000
0

*
PBS
Anti PD-1
Isotype/IL7v
Anti PD1/IL7v

3/ High synergistic efficacy of BICKI®IL7v in combination
with chemotherapeutic or Tyrosine Kinase Inhibitor agents

Combination with Cx
Oxaliplatin

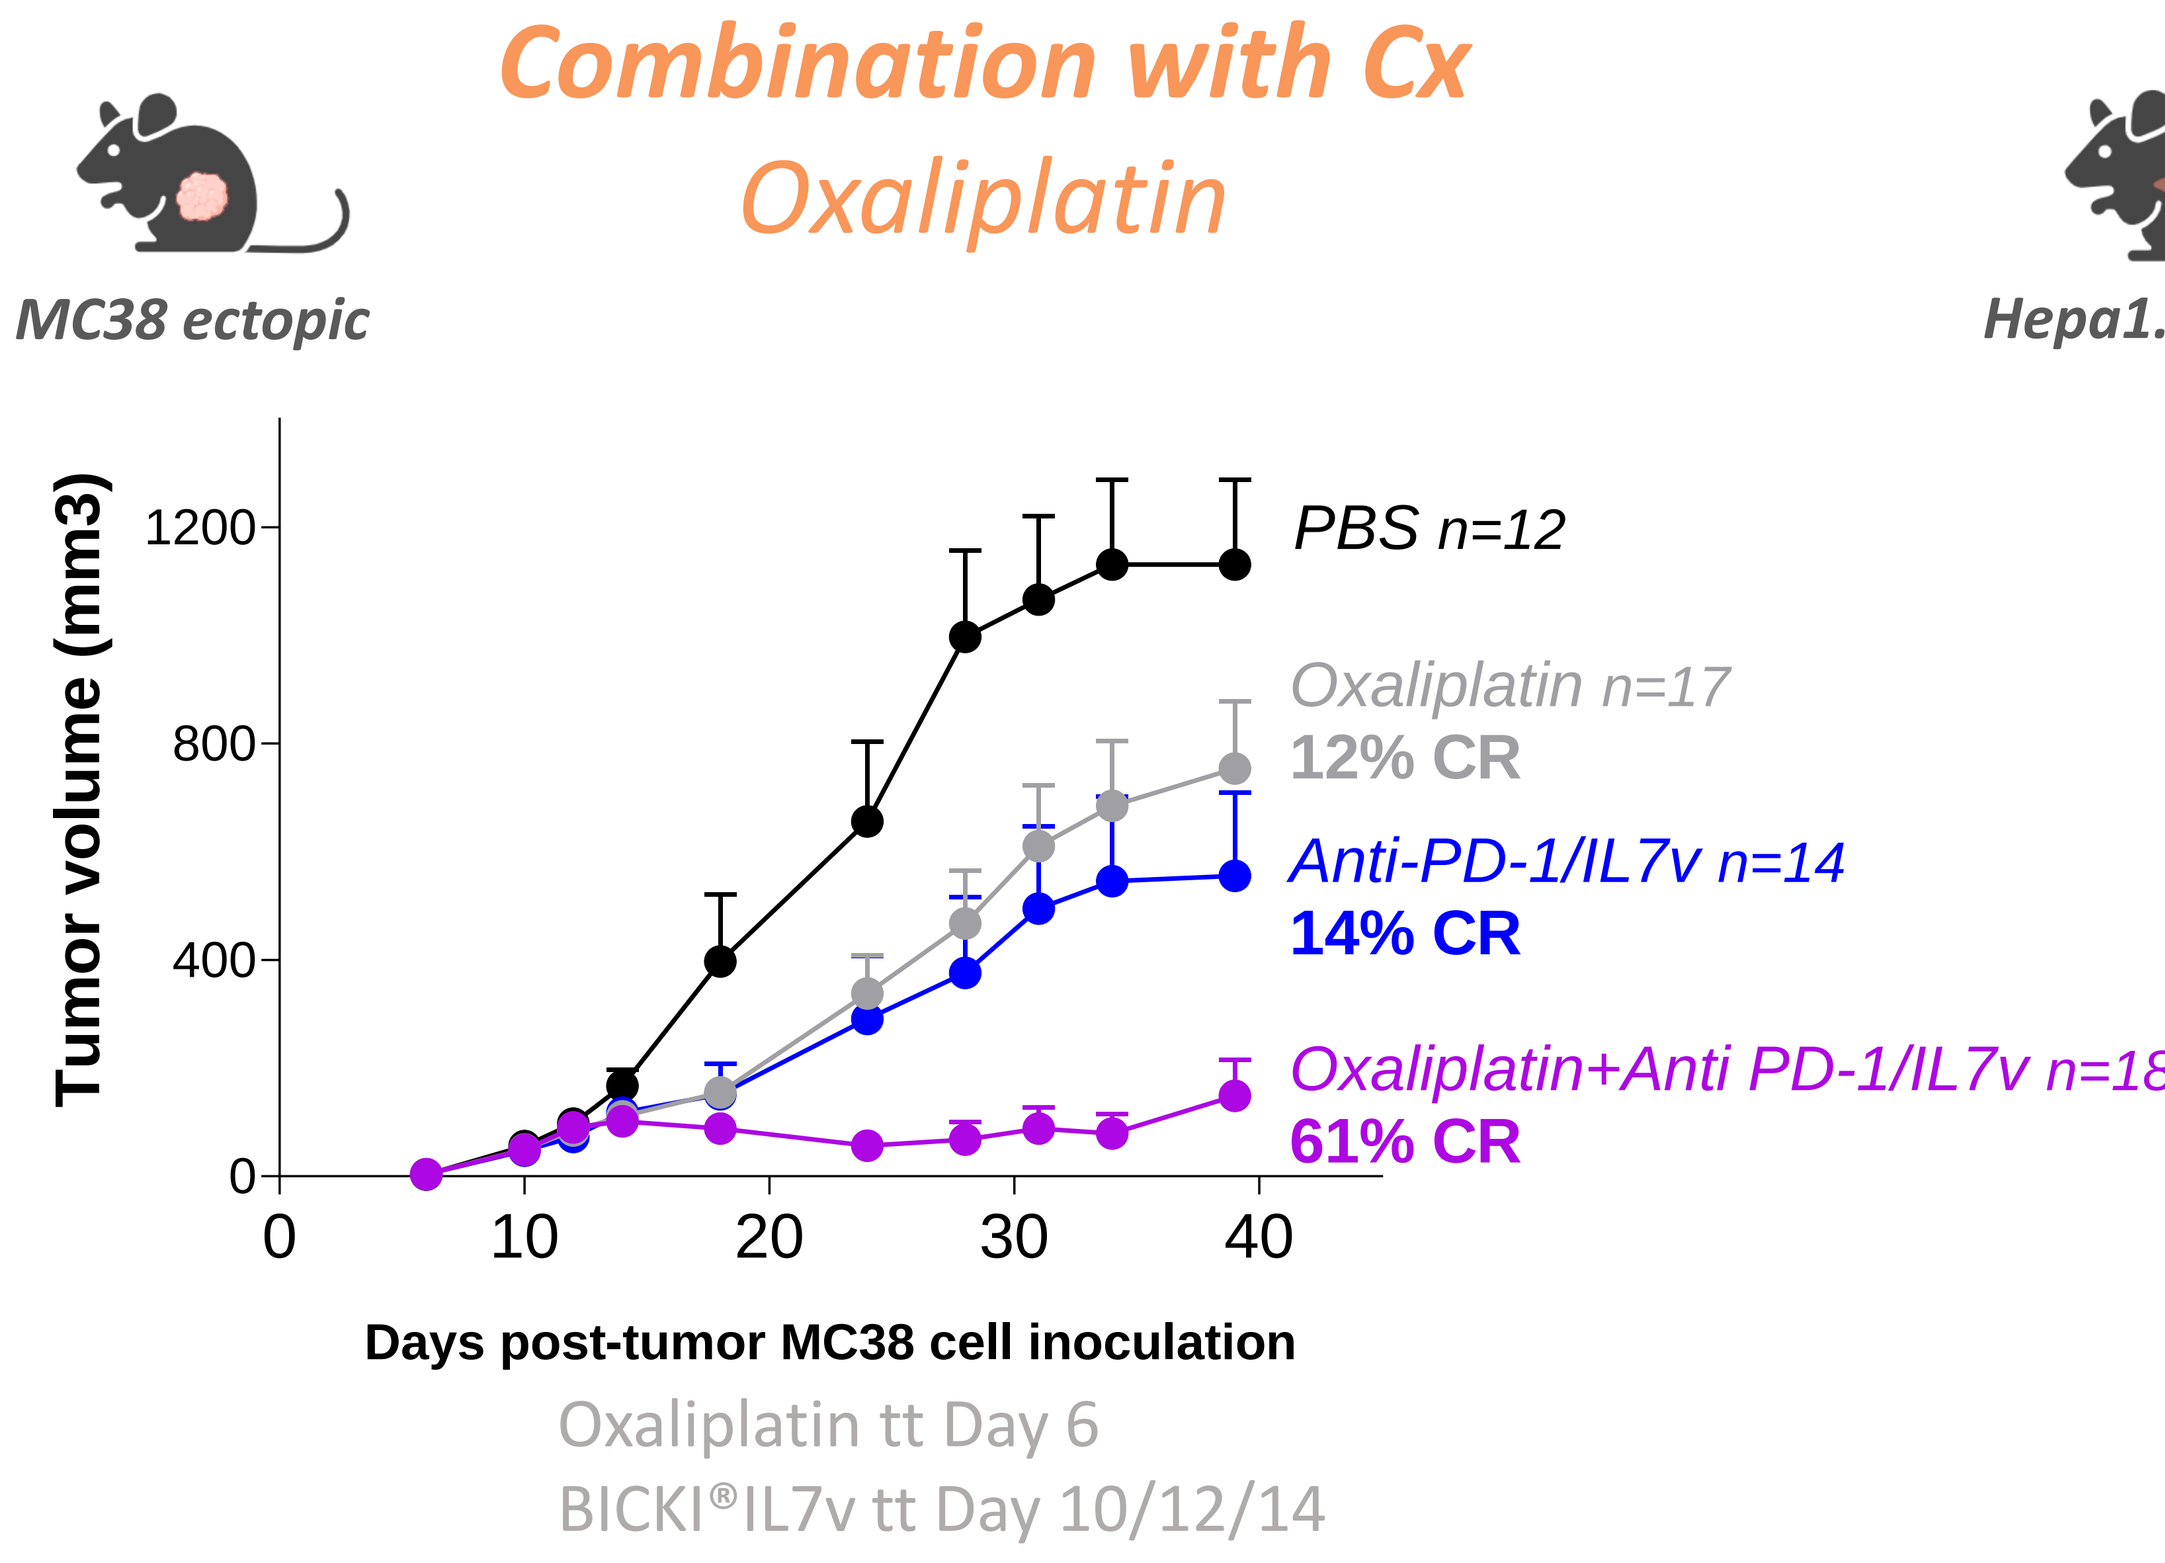
MC38 ectopic
Tumor volume (mm3)
1200
800
400
0
0
10
20
30
40
PBS n=12
Oxaliplatin n=17
12% CR
Anti-PD-1/IL7v n=14
14% CR
Oxaliplatin+Anti PD-1/IL7v n=18
61% CR
Days post-tumor MC38 cell inoculation
Oxaliplatin tt Day 6
BICKI®IL7v tt Day 10/12/14

Combination with TKI
Sorafenib

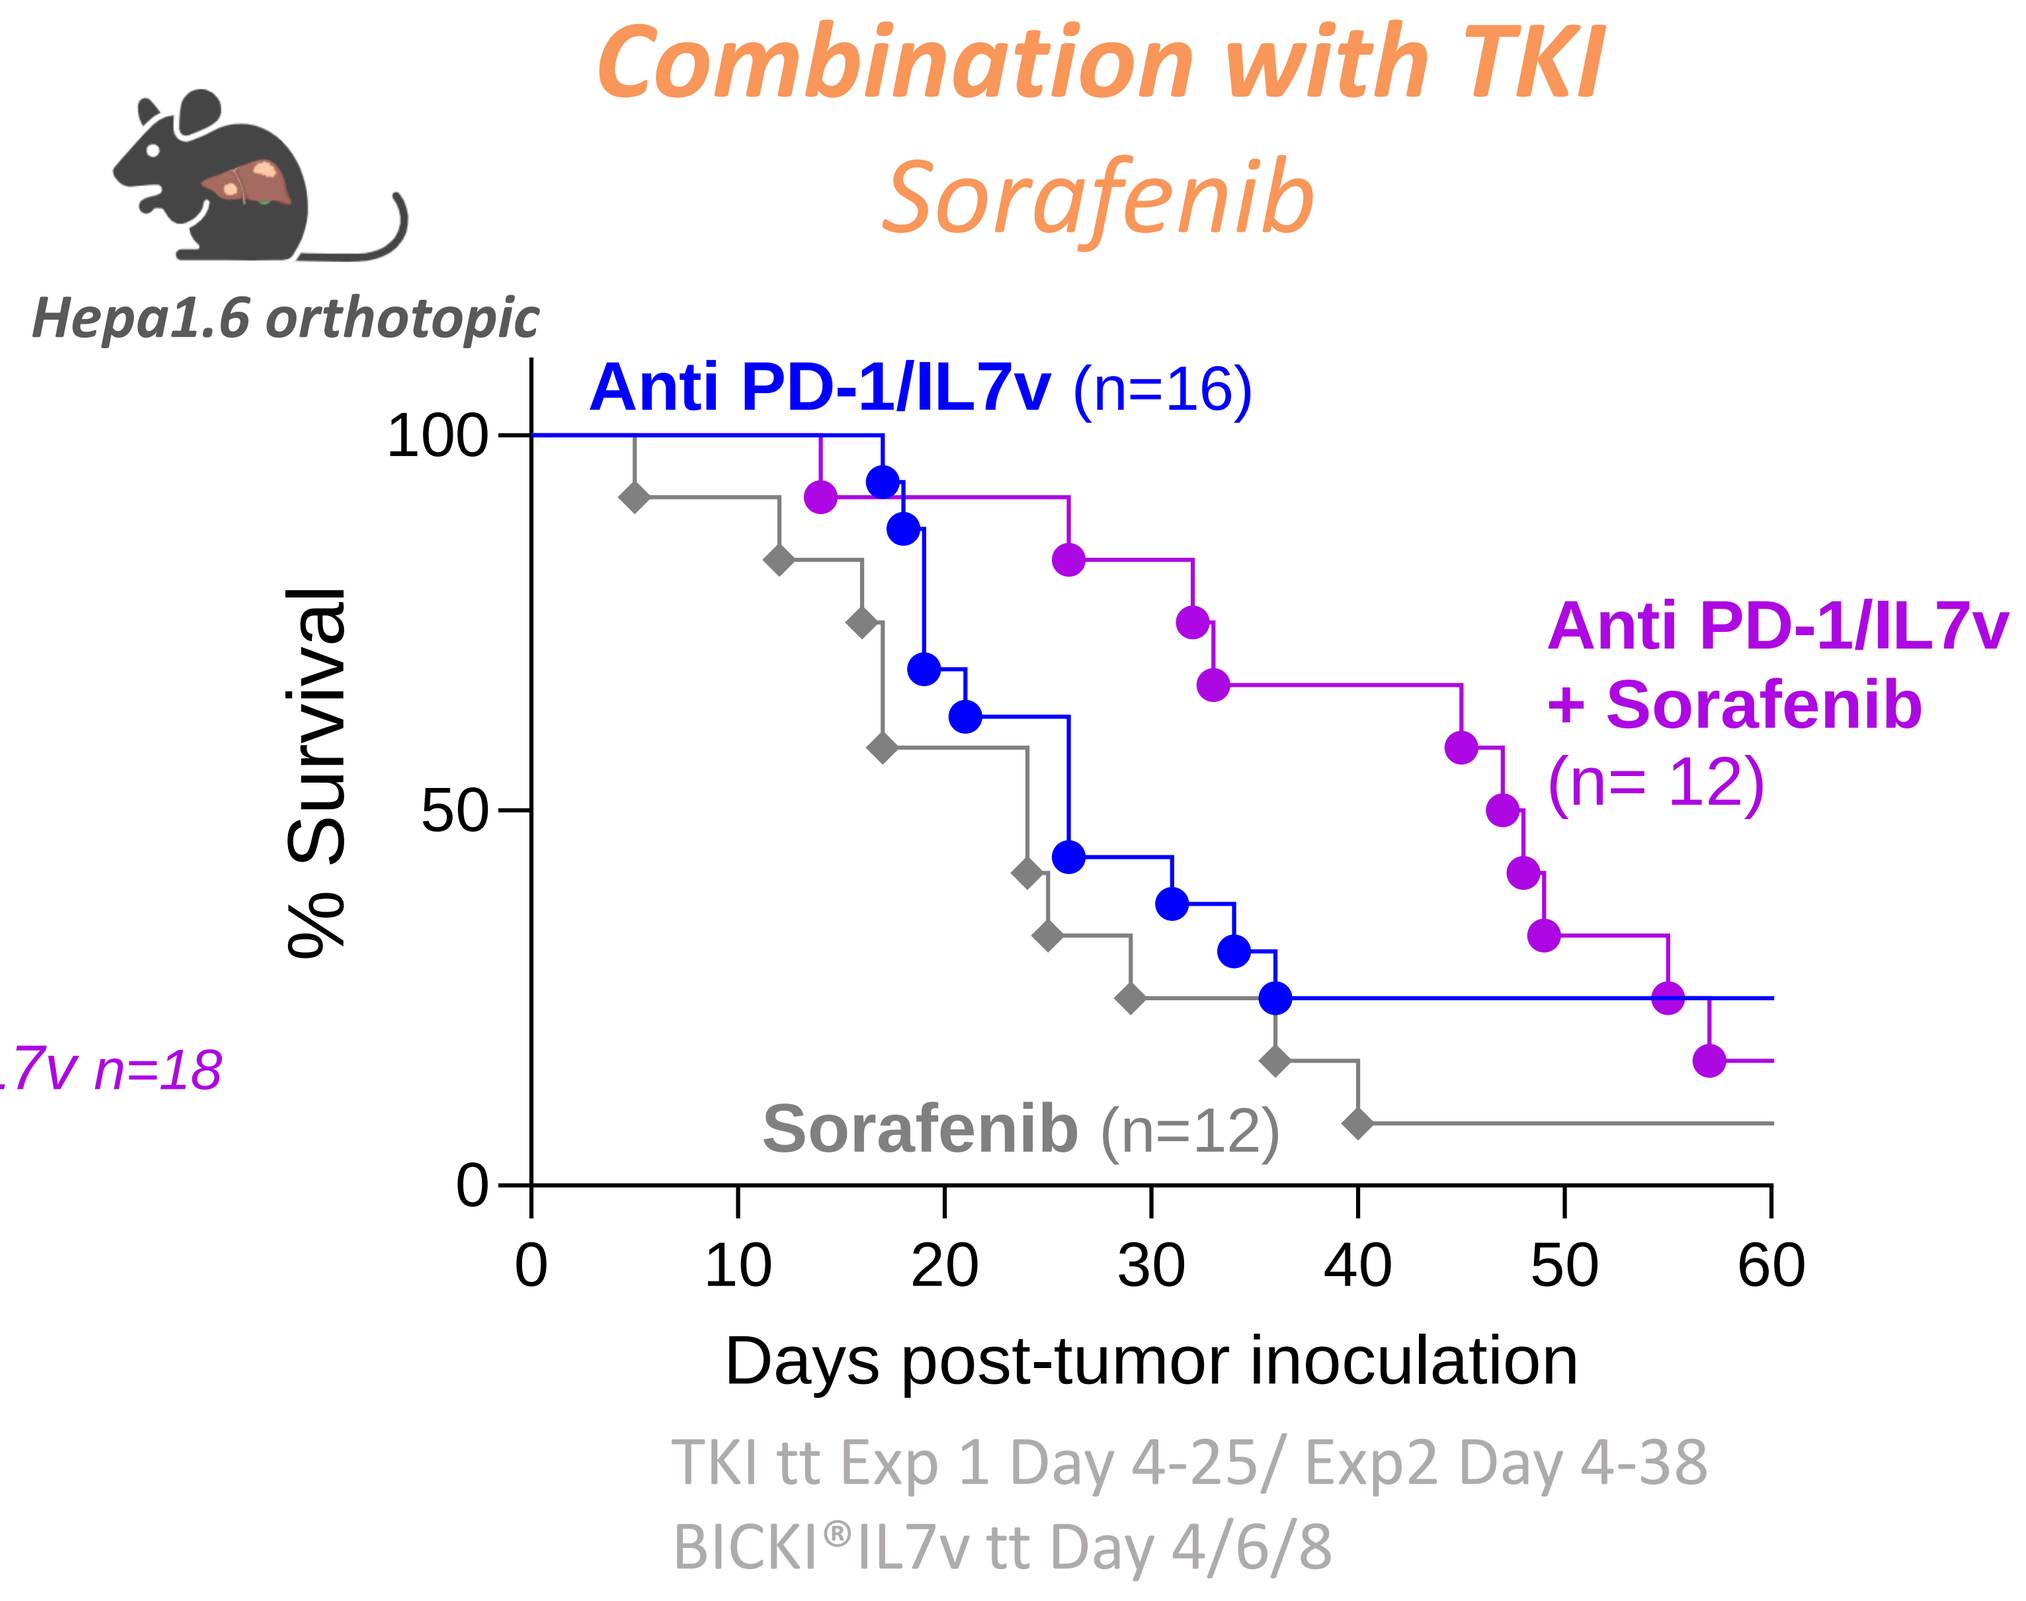
Hepa1.6 orthotopic
Anti PD-1/IL7v (n=16)
% Survival
100
50
0
0
10
20
30
40
50
60
Anti PD-1/IL7v
+ Sorafenib
(n= 12)
Sorafenib (n=12)
Days post-tumor inoculation
TKI tt Exp 1 Day 4-25/ Exp2 Day 4-38
BICKI®IL7v tt Day 4/6/8

Conclusion

BICKI®IL7v Anti PD-1/IL7v immunocytokine
To sustain a durable PD-1+ stem like CD8 T cell response

- Optimized molecule for Cis targeting activity
- Conserved high PD-1 binding and PD-1/PD-L1 antagonist activity
- Allows a selective delivery of IL-7 on PD-1+ cells
- Promotes proliferation of PD-1+ CD127+ TCF1+ stemness T cells
- High *in vivo* anti tumor efficacy in monotherapy or combo *in PD-1 sensitive, resistant syngeneic and humanized models (8 different models)*

Contacts
nicolas.poirier@ose-immuno.com
aurore.morello@ose-immuno.com